Poster 2524

34th ECCMID 2024
Barcelona, Spain
27–30 April, 2024

Contact: Jessica Sarda
Email: jessica.sarda@shionogi.eu

Real-world effectiveness and safety of long-term (>28 days) cefiderocol treatment in patients with Gram-negative bacterial infections in the PERSEUS study in Spain

Maria Jose Perez-Pedrero,[1] Jorge Calderón Parra,[2] **Jessica Sarda**,[3] A. Javier Gonzalez,[3] Stefano Verardi,[4]

1. Department of Critical Care Medicine, Complejo Hospitalario de Toledo, Toledo, Spain; 2. Internal Medicine Department, Hospital Universitario Puerta de Hierro-Majadahonda, Madrid, Spain; 3. Shionogi S.L.U., Madrid, Spain; 4. Shionogi B.V., London, UK

SHIONOGI

Abstract

Background: Cefiderocol was utilised through the Shionogi early access programme (EAP) for the treatment of patients with serious Gram-negative bacterial infections (GNBIs), who had no alternative treatment options, in Spain between 2018 and 2022. In the PERSEUS study, the effectiveness of cefiderocol in patients with GNBIs, excluding *Acinetobacter baumannii*, was evaluated in real-world settings in Spain. The current subgroup analysis assessed the long-term effectiveness and safety of cefiderocol administered for longer than 28 days.

Methods: PERSEUS was an observational, multicentre, retrospective, medical chart review study of hospitalised patients with confirmed GNBIs who participated in the Shionogi EAP and were treated with cefiderocol for the first time for ≥72 hours. Patients with documented *A. baumannii* infections were excluded by design. In this analysis, patients with a treatment duration of >28 days were included. Patient demographics, baseline clinical characteristics, clinical cure, all-cause mortality at Day 90 and safety were evaluated. Only descriptive statistics were used.

Results: A total of 13 patients received cefiderocol for >28 days. All patients were male, and the median age was 54 years (range: 40–65). In this subgroup, patients most frequently had intra-abdominal infection (30.8%, 4/13), osteoarticular infection (30.8%, 4/13) and respiratory tract infection (23.1%, 3/13). Bloodstream infection and vascular prosthesis infection were each found in one patient (7.7%). The causative pathogens were *Pseudomonas aeruginosa* (84.6%, 11/13), *Stenotrophomonas maltophilia* (7.7%, 1/13) and *Elizabethkingia miricola* (7.7%, 1/13). At baseline, the proportion of patients in the intensive care unit was 46.2% (6/13), and one patient (7.7%) had septic shock, five (38.5%) received renal replacement therapy and three (23.1%) had secondary bacteraemia. The median treatment duration was 40.0 days (range: 34–46). Overall, the clinical cure rate was 84.6% (11/13) and mortality rate at Day 90 was 23.1% (3/13). Cefiderocol was well tolerated without any adverse drug reaction in these patients.

Conclusions: Cefiderocol treatment administered for >28 days resulted in a high clinical cure rate and was well tolerated in patients with serious infections and no alternative treatment options.

OBJECTIVES

In the PERSEUS retrospective study, of the overall eligible population, who were treated with cefiderocol for ≥72 hours for a confirmed Gram-negative bacterial infection and primarily infected by *Pseudomonas aeruginosa*, 80.5% (210/261) achieved clinical cure and 21.5% (56/261) died by Day 28 [1]. The objective of this subgroup analysis was to describe the baseline characteristics and the clinical outcomes following cefiderocol treatment administered for >28 days in patients with Gram-negative bacterial infections enrolled into the PERSEUS study in Spain.

METHODS

Study design: a retrospective, observational study in patients receiving cefiderocol for the first time in the early access programme (EAP) in Spain.

Inclusion criteria: adult hospitalised patients treated with cefiderocol consecutively for ≥72 hours for a confirmed Gram-negative bacterial infection.

Exclusion criteria: confirmed *Acinetobacter* spp. at baseline; confirmed cefiderocol-resistant Gram-negative pathogen at baseline; incomplete medical records; enrolled into other clinical studies of investigational products.

Endpoints: baseline patient characteristics, Gram-negative species, clinical cure (cessation of antibiotic treatment due to clinical resolution of signs and symptoms) at end of treatment and all-cause mortality at Day 90.

RESULTS

Patient attrition

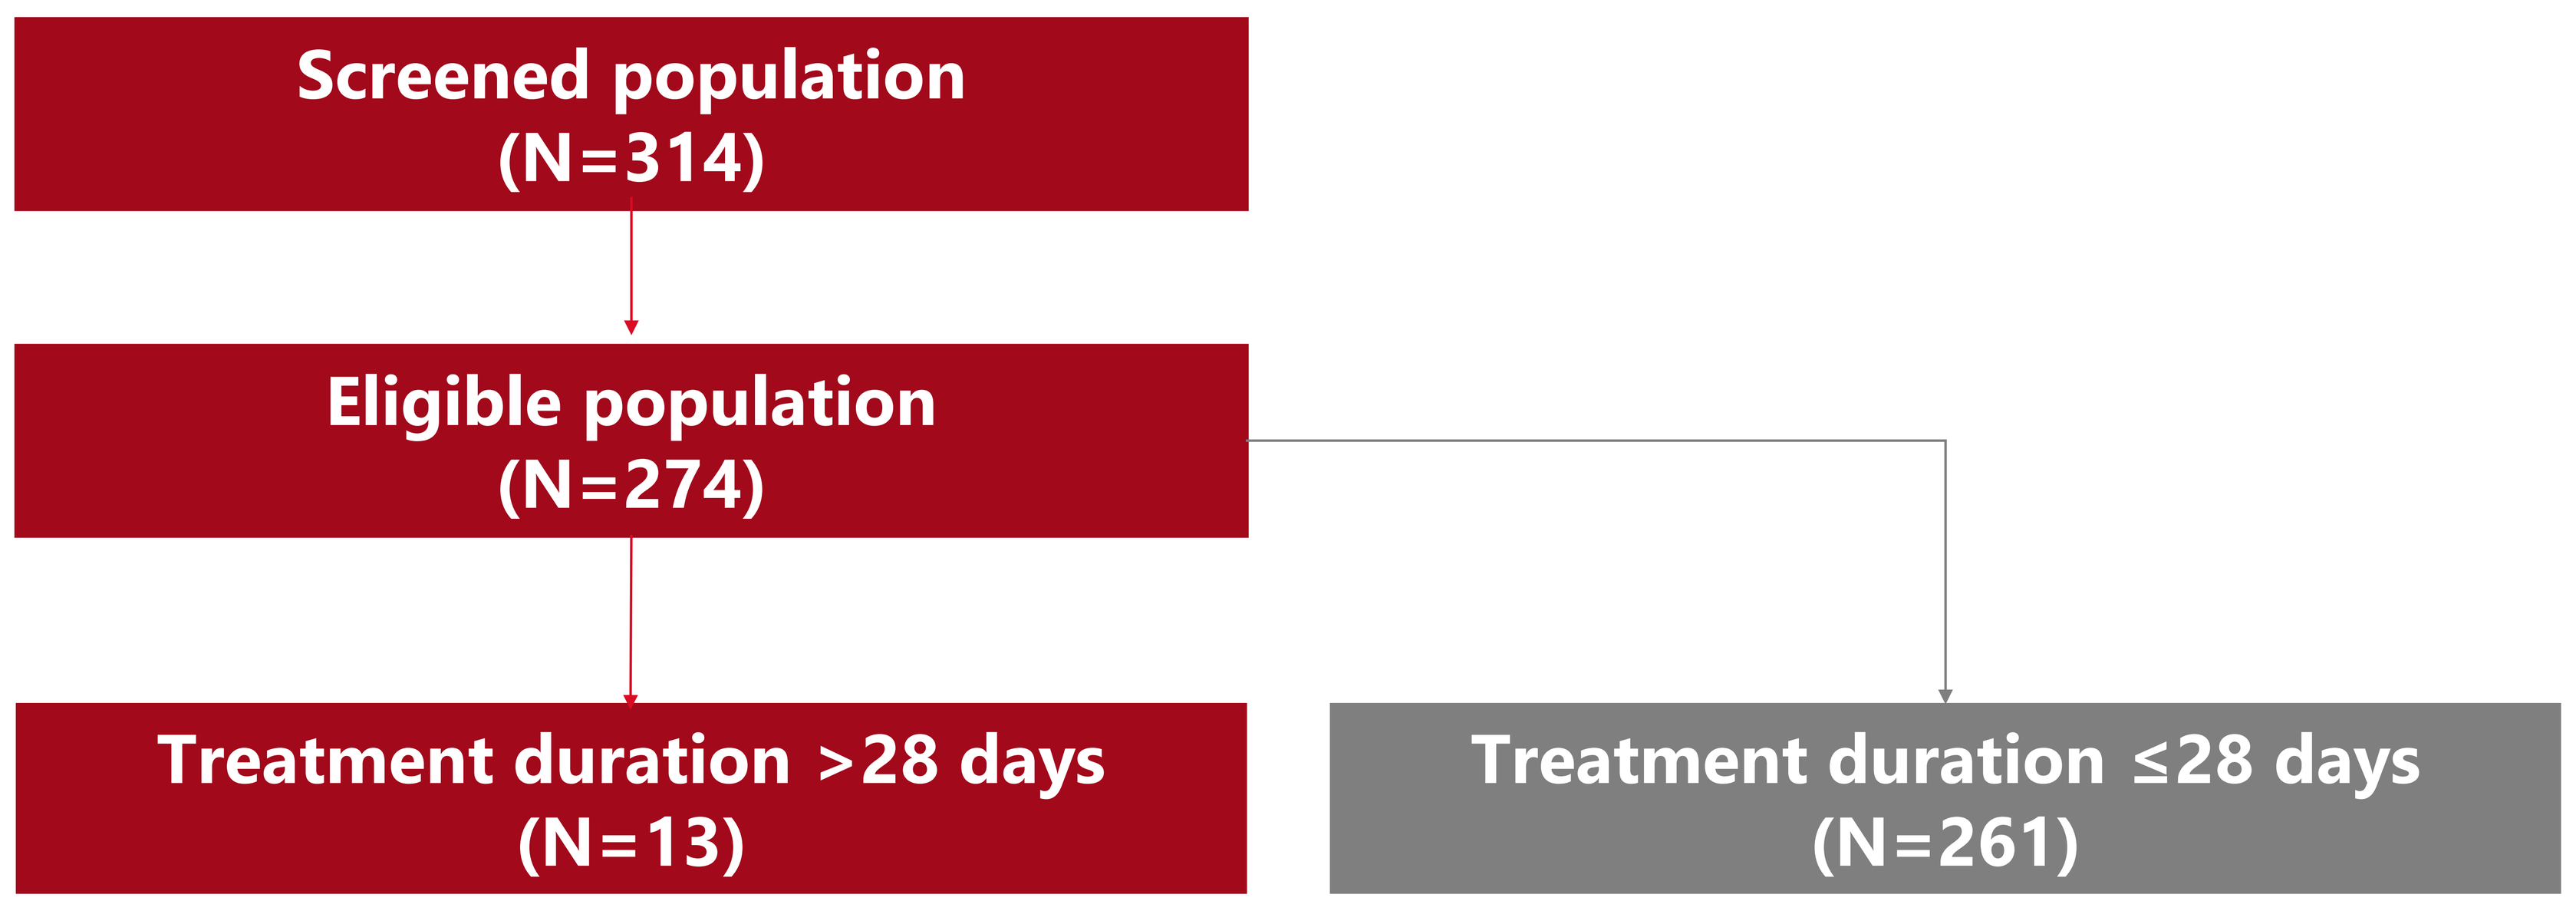
Screened population
(N=314)
Eligible population
(N=274)
Treatment duration >28 days
(N=13)
Treatment duration ≤28 days
(N=261)

Patient characteristics (N=13)		Main comorbidities	
Sex, male	13 (100%)	Tumour (solid/haematological)	5 (38.5%)
Age, median (Q1–Q3), years	54 (40–65)	Chronic renal disease	4 (30.8%)
CCI score, median (Q1–Q3)	2 (2–7)	Peripheral vascular disease	3 (23.1%)
APACHE II score, median (Q1–Q3)	16 (14–20)	Myocardial infarction	3 (23.1%)

46.2% (n=6)	38.5% (n=5)	30.8% (n=4)	7.7% (n=1)	7.7% (n=1)
ICU at the time of cefiderocol	Renal replacement therapy	Mechanical ventilation	Immuno-suppression	Septic shock

Site of infection

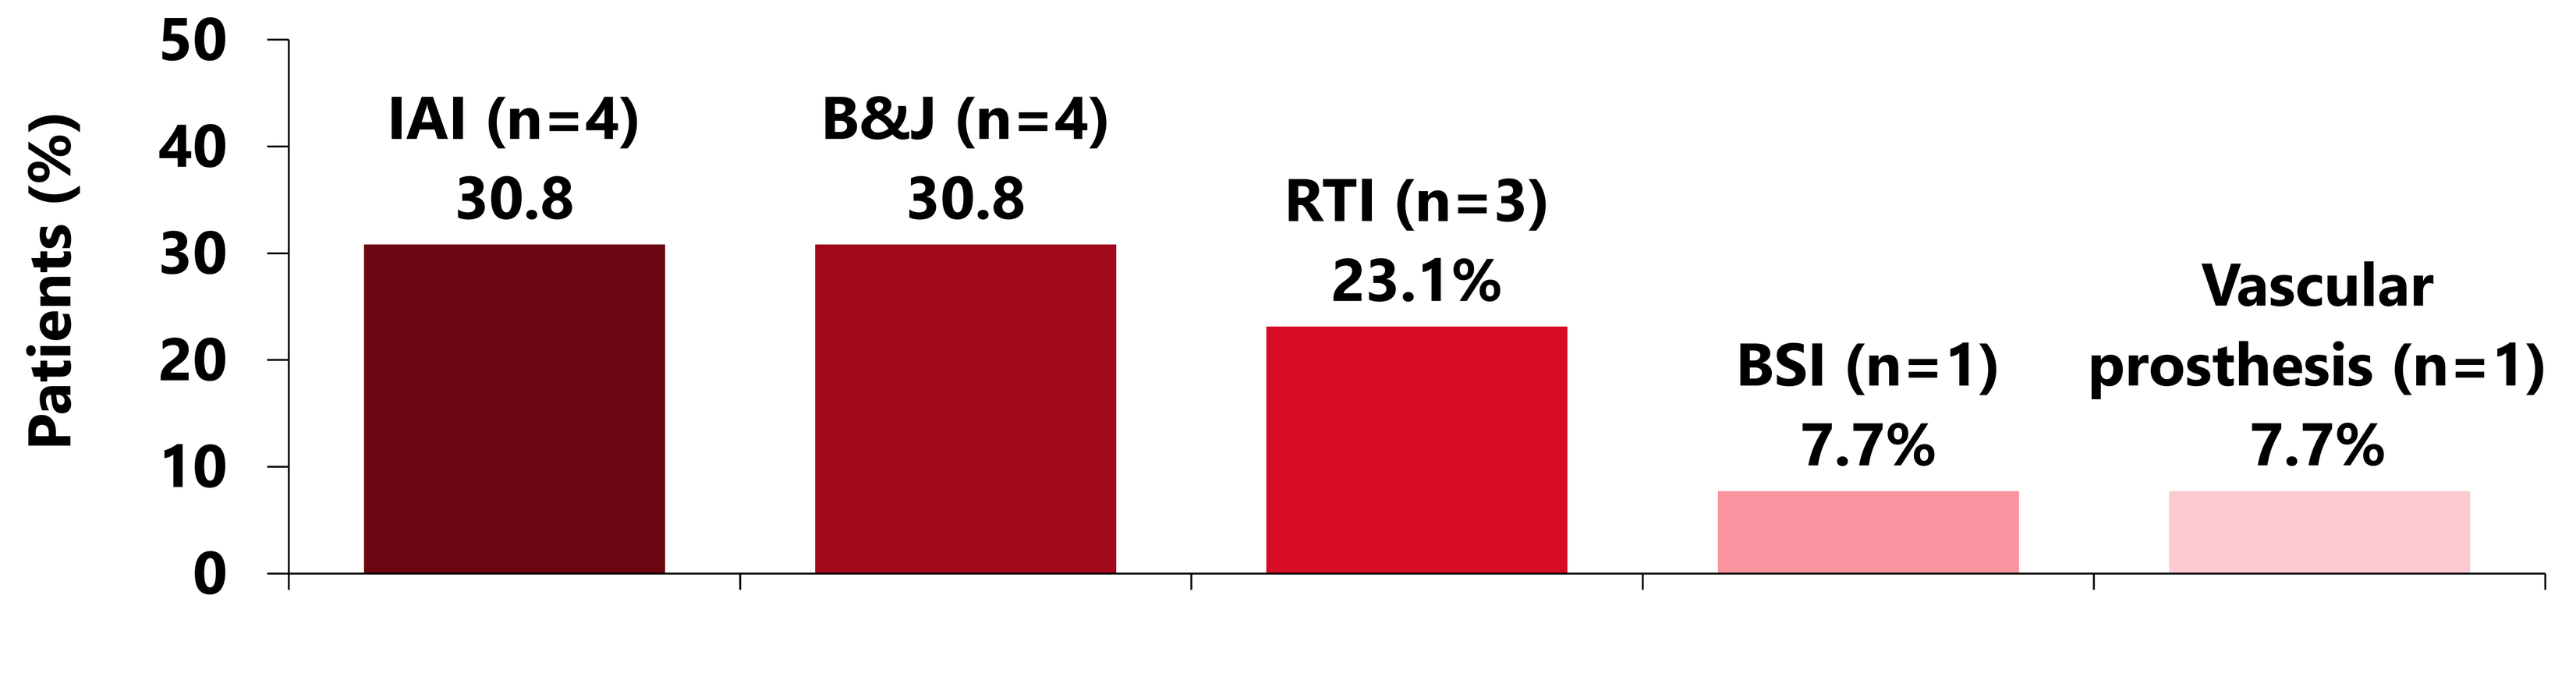
Patients (%)
50
40
30
20
10
0
IAI (n=4)
30.8
B&J (n=4)
30.8
RTI (n=3)
23.1%
BSI (n=1)
7.7%
Vascular prosthesis (n=1)
7.7%

RESULTS CONT'D

Baseline Gram-negative pathogens and rationale for cefiderocol administration in patients with treatment duration of >28 days (N=13)	
Gram-negative pathogen, n (%)	
Pseudomonas aeruginosa	11 (84.6)
Stenotrophomonas maltophilia	1 (7.7)
Elizabethkingia miricola	1 (7.7)
Secondary bacteraemia, n (%)	3 (23.1)
Polymicrobial infection, n (%)	2 (15.4)
Previously colonised with the same pathogen, n (%)	8 (61.5)
Other pathogens isolated, n (%)	6 (46.2)
Gram-positive pathogen, n (%)	5 (83.3)
Fungi, n (%)	3 (50.0)
Anaerobes, n (%)	1 (16.7)
Previous treatment with antibiotics, n (%)	11 (84.6)
Rationale for administering cefiderocol[a]	
Resistance to all tested antibiotics	7 (53.8)
Treatment failure with prior antibiotics	5 (38.5)
Adverse events to other susceptible antibiotics	2 (15.4)
Other	3 (23.1)
Cefiderocol treatment duration, median (range), days	40 (34–46)
Cefiderocol combination therapy, n (%)	7 (53.8)
Adverse drug reactions, n (%)	0 (0)

[a]Not mutually exclusive.

Clinical outcomes

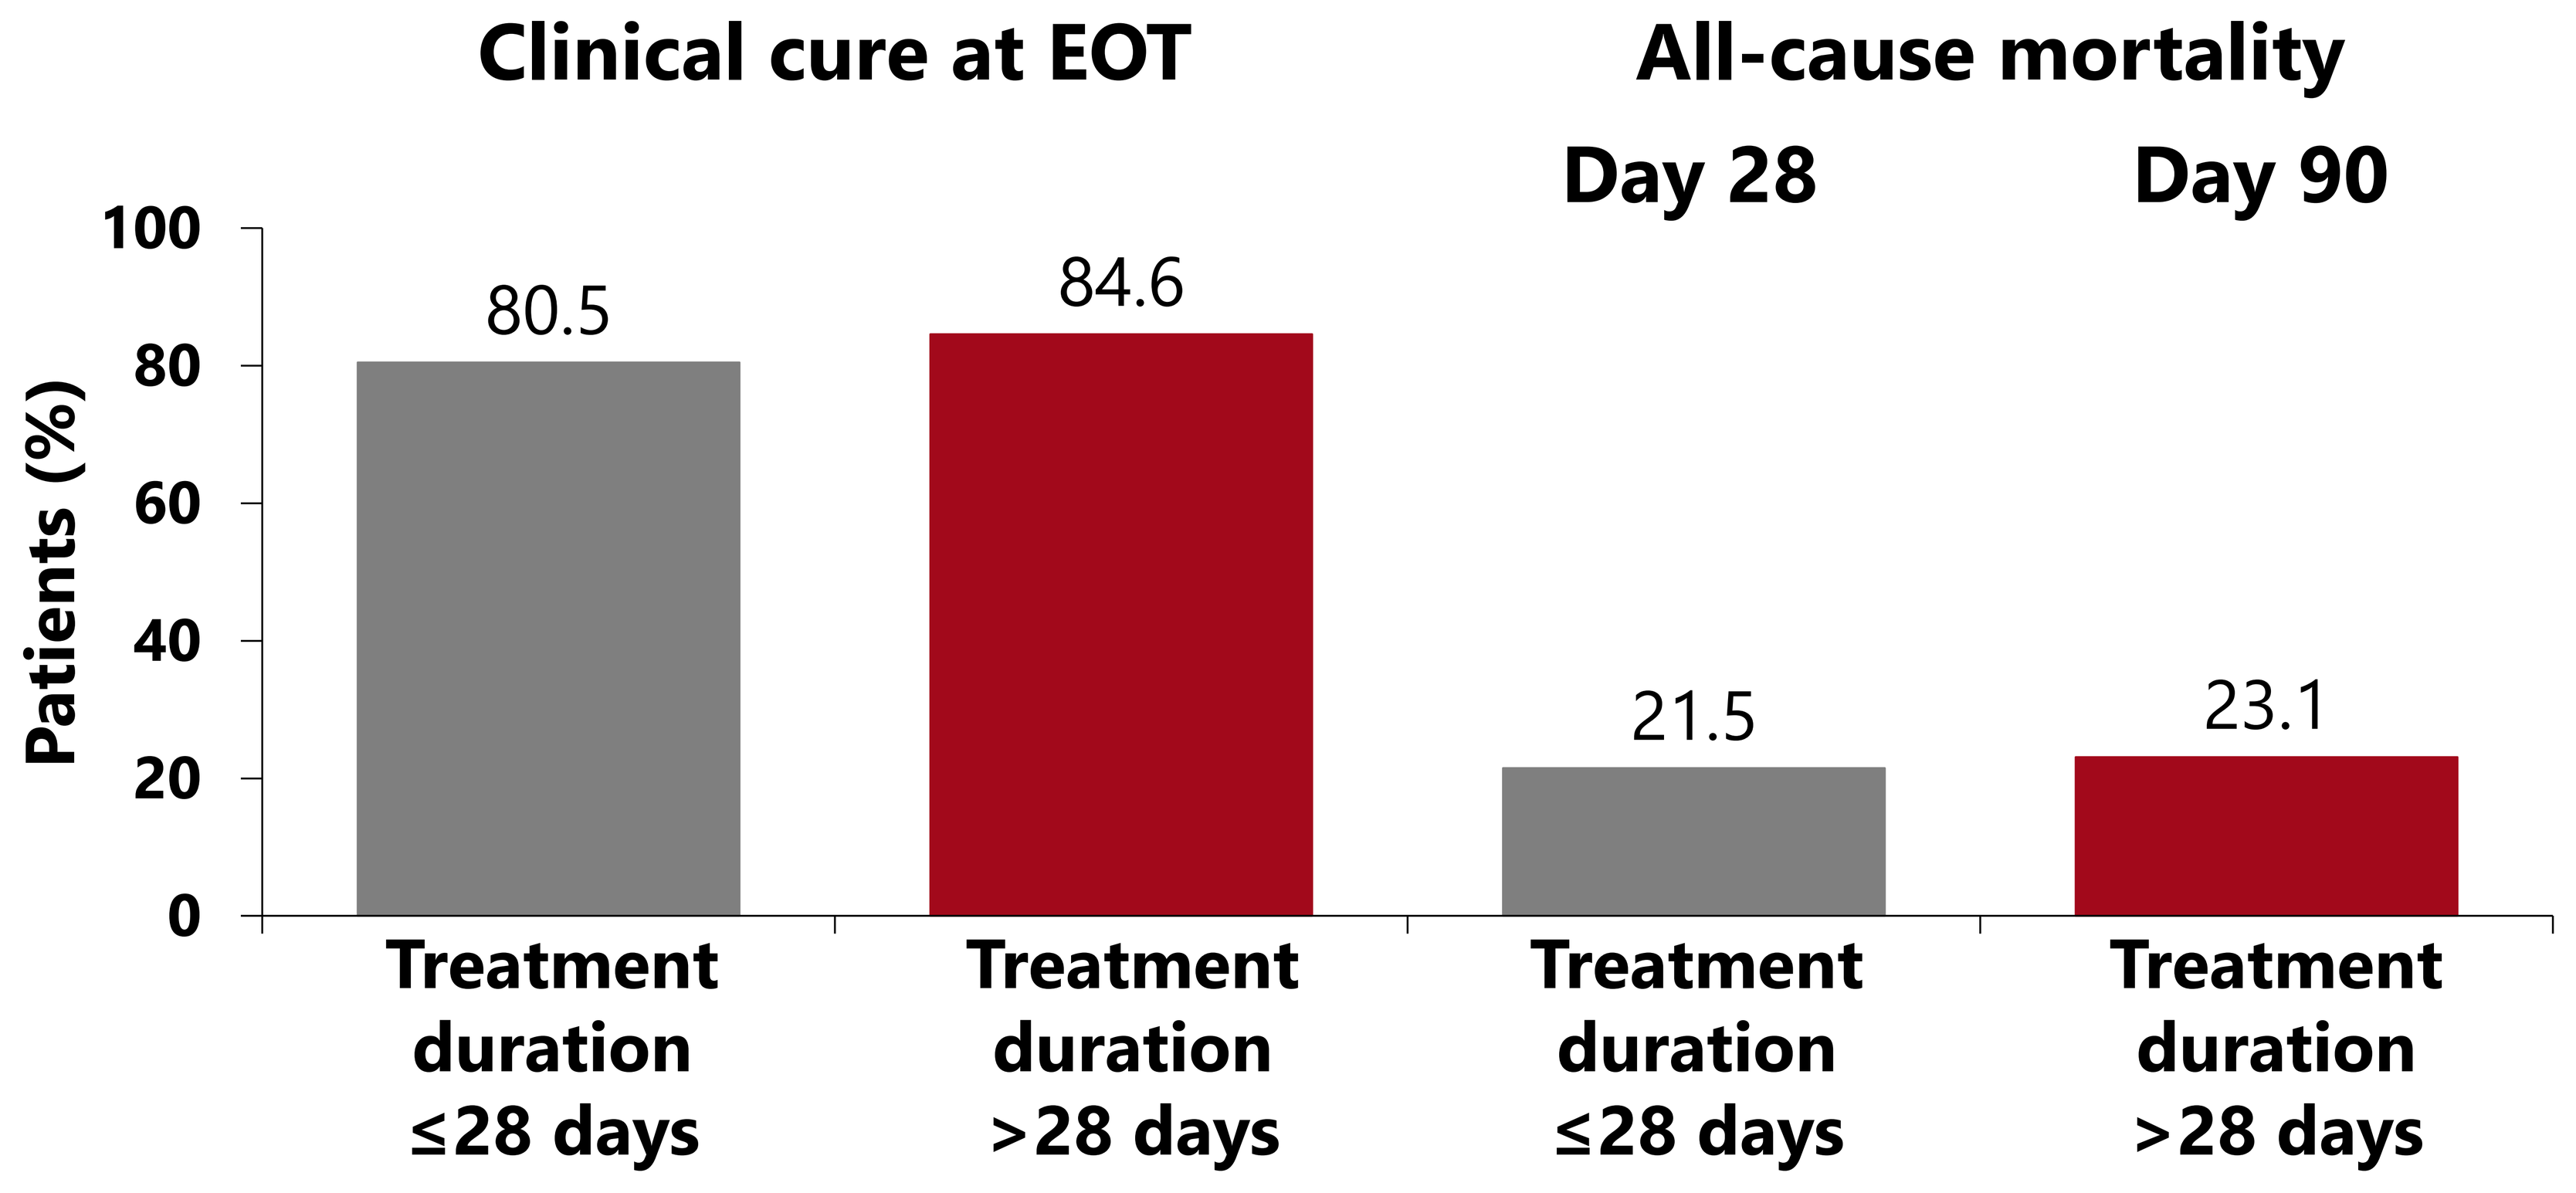
Clinical cure at EOT
All-cause mortality
Day 28
Day 90
Patients (%)
100
80
60
40
20
0
80.5
84.6
21.5
23.1
Treatment duration ≤28 days
Treatment duration >28 days
Treatment duration ≤28 days
Treatment duration >28 days

CONCLUSIONS

- Where cefiderocol treatment was administered for long term (i.e., >28 days), it was driven by deep seated infections, such as IAI and B&J infections. In these patients, clinical cure rate was high and mortality rate at day 90 was <25% in this subgroup of the PERSEUS study.
- Cefiderocol was well tolerated in patients with serious infections and limited treatment options that required prolonged treatment.

Reference

1. Ramirez P, et al. Real-world effectiveness and safety of cefiderocol in patients with Gram-negative bacterial infections in the early access programme in Spain: results of the PERSEUS study. Presented at 34th ECCMID, Barcelona, Spain; 27–30 April 2024. Poster 2523.

Acknowledgements

The PERSEUS study was funded by Shionogi. Sarda J, Verardi S, Gonzalez AJ are employees of Shionogi. Editorial support was provided by Highfield, Oxford, UK and this support was funded by Shionogi.

Abbreviations

APACHE, Acute Physiology and Chronic Health Evaluation; B&J, bone and joint infection; BSI, bloodstream infection; CCI, Charlson Comorbidity Index; EOT, end of treatment; IAI, intra-abdominal infection; ICU, intensive care unit; LOS, length of stay; Q, quartile; RTI, respiratory tract infection.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ECCMID 2024 and the authors of the poster.