Poster 1475

IDWeek 2024, Los Angeles, CA, USA
October 16–19, 2024

Contact: Bin Cai, MD
Email: bin.cai@shionogi.com

Real-World Effectiveness and Safety of Cefiderocol in the Treatment of Patients with Serious Gram-Negative Bacterial Infections: Results of the PROVE Chart Review Study

Cornelius Clancy[1], Oliver A. Cornely[2], Christine Slover[3], Sean T. Nguyen[3], Frank Kung[3], Stefano Verardi[4], Christopher Longshaw[4], Stephen Marcella[3], and Bin Cai[3]

[1]University of Pittsburgh, Pittsburgh, PA, USA; [2]University of Cologne, Cologne, Germany; [3]Shionogi Inc., Florham Park, NJ, USA; [4]Shionogi B.V., London, UK

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from IDWeek 2024 and the authors of the poster.

SHIONOGI

Abstract (Revised data)

Background: The PROVE retrospective chart review study aimed to assess real-world outcomes of cefiderocol treatment in patients with serious Gram-negative bacterial infections.
Methods: We analyzed data of 1075 hospitalized patients with suspected or confirmed Gram-negative bacterial infections in Europe and the USA treated with cefiderocol for the first time for ≥72 hours (Nov 2020–Feb 2024). Baseline demographics, clinical characteristics, cefiderocol use, clinical cure, clinical response at end of treatment (EOT), in-hospital all-cause mortality (ACM), and adverse drug reactions (ADRs) were assessed.
Results: Of 1075 patients analyzed, 705 (65.6%) were men and the median age (interquartile range [IQR]) was 60 (46–69) years; 88.7% of patients had ≥1 risk factor for carbapenem-resistant infections. Overall, 56.6% were in the intensive care unit (ICU) and 44.3% received organ support. Cefiderocol was used for a documented infection in 71.2% of patients; the median (IQR) duration was 11 (7–16) days. The most frequent infection site was the respiratory tract (RTI; 53.1%), and the most frequent species in monomicrobial infections was *Pseudomonas aeruginosa* (35.9%), followed by *Acinetobacter baumannii* (18.1%). Overall, 67.5% of patients had clinical cure and 75.1% responded to cefiderocol treatment at EOT. ACM rate at day 30 was 23.3%. Clinical cure and clinical response rates were 61.8% and 71.6% in patients with RTI, and 68.5% and 74.1% in patients with bloodstream infection (BSI), respectively. Among patients with RTI or BSI, 29.2% and 19.4%, respectively, died by Day 30. Among patients with *P. aeruginosa* infection, 72.0% and 79.3% had clinical cure and clinical response, respectively, and 19.9% died by Day 30. In monomicrobial and polymicrobial infections, clinical cure rates were 67.8% and 66.1%, while ACM rates were 23.5% and 22.9% at Day 30, respectively. A total of 25 patients had 29 ADRs or serious ADRs, which led to discontinuation of cefiderocol for 16 events.
Conclusions: Cefiderocol was efficacious and well tolerated in the treatment of serious Gram-negative bacterial infections in real-world settings in a patient population with high rates of ICU admission and organ support.

Objectives

- The main objectives of the PROVE study were to describe the patient characteristics and clinical outcomes in patients treated with cefiderocol for Gram-negative bacterial infections in real-world settings between 2020 and 2024.

Methods

- PROVE was an international, multicenter, retrospective medical chart review study of first-time cefiderocol use in patients with Gram-negative bacterial infections.[1]
- Eligibility criteria included having a documented Gram-negative bacterial infection, which prompted the use of cefiderocol for at least 72 hours, and availability of data on cefiderocol starting dose, description of the Gram-negative bacterial infection for which cefiderocol was prescribed, and discharge data after hospitalization.[1]
- Clinical cure was defined as resolution or improvement in infection signs and symptoms, without evidence of a later relapse or death. Clinical response was defined as resolution or improvement of signs/symptoms at EOT as judged by the physician, excluding patients who died during therapy.[1]
- Only descriptive data are collected. Further data analyses are ongoing.

Conclusions

- Cefiderocol treatment, administered primarily for patients with documented carbapenem-resistant Gram-negative bacterial infections and including a large proportion of critically ill patients, was effective and well tolerated in real-world settings in the USA and Europe.
- Similar effectiveness was demonstrated across a range of Gram-negative infections, including those that were polymicrobial.

Results

- A total of 1075 hospitalized patients were enrolled, and 65.6% were male.
- The median age was 60 years (IQR: 46–69) and the median CCI was 2 (IQR: 1–4).
- Participating countries: USA 47.3%, Spain 14.8%, France 11.9%, Italy 11.3%, UK 7.5%, Germany 7.3%.
- The median cefiderocol treatment duration was 11 days (IQR: 7–16), and 33.9% of patients received combination therapy.

Comorbidities

20.4%	Chronic pulmonary disease	11.5%	Diabetes with end-organ damage
18.5%	Uncomplicated diabetes	9.4%	Myocardial infarction
17.0%	COVID-19	4.1%	Cystic fibrosis
16.9%	Moderate-severe renal disease	3.9%	Burns
14.0%	Congestive heart failure	2.5%	Bronchiectasis

Infection characteristics

Distribution of primary infection sites

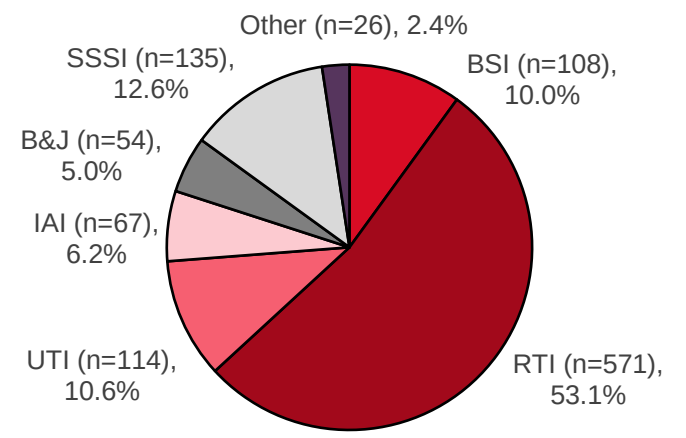
Other (n=26), 2.4%
BSI (n=108), 10.0%
SSSI (n=135), 12.6%
B&J (n=54), 5.0%
IAI (n=67), 6.2%
UTI (n=114), 10.6%
RTI (n=571), 53.1%

Distribution of baseline Gram-negative pathogens

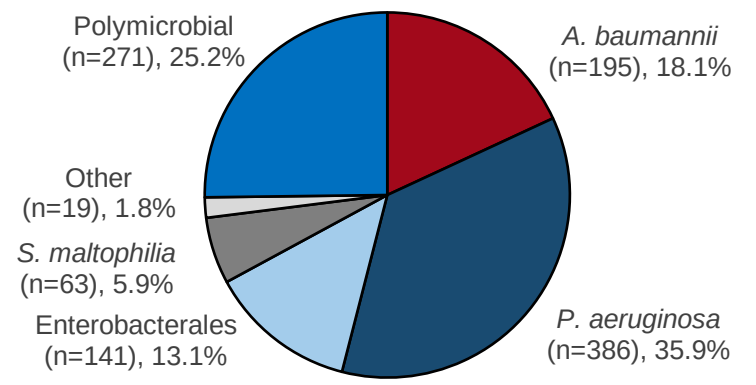
Polymicrobial (n=271), 25.2%
A. baumannii (n=195), 18.1%
Other (n=19), 1.8%
S. maltophilia (n=63), 5.9%
Enterobacterales (n=141), 13.1%
P. aeruginosa (n=386), 35.9%

*Other: bone and joint, skin and skin structure, intra-abdominal, and other sites.

Disease severity

56.6% were treated with cefiderocol in the ICU

11.0% had secondary bacteremia

38.5% required mechanical ventilation

25.3% received vasopressor support

Carbapenem susceptibility status

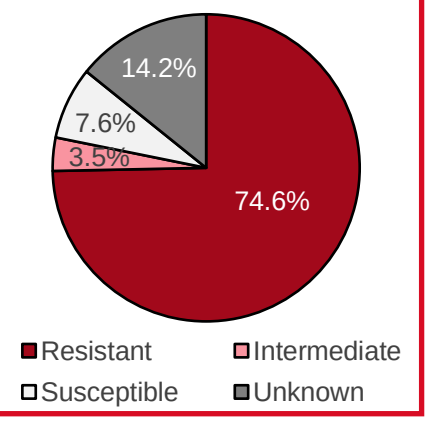
14.2%
7.6%
3.5%
74.6%
Resistant
Intermediate
Susceptible
Unknown

▼ 25 patients had a total of 29 ADRs, of which 3 were serious ADRs (i.e., interstitial nephritis and acute kidney injury; elevation of liver enzyme; hepatic cytolysis). Cefiderocol was discontinued for 16 events (in 13 patients).

Reason for cefiderocol use*

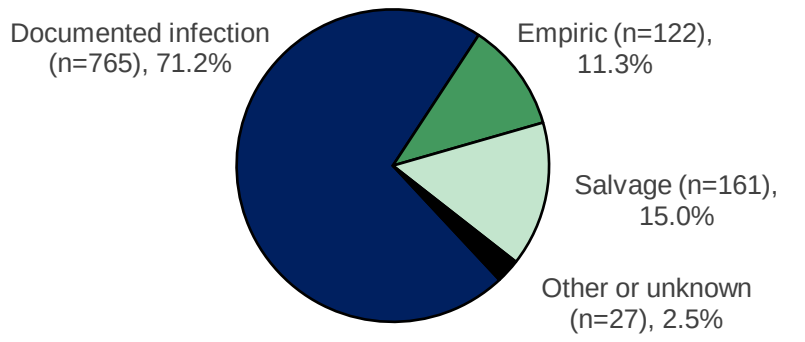
Documented infection (n=765), 71.2%
Empiric (n=122), 11.3%
Salvage (n=161), 15.0%
Other or unknown (n=27), 2.5%

Prior colonization/Prior infection status

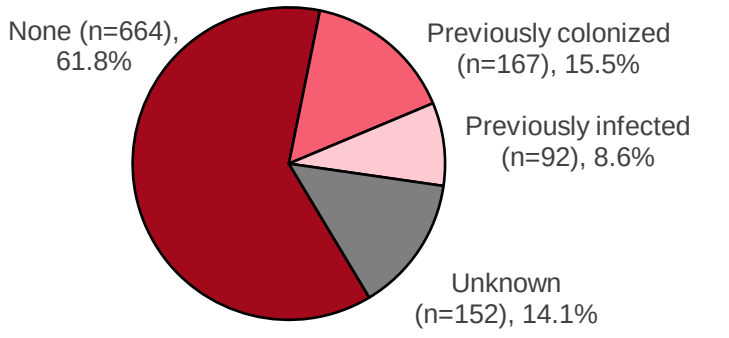
None (n=664), 61.8%
Previously colonized (n=167), 15.5%
Previously infected (n=92), 8.6%
Unknown (n=152), 14.1%

*Reported by the investigator. Documented infection: the pathogen and susceptibilities were known, and cefiderocol use was not due to failure of a prior antibiotic regimen. Empiric use: cefiderocol use started before the pathogen and/or susceptibilities were available to help guide the choice. Salvage therapy: cefiderocol was used after a prior antibiotic regimen had failed; patients usually received at least 3 days of prior antibiotic therapy to see if there was an adequate response before the next course of antibiotic would be considered salvage. Other, including unknown: any other reason provided by the physician.

Clinical cure and clinical response at EOT by infection site

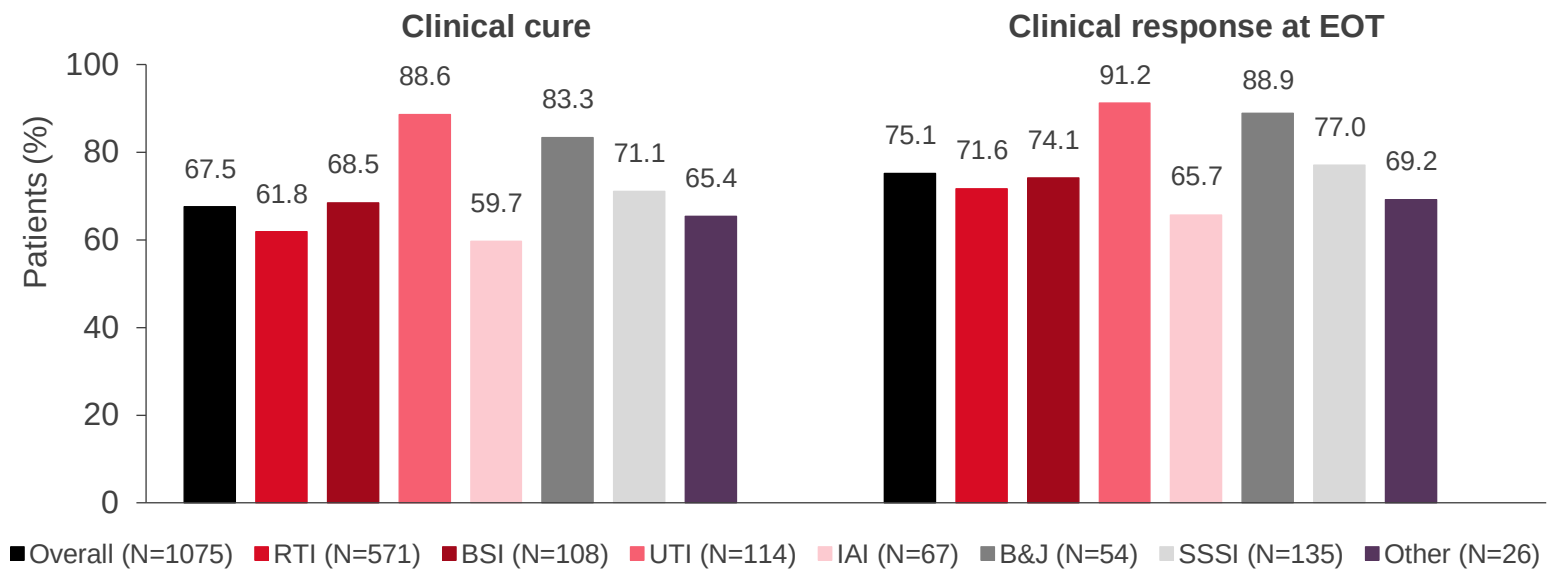
Clinical cure
Clinical response at EOT
Patients (%)
100
80
60
40
20
0
67.5
61.8
68.5
88.6
59.7
83.3
71.1
65.4
75.1
71.6
74.1
91.2
65.7
88.9
77.0
69.2
Overall (N=1075)
RTI (N=571)
BSI (N=108)
UTI (N=114)
IAI (N=67)
B&J (N=54)
SSSI (N=135)
Other (N=26)

Clinical cure and 30-day ACM by pathogen

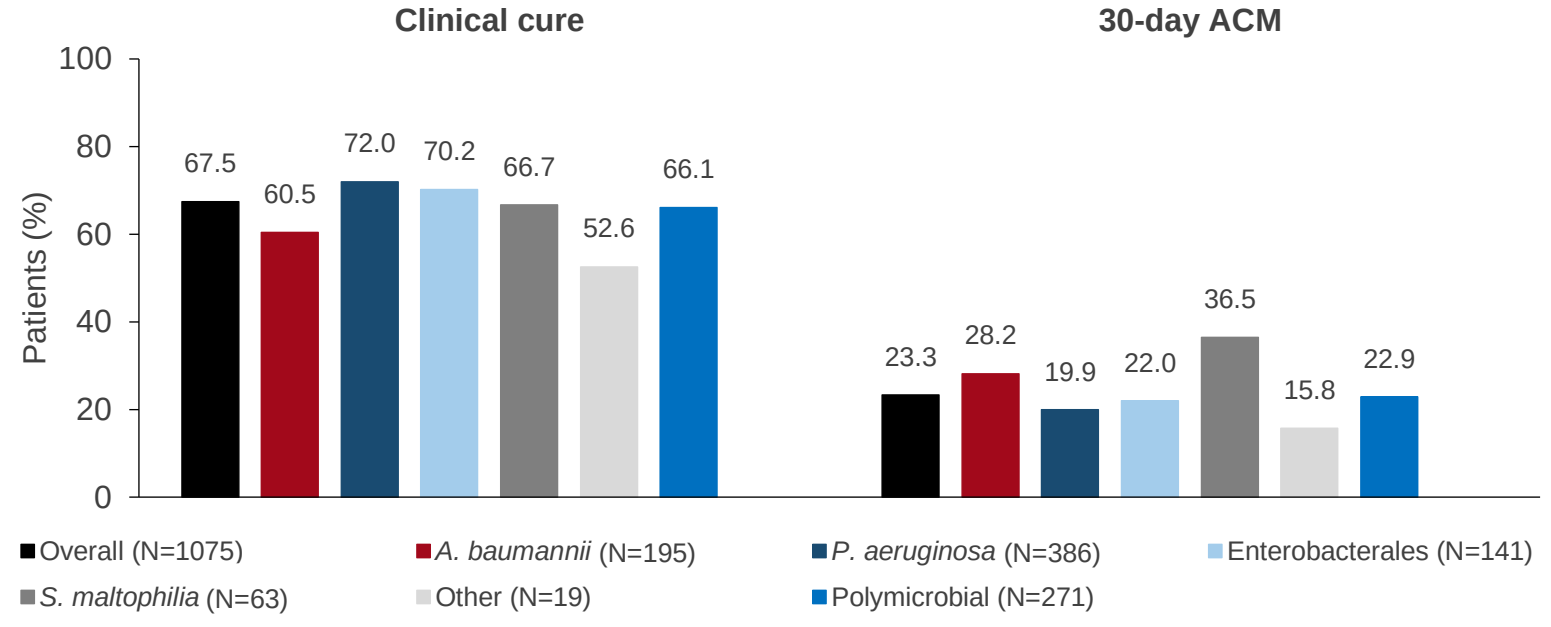
Clinical cure
30-day ACM
Patients (%)
100
80
60
40
20
0
67.5
60.5
72.0
70.2
66.7
52.6
66.1
23.3
28.2
19.9
22.0
36.5
15.8
22.9
Overall (N=1075)
A. baumannii (N=195)
P. aeruginosa (N=386)
Enterobacterales (N=141)
S. maltophilia (N=63)
Other (N=19)
Polymicrobial (N=271)

Abbreviations: ACM, all-cause mortality; B&J, bone and joint infection; BSI, bloodstream infection; CCI, Charlson Comorbidity Index; COVID-19, coronavirus disease-2019; EOT, end of treatment; IAI, intra-abdominal infection; ICU, intensive care unit; IQR, interquartile range; RTI, respiratory tract infection; SSSI, skin and skin structure infection; UTI, urinary tract infection.

Reference. 1. EUPAS40551. Retrospective chart review study of cefiderocol real world outcomes and safety in the treatment of patients with Gram-negative bacterial infections (GNBI) in the US and Europe (PROVE). https://catalogues.ema.europa.eu/node/2635/administrative-details.

This study was funded by Shionogi & Co., Ltd., Osaka, Japan. CS, STN, FK, SV, CL, SM, BC are or were employees of Shionogi during the conduct of the study.