Poster 3734

34th ECCMID 2024
Barcelona, Spain
27–30 April, 2024

Contact: Jessica Sarda
Email: jessica.sarda@shionogi.eu

Effectiveness of cefiderocol in immunosuppressed patients with serious Gram-negative bacterial infections in the PERSEUS study in Spain

Maria Rosa Oltra,[1] **Jessica Sarda**,[2] A. Javier Gonzalez,[2] Stefano Verardi,[3] Belen Loeches[4]

1. Infectious Disease Unit, Valencia University Clinical Hospital, València, Spain; 2. Shionogi S.L.U., Madrid, Spain; 3. Shionogi B.V., London, UK; 4. Clinical Unit of Infectious Diseases, La Paz University Hospital, Madrid, Spain

SHIONOGI

Revised abstract

Background: Cefiderocol has demonstrated potent *in vitro* activity against carbapenem-resistant and multidrug-resistant Gram-negative bacteria, including Enterobacterales, *Pseudomonas aeruginosa*, *Stenotrophomonas maltophilia* and other non-fermenting species. Cefiderocol was accessible in the Shionogi early access programme (EAP) for the treatment of patients with serious infections with no alternative treatment options in Spain (2018–2022). In this analysis, the real-world effectiveness of cefiderocol treatment in immunosuppressed patients was evaluated.

Methods: PERSEUS was a retrospective, multicentre, observational, medical chart review study enrolling hospitalised patients in the EAP with confirmed Gram-negative bacterial infections, who received first-time cefiderocol treatment for ≥72 hours. Patients with *Acinetobacter baumannii* infections were not enrolled in this study by design. Data included patient baseline characteristics, clinical success, clinical cure, all-cause mortality rates at Day 28 and adverse drug reactions. Only descriptive statistics were used.

Results: Of 261 eligible patients in the PERSEUS study, 79 (30.3%) had immunosuppression. Immunosuppressed patients had a median age of 59 years (range: 45–66) and 70.9% (n=56) were male. Comorbid conditions were present in 89.9% of patients, most commonly solid/haematological cancer (44.3%), chronic renal disease (19.0%) and diabetes mellitus (16.5%). At baseline, 49.4% of patients were in the intensive care unit, 24.1% had septic shock, 25.3% received renal replacement therapy and 7.6% had secondary bacteraemia. Immunosuppressed patients most frequently had respiratory tract infection (35.4%), urinary tract infection (20.3%) and intra-abdominal infection (19.0%). The most frequent pathogens were *P. aeruginosa* (51.9%), *S. maltophilia* (13.9%), *Pseudomonas* spp. (11.4%) and other non-fermenters (11.4%). Polymicrobial infections were present in 11.4% of patients. The median duration of treatment was 10.0 days (range: 6.0–14.0). In this subgroup of patients, the overall clinical success rate was 81.0% (64/79), clinical cure rate was 77.2% (61/79) and mortality rate at Day 28 was 22.8% (18/79). Two patients (2.5%) reported adverse drug reactions; both events were mild, and both patients recovered.

Conclusions: Cefiderocol was effective, with a high clinical cure rate and rare adverse drug reactions, in immunosuppressed patients with serious infections caused mainly by *P. aeruginosa* and other non-fermenters.

OBJECTIVES

In the PERSEUS study, patients were treated with cefiderocol for ≥72 hours for a confirmed Gram-negative bacterial infection and were mainly infected by *Pseudomonas aeruginosa* [1]. Of 261 eligible patients, 80.5% (210/261) achieved clinical cure and 21.5% (56/261) died by Day 28 [1]. The objective of this subgroup analysis of the PERSEUS study was to describe the baseline characteristics and the clinical outcomes in patients with immunosuppression at baseline, who were treated with cefiderocol for up to 28 days.

METHODS

Study design: a retrospective, multicentre, observational study in patients receiving cefiderocol for the first time in the early access programme in Spain.

Inclusion criteria: adult hospitalised patients treated with cefiderocol consecutively for ≥72 hours for a confirmed Gram-negative bacterial infection, with tested sensitivity to cefiderocol.

Exclusion criteria: confirmed *Acinetobacter* spp. at baseline; confirmed cefiderocol-resistant Gram-negative pathogen at baseline; incomplete medical records; enrolled into other clinical studies of investigational products.

Endpoints: baseline patient characteristics, Gram-negative bacterial pathogens, clinical success (composite of clinical cure and/or survival at Day 28), clinical cure (cessation of antibiotic treatment due to clinical resolution of signs and symptoms) at end of treatment and all-cause mortality at Day 28.

RESULTS

Patient attrition

Screened population (N=314)
↓
Eligible population (N=274)
↓
Primary analysis population (N=261)
↓
Immunosuppression* (N=79)

*Transplant recipient, immunosuppressive treatment (e.g. high-dose corticosteroids, calcineurin inhibitors, anti-CD20, IL-1 inhibitors, and IL-6 inhibitors).

Patient characteristics (N=79)

Sex, male	56 (70.9%)
Age, median (Q1–Q3), years	59 (45–66)
CCI score, median (Q1–Q3)	4 (2–5)
Symptomatic COVID-19	15 (19.0%)

Main comorbidities

Transplant recipient	53 (67.1%)
Tumor (solid/haematological)	35 (44.3%)
Chronic renal disease	15 (19.0%)
Diabetes	13 (16.5%)

49.4% (n=39) — **ICU at the time of cefiderocol**

41.8% (n=33) — **Mechanical ventilation**

25.3% (n=20) — **Renal replacement therapy**

24.1% (n=19) — **Septic shock**

RESULTS CONT'D

Site of infection

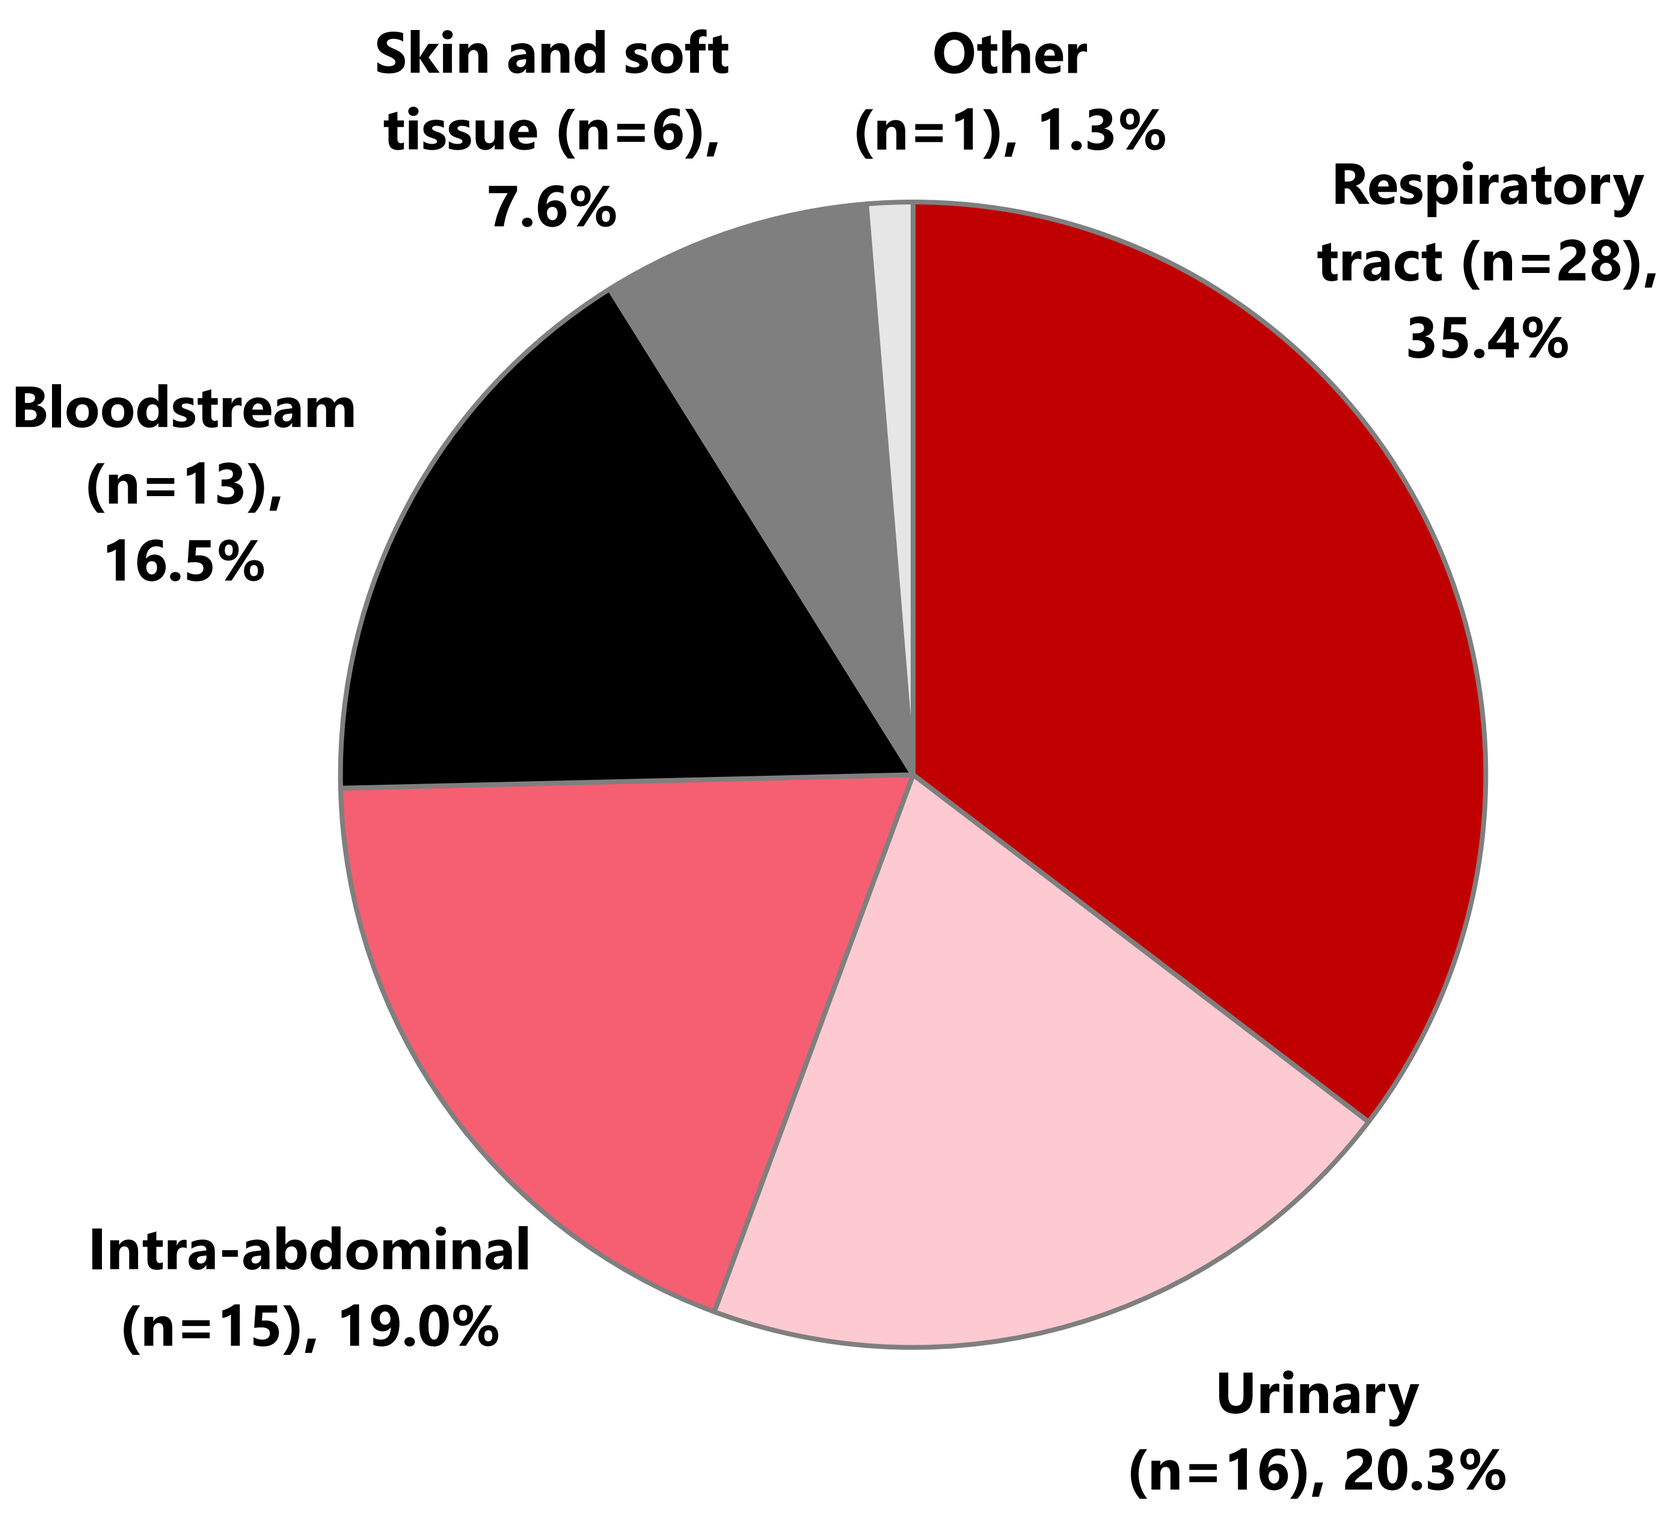
Skin and soft tissue (n=6), 7.6%
Other (n=1), 1.3%
Respiratory tract (n=28), 35.4%
Bloodstream (n=13), 16.5%
Intra-abdominal (n=15), 19.0%
Urinary (n=16), 20.3%

Distribution of pathogens

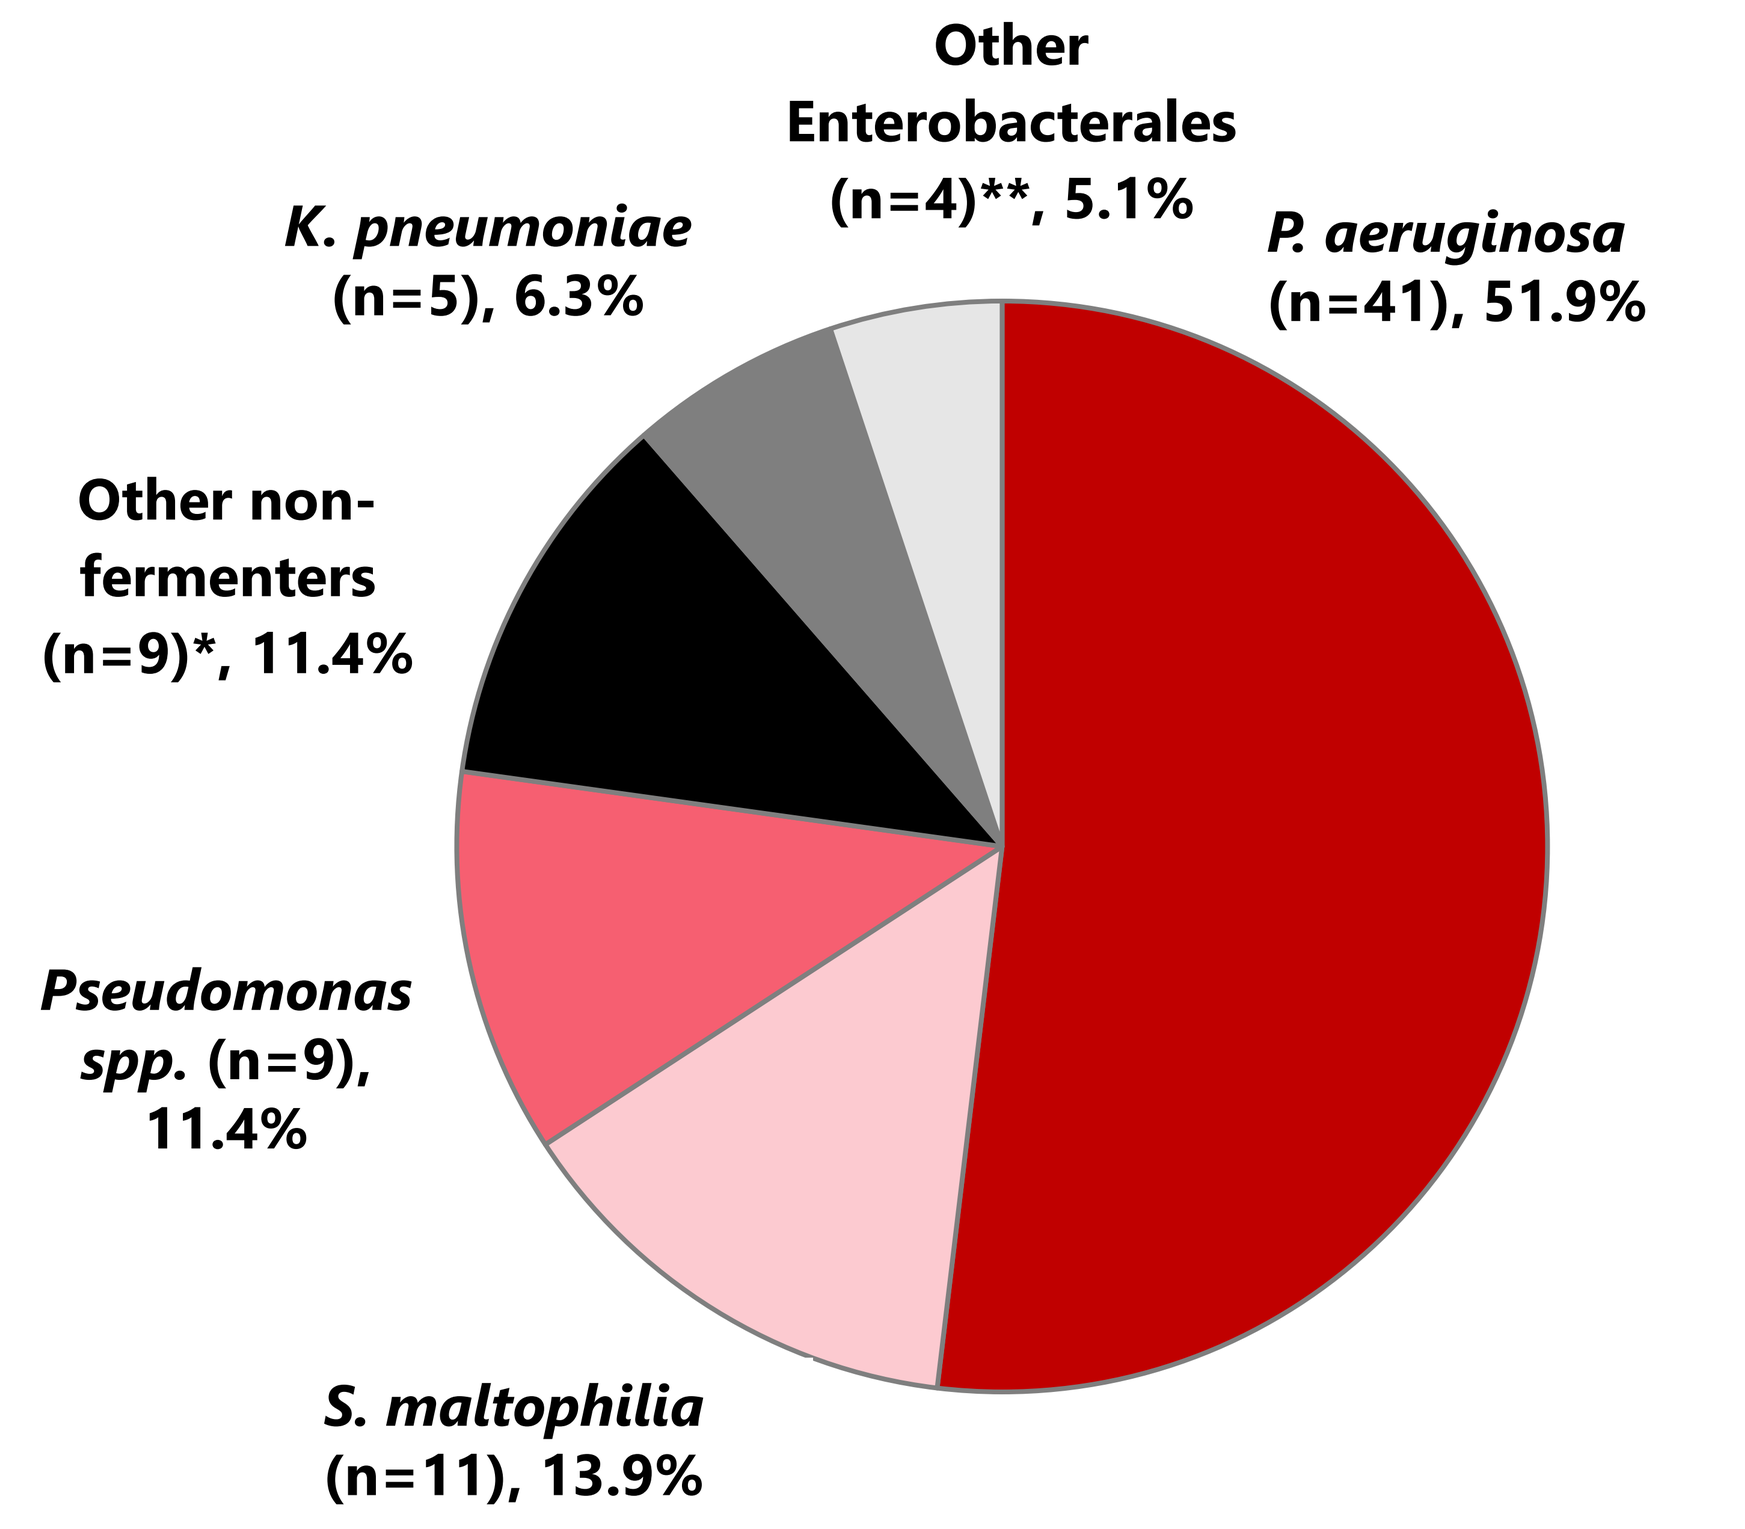
Other Enterobacterales (n=4)**, 5.1%
K. pneumoniae (n=5), 6.3%
P. aeruginosa (n=41), 51.9%
Other non-fermenters (n=9)*, 11.4%
Pseudomonas spp. (n=9), 11.4%
S. maltophilia (n=11), 13.9%

**Burkholderia cepacia* complex (5), *Achromobacter* spp. (3), *Ralstonia mannitolilytica* (1);
***Klebsiella oxytoca* (2), *Citrobacter freundii* (1), *Enterobacter cloacae* (1);

Baseline infection characteristics and rationale for cefiderocol administration in patients with immunosuppression[a] (N=79)	
Secondary bacteraemia, n (%)	6 (7.6)
Polymicrobial infection, n (%)	9 (11.4)
Previous colonisation with the same pathogen, n (%)	41 (51.9)
Previous treatment with antibiotics, n (%)	66 (83.5)
Rationale for administering cefiderocol[b]	
Resistance to all tested antibiotics	53 (67.1)
Treatment failure with prior antibiotics	35 (44.3)
Adverse events to other susceptible antibiotics	8 (10.1)
Other	5 (6.3)
Cefiderocol treatment duration, median (range), days	10.0 (6.0–14.0)
Cefiderocol combination therapy, n (%)	35 (44.3)
Adverse drug reactions, n (%)	2 (2.5)

[a]Transplant recipient, immunosuppressive treatment (e.g. high-dose corticosteroids, calcineurin inhibitors, anti-CD20, IL-1 inhibitors, and IL-6 inhibitors); [b]Not mutually exclusive.

Clinical outcomes

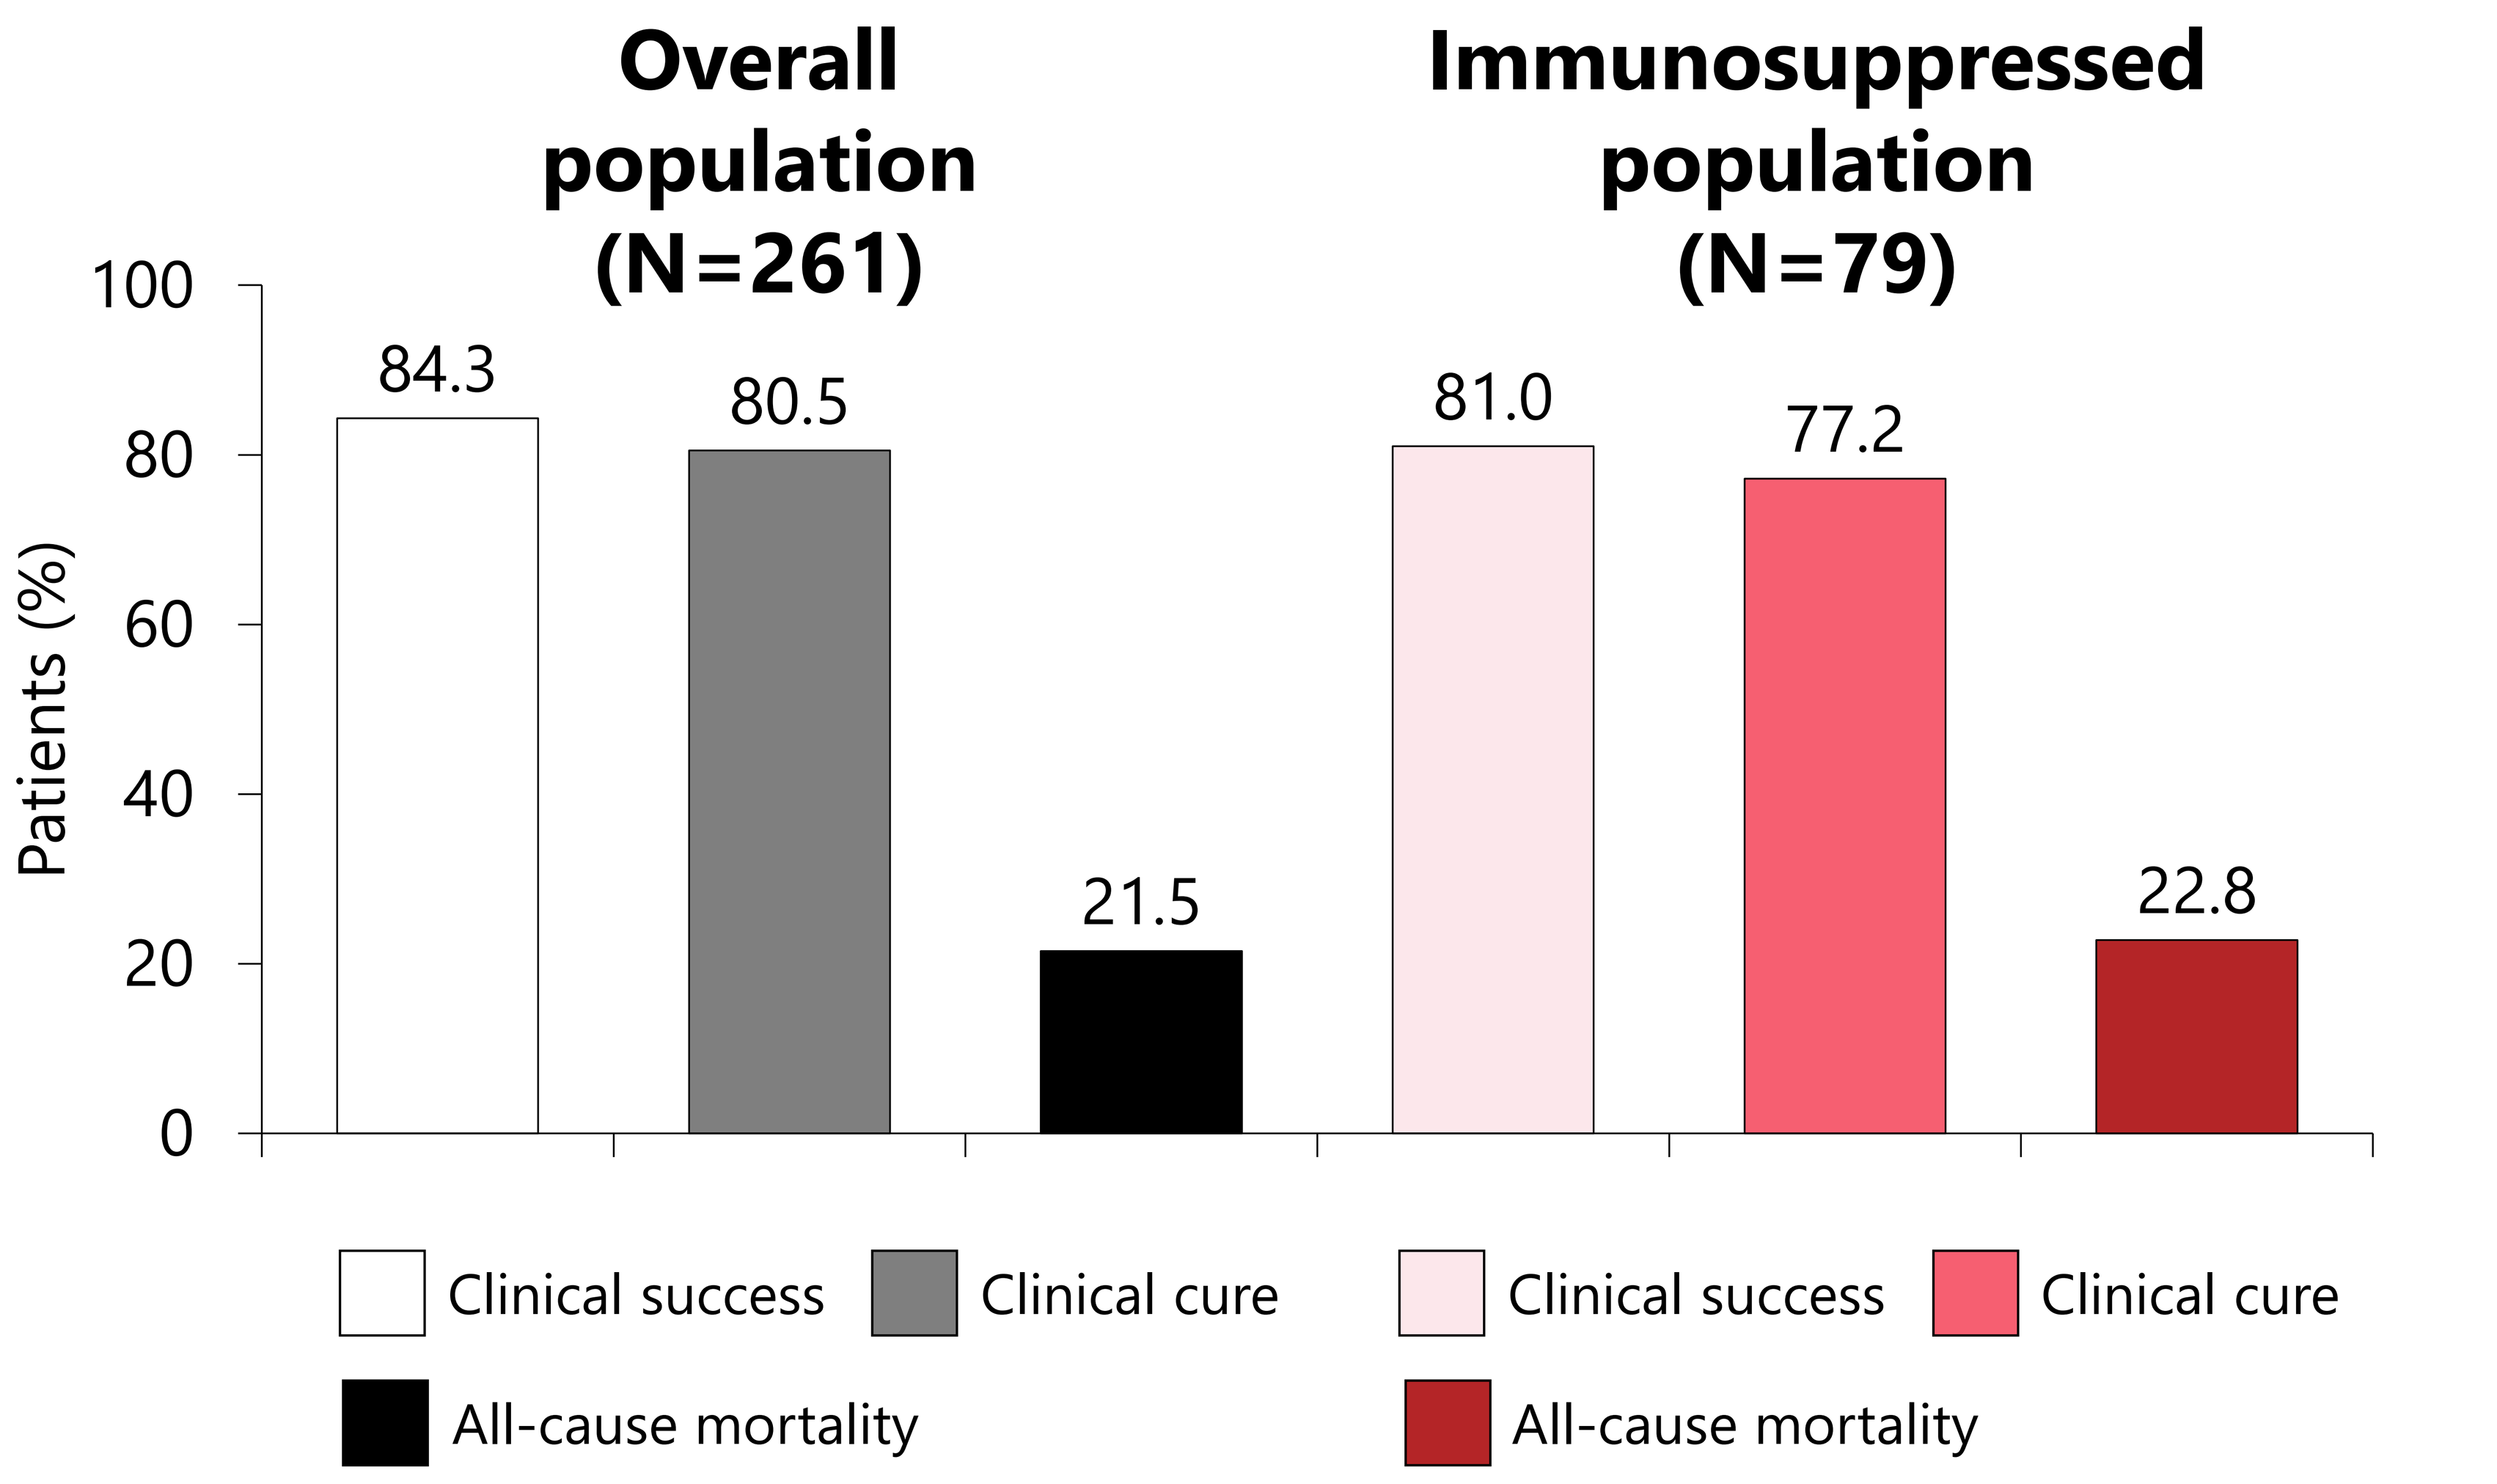
Overall population (N=261)
Immunosuppressed population (N=79)
Patients (%)
100
80
60
40
20
0
84.3
80.5
21.5
81.0
77.2
22.8
Clinical success
Clinical cure
All-cause mortality
Clinical success
Clinical cure
All-cause mortality

CONCLUSIONS

Cefiderocol was effective with a high clinical cure rate in immunosuppressed patients with serious infections caused mainly by *P. aeruginosa* and other non-fermenters. Adverse drug reactions were rare.

Reference

1. Ramirez P, et al. Real-world effectiveness and safety of cefiderocol in patients with Gram-negative bacterial infections in the early access programme in Spain: results of the PERSEUS study. Presented at 34th ECCMID, Barcelona, Spain; 27–30 April 2024. Poster 2523.

Acknowledgements

The PERSEUS study was funded by Shionogi. Sarda J, Verardi S, Gonzalez AJ are employees of Shionogi. Editorial support was provided by Highfield, Oxford, UK and this support was funded by Shionogi.

Abbreviations

CCI, Charlson Comorbidity Index; COVID-19, coronavirus disease 2019; ICU, intensive care unit; IL, interleukin; Q, quartile.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ECCMID 2024 and the authors of the poster.