P-1110

The Impact of Xeruborbactam on *in vitro* Activity of Cefiderocol against a Challenge Panel of Enterobacterales Enriched with Isolates with Increased Cefiderocol MICs

IDWeek 2024
Los Angeles, CA
October 16–19, 2024

Takafumi Hara[1], Naoki Ishibashi[1], Dai Miyagawa[1], Motoyasu Onishi[1], Olga Lomovskaya[2], Yoshinori Yamano[1]

[1]Shionogi & Co., Ltd., Osaka, Japan; [2]Qpex Biopharma Inc., San Diego, USA

Contact information:
Takafumi Hara
3-1-1, Futaba-cho, Toyonaka,
Osaka 561-0825, Japan
Phone: +81-70-7812-7383
Email: Takafumi.hara@shionogi.co.jp

SHIONOGI

BACKGROUND

- Cefiderocol (FDC) is a siderophore cephalosporin with potent activity against resistant gram-negative bacteria. According to SIDERO-WT 2014 to 2019 surveillance studies that involved testing of over 30,000 Enterobacterales (ENT) isolates collected in North America and Europe, 99.8% were susceptible (S) to FDC (by CLSI breakpoint) (MIC_{90}=1 µg/mL). Against meropenem non-susceptible (non-S) ENT, 96.5% were susceptible to FDC (MIC_{90}=4 µg/mL).
- Resistance to FDC in ENT does not appear frequently, but when it occurs it appears to be multifactorial and requiring production of certain serine (SBL) or metallo β-lactamases (MBL) (J Glob Antimicrob Resist. 2020 22:738-741).
- Xeruborbactam (XER) is a novel β-lactamase inhibitor that inhibits numerous SBL and MBL. Previous study demonstrated that XER enhanced potency of multiple β-lactam antibiotics against meropenem non-susceptible Enterobacterales that produced various SBL and MBL β-lactamases (Antimicrob Agents Chemother. 2023 67(11):e0044023).

OBJECTIVE

- Evaluate the impact of XER on the activity of FDC against a challenge panel of ENT highly enriched in isolates with elevated FDC minimum inhibitory concentration (MIC) values.

MATERIALS AND METHODS

- A challenge panel of 165 ENT isolates was selected from SIDERO-WT and clinical isolates. The panel contained 139 carbapenem resistant isolates. 79 (47.9%) and 41 (24.8%) of isolates had FDC MIC values >2 µg/mL and >4 µg/mL (non-S by EUCAST/CLSI breakpoint), respectively.
- The β-lactamase profile for each isolate was determined by PCR or whole genome sequencing.
- MICs were determined for FDC alone, in combination with 1, 2 and 4 µg/mL of XER according to CLSI guidelines for FDC using iron-depleted cation-adjusted Mueller-Hinton broth (CAMHB). XER alone was also tested using iron-depleted CAMHB. The other antimicrobials were tested using CAMHB.

RESULTS

- MIC_{50}/MIC_{90} of FDC alone and XER alone against the challenge panel were 2/16 and 16/>16 µg/mL, respectively. Dose-dependent shifts to lower FDC MICs were observed in the presence of increasing concentrations of XER for the challenge panel and individual subsets (Figure 1, Table 1).
- For isolates non-S to FDC (MIC >4 µg/mL), 4 µg/mL of XER reduced the FDC MIC_{90} from 32 to 1 µg/mL and the MIC_{50} from 16 to 0.25 µg/mL.
- FDC-XER (at fixed 4 µg/mL XER) showed the highest activity against this challenge panel compared to other agents tested (Table 2).

Figure 1: MIC distribution of cefiderocol alone and with xeruborbactam against challenge panel enriched in cefiderocol-non-susceptible isolates (N=165)

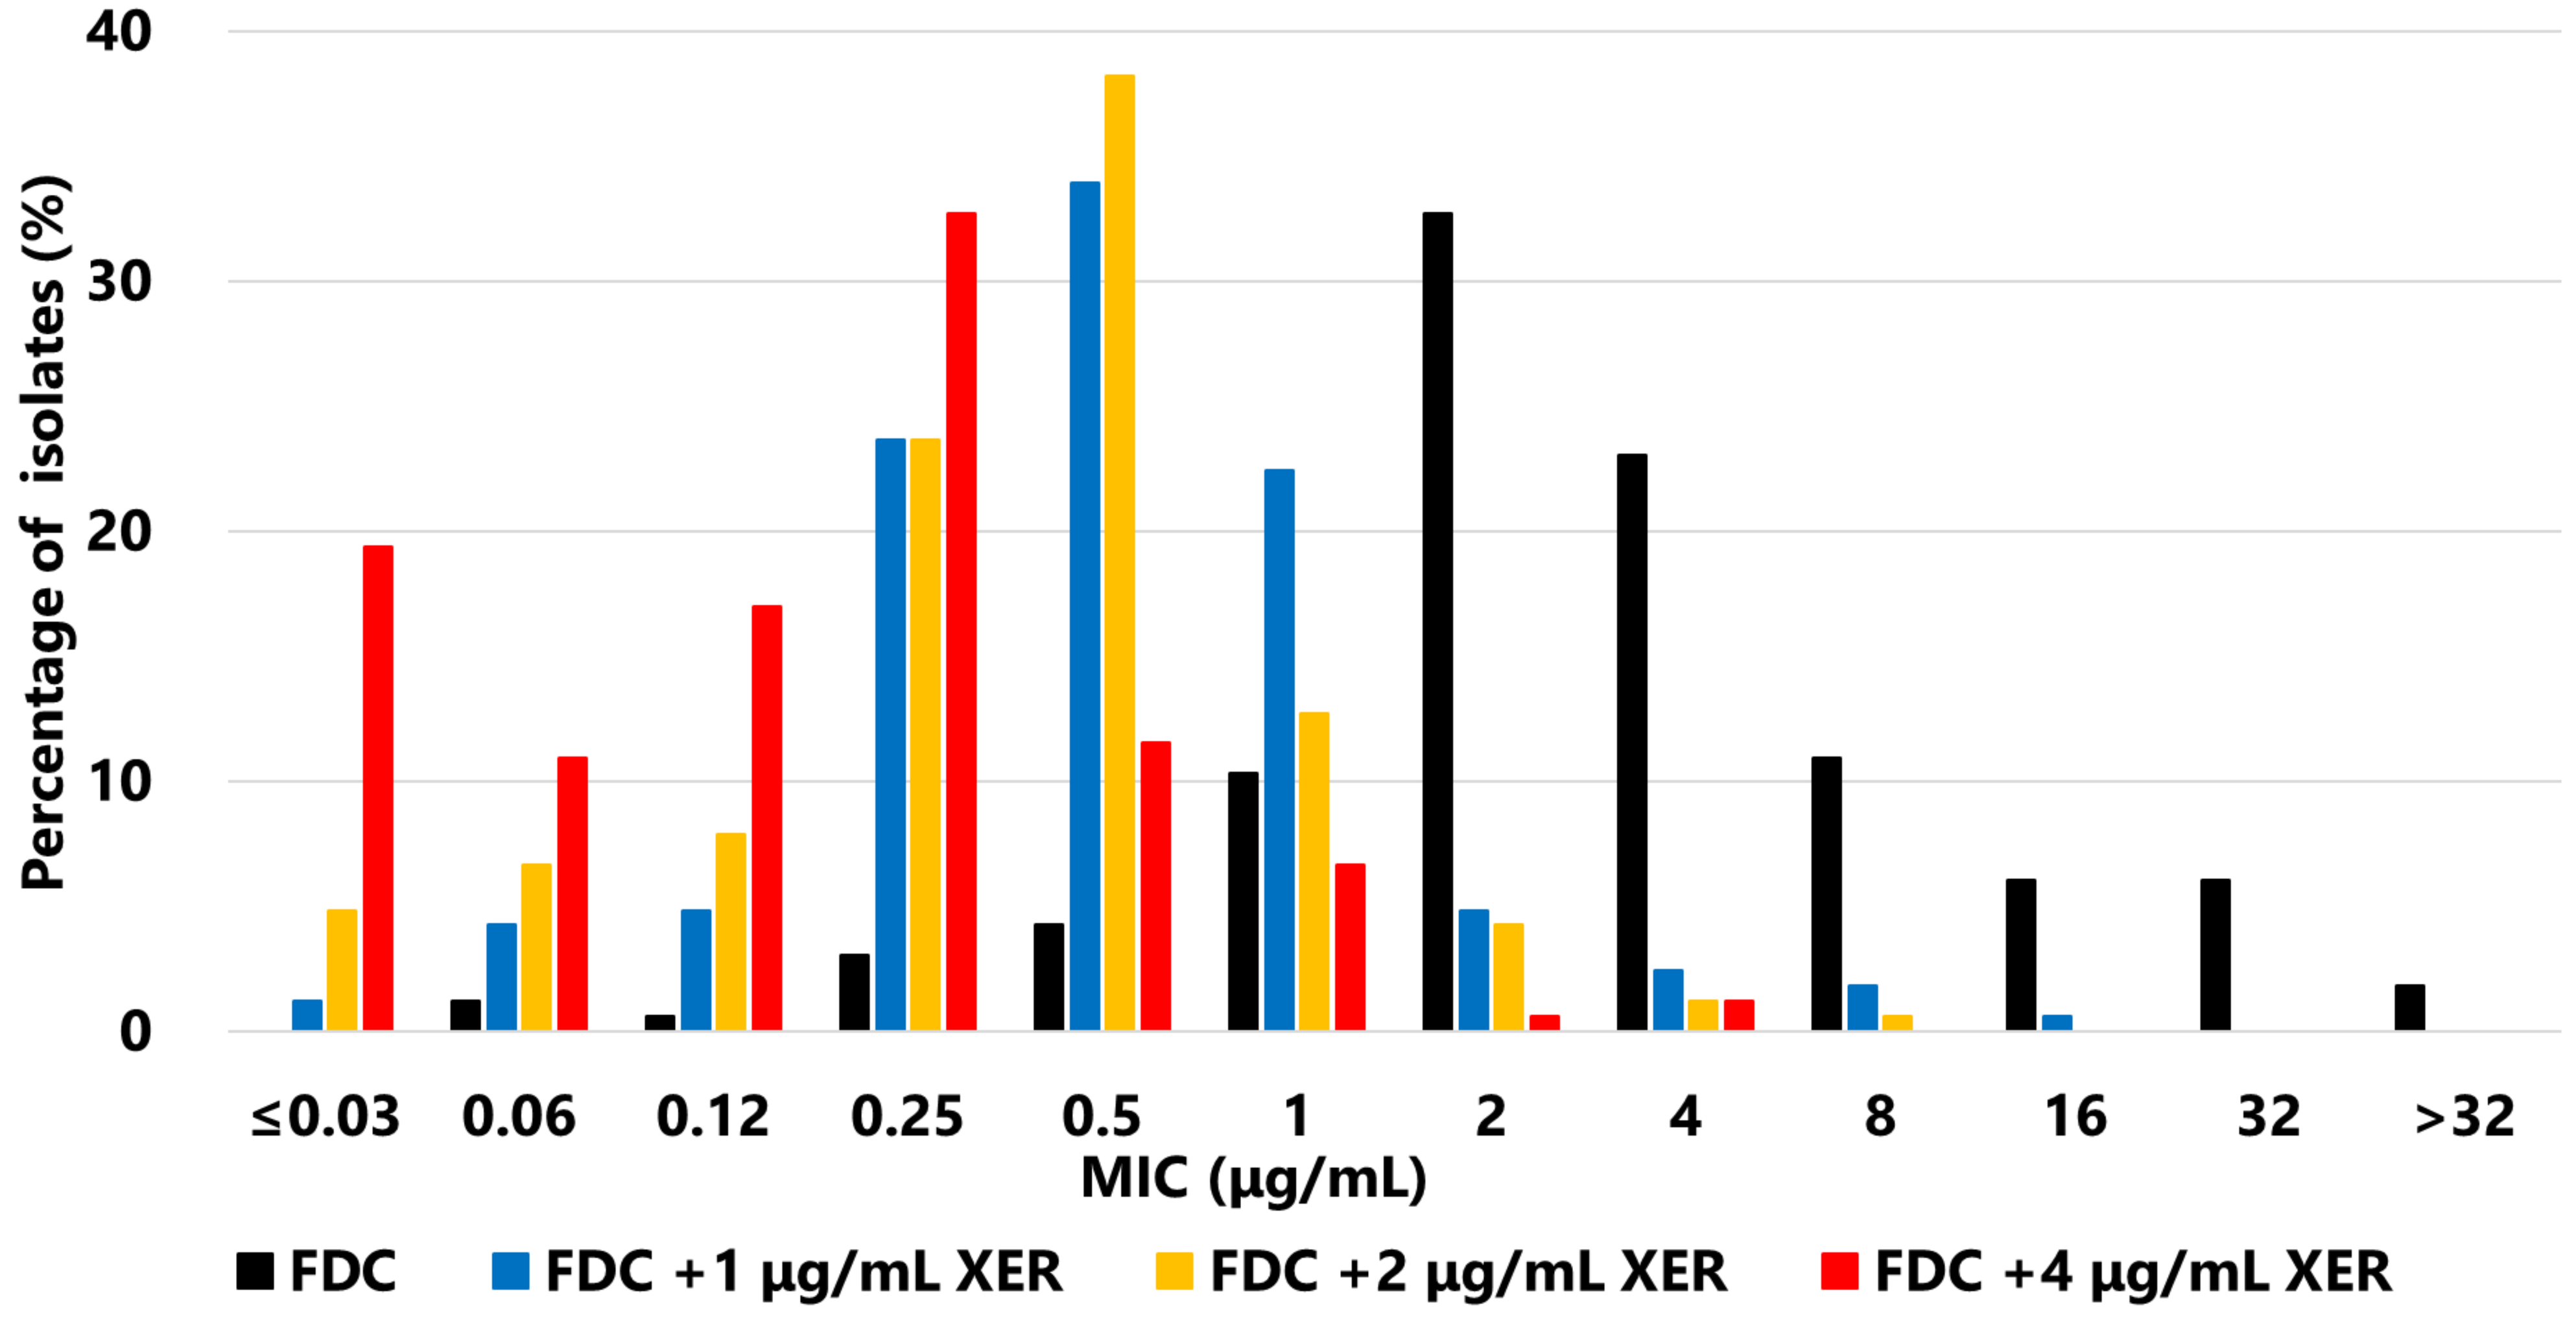
Percentage of isolates (%)
0
10
20
30
40
≤0.03
0.06
0.12
0.25
0.5
1
2
4
8
16
32
>32
MIC (µg/mL)
FDC
FDC +1 µg/mL XER
FDC +2 µg/mL XER
FDC +4 µg/mL XER

FDC: cefiderocol, XER: xeruborbactam

Table 1: $MIC_{50/90}$ (µg/mL) of cefiderocol alone and with xeruborbactam against challenge panel enriched in cefiderocol-non-susceptible isolates

	Total (N = 165)	FDC MIC>4 (N=41)	MBL (N = 67)
FDC	2 / 16	16 / 32	4 / 32
FDC +1 µg/mL XER	0.5 / 1	1 / 4	0.5 / 2
FDC +2 µg/mL XER	0.5 / 1	0.5 / 2	0.5 / 1
FDC +4 µg/mL XER	0.25 / 0.5	0.25 / 1	0.12 / 0.5
XER	16 / >16	16 / >16	16 / >16

FDC: cefiderocol, XER: xeruborbactam; MBL: metallo-β-lactamase
Of the 41 isolates with FDC MIC>4, 28 had NDM-type MBL.

Acknowledgments
This study has been funded in whole or in part with Federal funds from the Department of Health and Human Services; Office of the Assistant Secretary for Preparedness and Response; Biomedical Advanced Research and Development Authority (BARDA), under OTA number HHSO100201600026C.

Table 2: $MIC_{50/90}$ (µg/mL) of antimicrobials against challenge panel enriched in cefiderocol-non-susceptible isolates

	Total (N = 165)	FDC MIC>4 (N=55)	MBL (N = 67)	KPC (N = 60)	OXA-48 (N = 11)	Carbapenemase negative (N=27)
FDC-XER	0.12 / 1	0.5 / 2	0.25 / 1	0.12 / 0.5	0.5 / 1	0.06 / 0.5
MVB	4 / >64	64 / >64	64 / >64	0.5 / 4	8 / 16	0.12 / >64
I-R	4 / 64	32 / 64	32 / >64	1 / 4	4 / 4	1 / 16
CZA	4 / >64	>64 / >64	>64 / >64	2 / 8	2 / 8	1 / >64
C/T	>64 / >64	>64 / >64	>64 / >64	>64 / >64	>64 / >64	4 / >64
ATM-AVI	0.25 / 2	0.25 / 8	0.25 / 1	0.25 / 1	0.25 / 16	0.25 / 16
CST	2 / 32	1 / 32	1 / 32	2 / 32	2 / 16	2 / 16
TGC	1 / 4	1 / 4	1 / 4	1 / 2	1 / 2	1 / 2

FDC: cefiderocol, XER: xeruborbactam, MVB: meropenem-vaborbactam, I-R: imipenem-relebactam,
CZA: ceftazidime-avibactam, C/T: ceftolozane-tazobactam, ATM: aztreonam, AVI: avibactam, CST: colistin,
TGC: tigecycline, MBL: metallo-β-lactamase
All β-lactamase inhibitors except for vaborbactam were used at fixed 4 µg/mL, and only vaborbactam was used at fixed 8 µg/mL.
Of the 55 isolates with FDC MIC>4, 37 had NDM-type MBL.
Of the 67 MBL isolates, 58 had NDM-type MBL and the other 9 had VIM-1.

CONCLUSIONS

- XER strongly improved FDC activity against rarely encountered isolates of ENT with increased FDC MIC and showed the highest activity compared to other agents tested.
- FDC-XER is a promising combination with good activity against highly resistant ENT isolates that warrants further development

＜**Conflict of interest statement**＞
Funding: This research was sponsored by Shionogi & Co., Ltd. and Qpex Biopharma Inc.
Conflict of interest: TH, NI, DM, MO, OL and YY are employees of Shionogi & Co., Ltd. or Qpex Biopharma Inc.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from IDWeek 2024 and the authors of the poster.