Poster 2523

34th ECCMID 2024
Barcelona, Spain
27–30 April, 2024

Contact: Jessica Sarda
Email: jessica.sarda@shionogi.eu

Real-world effectiveness and safety of cefiderocol in patients with Gram-negative bacterial infections in the early access programme in Spain: results of the PERSEUS study

Paula Ramirez,[1] Esperanza Merino,[2] **Jessica Sarda**,[3] A. Javier Gonzalez,[3] Stefano Verardi,[4] Jesus Fortun[5]

1. Servicio de Medicina Intensiva, Hospital Universitario y Politécnico la Fe, Valencia, Spain; 2. Infectious Diseases Unit, Alicante General University Hospital-Alicante Institute of Sanitary and Biomedical Research (ISABIAL), Alicante, Spain; 3. Shionogi S.L.U., Madrid, Spain; 4. Shionogi B.V., London, UK; 5. Hospital Universitario Ramón y Cajal, CIBERINFEC, IRYCIS, Madrid, Spain

SHIONOGI

Revised abstract

Background: Cefiderocol was utilised for the treatment of life-threatening Gram-negative bacterial infections (GNBIs) through the Shionogi early access programme (EAP) in Spain. In the PERSEUS study, the effectiveness and safety of cefiderocol in patients with GNBIs were evaluated in real-world settings in Spain.

Methods: PERSEUS was a retrospective, multicentre, observational, medical chart review study (2018–2022). Hospitalised patients with confirmed GNBIs in the EAP were treated with cefiderocol for the first time for ≥72 hours. Patient characteristics, clinical success, clinical cure, all-cause mortality rates at Day 28 and safety were evaluated. The primary endpoint population included patients with a treatment duration of ≤28 days. Patients with *Acinetobacter baumannii* were not enrolled by design. Only descriptive statistics were used.

Results: Of 261 patients, 77.4% were male, and the median age was 61 years (range: 49–68). Patients most frequently had respiratory tract infection (RTI; 47.9%), intra-abdominal infection (14.6%) and urinary tract infection (UTI; 14.6%). The most frequent pathogens were *Pseudomonas aeruginosa* (66.7%), *Klebsiella pneumoniae* (10.0%) and *Stenotrophomonas maltophilia* (7.7%). The median treatment duration was 10.0 days (range: 7.0–14.0). At baseline, 63.2% of patients were in the intensive care unit (ICU) and 28.0% had septic shock. Overall, the clinical success rate was 84.3% (220/261), clinical cure rate was 80.5% (210/261) and 21.5% (56/261) of patients died by Day 28. Clinical success was achieved in 80.0% (100/125) of patients with RTI, 83.3% (20/24) of patients with bloodstream infection and 94.7% (36/38) of patients with UTI. Among patients with *P. aeruginosa* infections, the clinical cure rate and mortality rate at Day 28 were 84.5% and 17.2%, respectively. Six patients out of 261 experienced adverse drug reactions (mild/moderate/severe: 4/1/1); cefiderocol was withdrawn for three patients. The outcome was recovery for five patients, and one case was fatal (patient experienced toxic epidermal necrolysis).

Conclusions: In patients with a range of GNBIs in the EAP who were predominantly infected by *P. aeruginosa* and/or treated in the ICU, cefiderocol treatment was effective and well tolerated, with high clinical success and low mortality rates.

OBJECTIVES

In the PERSEUS retrospective study, patients were treated with cefiderocol through the early access programme (EAP) in Spain [1]. The key objectives of this study were to assess the baseline characteristics and the clinical outcomes in patients who were treated with cefiderocol for up to 28 days in the PERSEUS study.

METHODS

Study design: a retrospective, multicentre, observational study in patients receiving cefiderocol for the first time in the EAP in Spain.

Inclusion criteria: adult hospitalised patients treated with cefiderocol consecutively for ≥72 hours for a confirmed Gram-negative bacterial infection.

Exclusion criteria: confirmed *Acinetobacter* spp. at baseline; confirmed cefiderocol-resistant Gram-negative pathogen at baseline; incomplete medical records; enrolled into other clinical studies of investigational products.

Endpoints: baseline patient characteristics, Gram-negative bacterial pathogens, cefiderocol use pattern, clinical success (composite of clinical cure and/or survival at Day 28), clinical cure (cessation of antibiotic treatment due to improvement of signs and symptoms) at end of treatment and all-cause mortality at Day 28.

RESULTS

Patient characteristics	(N=261)	Main comorbidities	
Sex, male	202 (77.4%)	Immunosuppression	79 (30.3%)
Age, median (Q1–Q3), years	61 (49–68)	Tumour (solid/haematological)	62 (23.8%)
CCI score, median (Q1–Q3)	3 (2–4)	Diabetes	58 (22.2%)
APACHE II score, median (Q1–Q3)	15.0 (10.5–22)	Transplant recipient	54 (20.7%)
Symptomatic COVID-19	63 (24.1%)	Chronic renal disease	34 (13.0%)
ECMO	12 (4.6%)	COPD	27 (10.3%)

63.2% (n=165)
ICU at the time of cefiderocol

47.1% (n=123)
Mechanical ventilation

28.0% (n=73)
Septic shock

27.2% (n=71)
Renal replacement therapy

Baseline Gram-negative pathogens and rationale for cefiderocol administration	Overall (N=261)
Gram-negative pathogen, n (%)	
Pseudomonas aeruginosa	174 (66.7)
Stenotrophomonas maltophilia	20 (7.7)
***Pseudomonas* spp.**	15 (5.7)
Other non-fermenters[a]	14 (5.4)
Klebsiella pneumoniae	26 (10.0)
Other Enterobacterales[b]	12 (4.6)
Secondary bacteraemia, n (%)[c]	45 (18.9)
Polymicrobial infection, n (%)	51 (19.5)
Previous colonisation, n (%)[d]	135 (52.9)
Previous treatment with antibiotics, n (%)	219 (83.9)
Rationale for administering cefiderocol[e]	
Resistance to all tested antibiotics	169 (64.8)
Treatment failure with prior antibiotics	116 (44.4)
Adverse events to other susceptible antibiotics	21 (8.0)
Other	26 (10.0)
Cefiderocol treatment duration, median (range), days	10.0 (7.0–14.0)
Cefiderocol combination therapy, n (%)	91 (34.9)
Adverse drug reactions, n (%)	6 (2.3)
Serious adverse drug reactions, n (%)	3 (1.1)
Serious adverse drug reactions leading to death, n (%)	1 (0.4)

[a]*Burkholderia cepacia* complex (8); *Achromobacter* spp. (5); *Ralstonia mannitolilytica* (1); [b]*Serratia marcescens* (5); *Enterobacter cloacae* (3); *Klebsiella oxytoca* (2); *Citrobacter freundii* (1); other *Serratia* sp. (1); [c]Missing (23); [d]Missing (6); [e]Not mutually exclusive.

RESULTS CONT'D

Patient attrition

Screened population
(N=314)
↓
Eligible population
(N=274)
↓
Primary analysis population
(N=261)

Site of infection

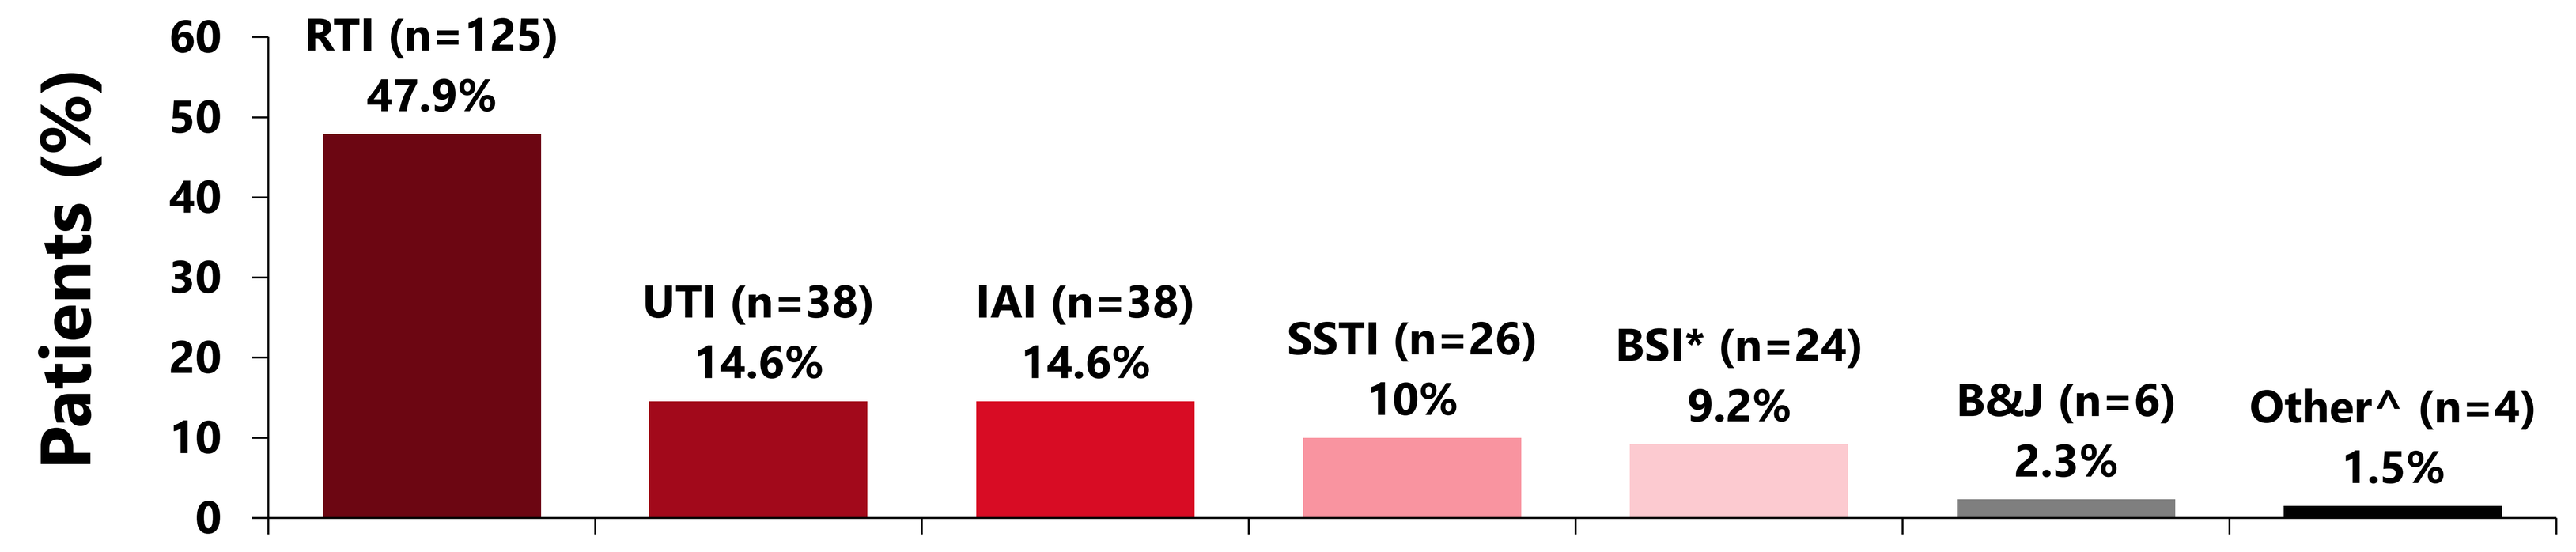
Patients (%)
60
50
40
30
20
10
0
RTI (n=125)
47.9%
UTI (n=38)
14.6%
IAI (n=38)
14.6%
SSTI (n=26)
10%
BSI* (n=24)
9.2%
B&J (n=6)
2.3%
Other^ (n=4)
1.5%

*Includes catheter-related (15), unknown source (9). ^Central nervous system infection (2); mediastinitis (2).

Clinical outcomes

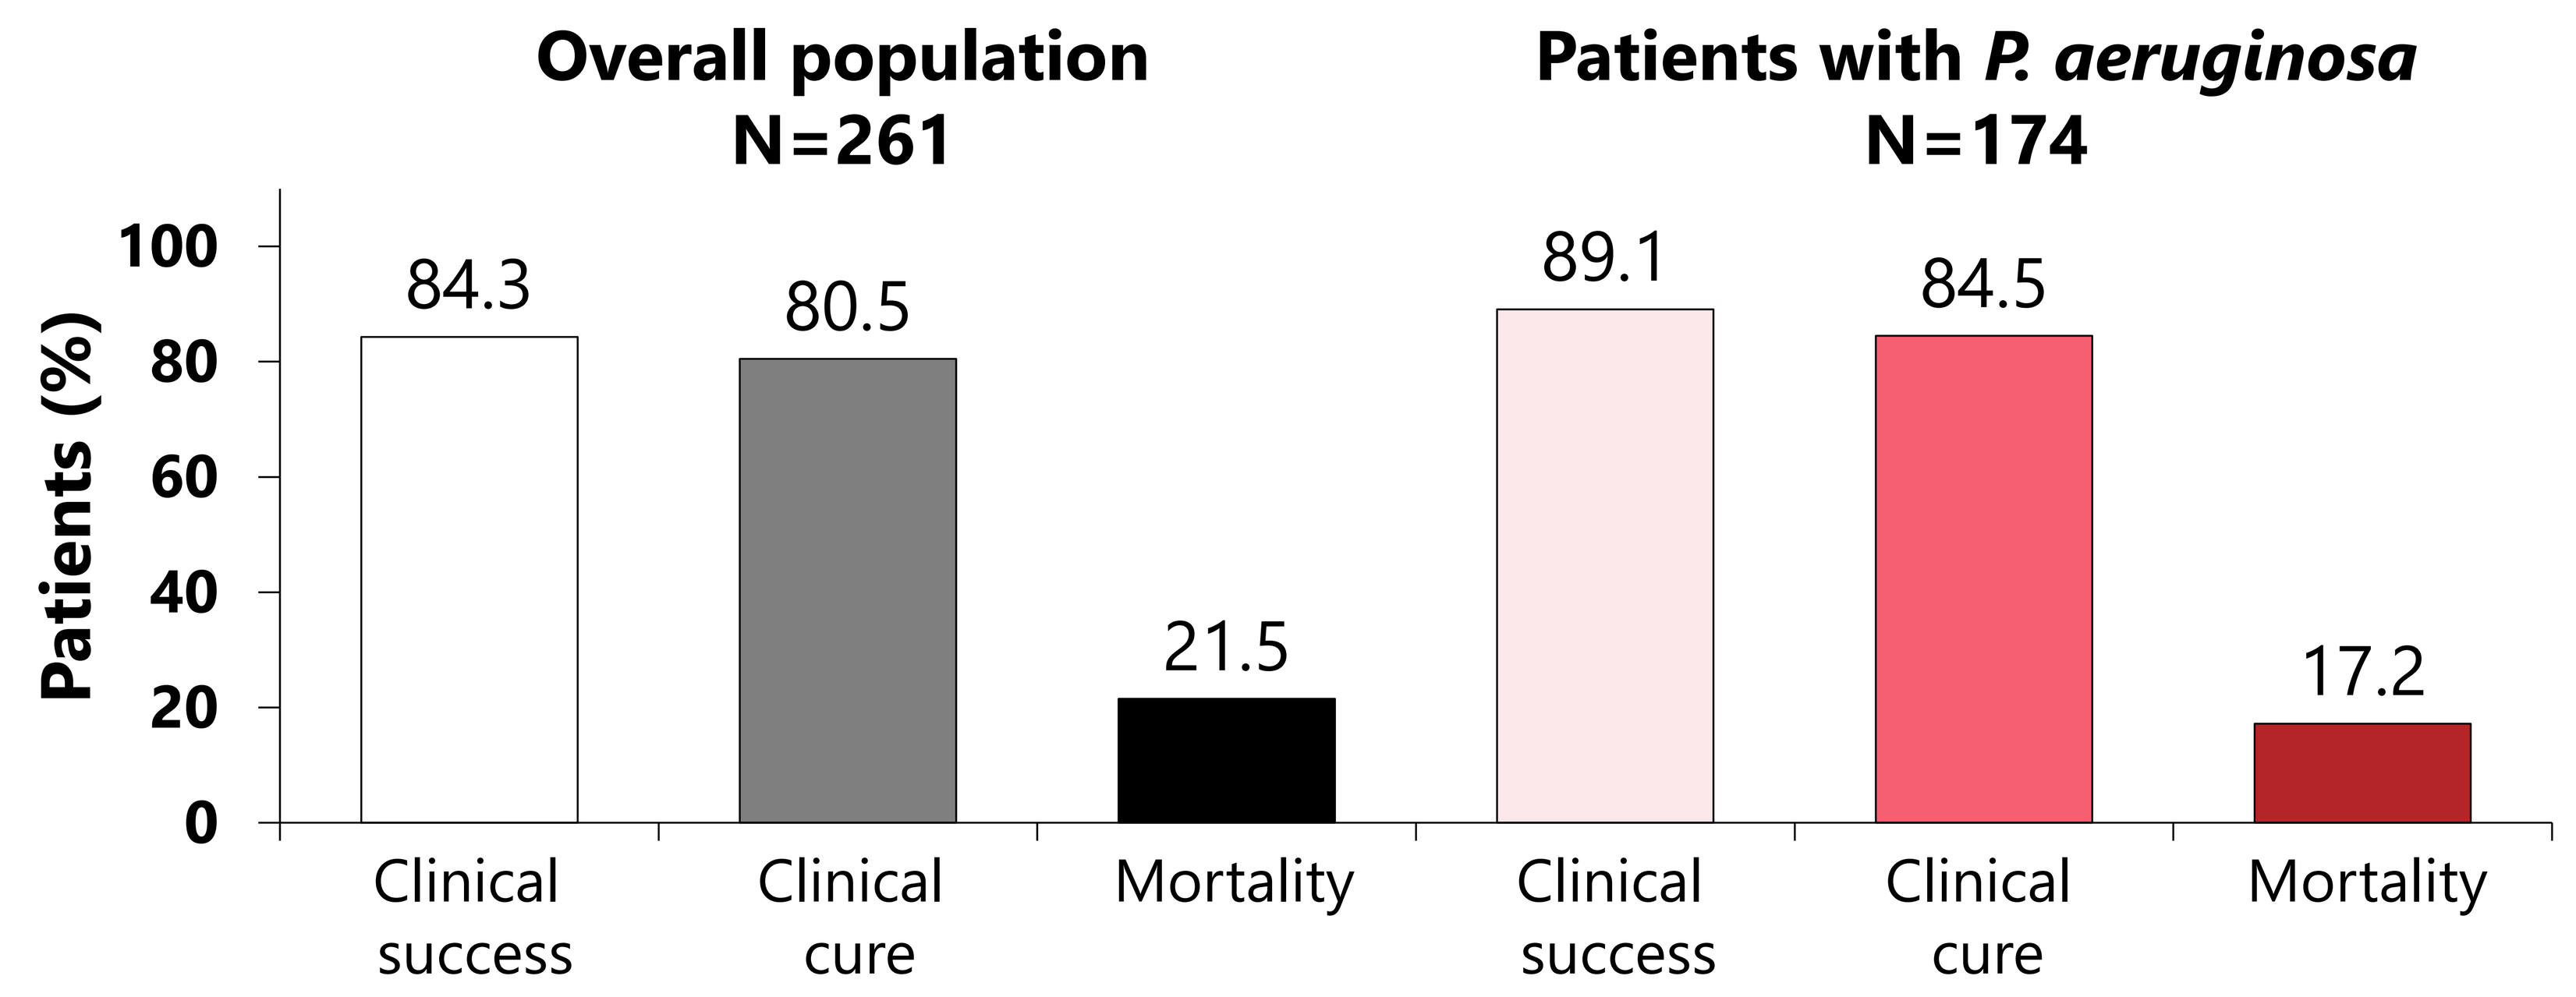
Overall population
N=261
Patients with *P. aeruginosa*
N=174
Patients (%)
100
80
60
40
20
0
84.3
80.5
21.5
89.1
84.5
17.2
Clinical success
Clinical cure
Mortality
Clinical success
Clinical cure
Mortality

Clinical outcomes by infection site

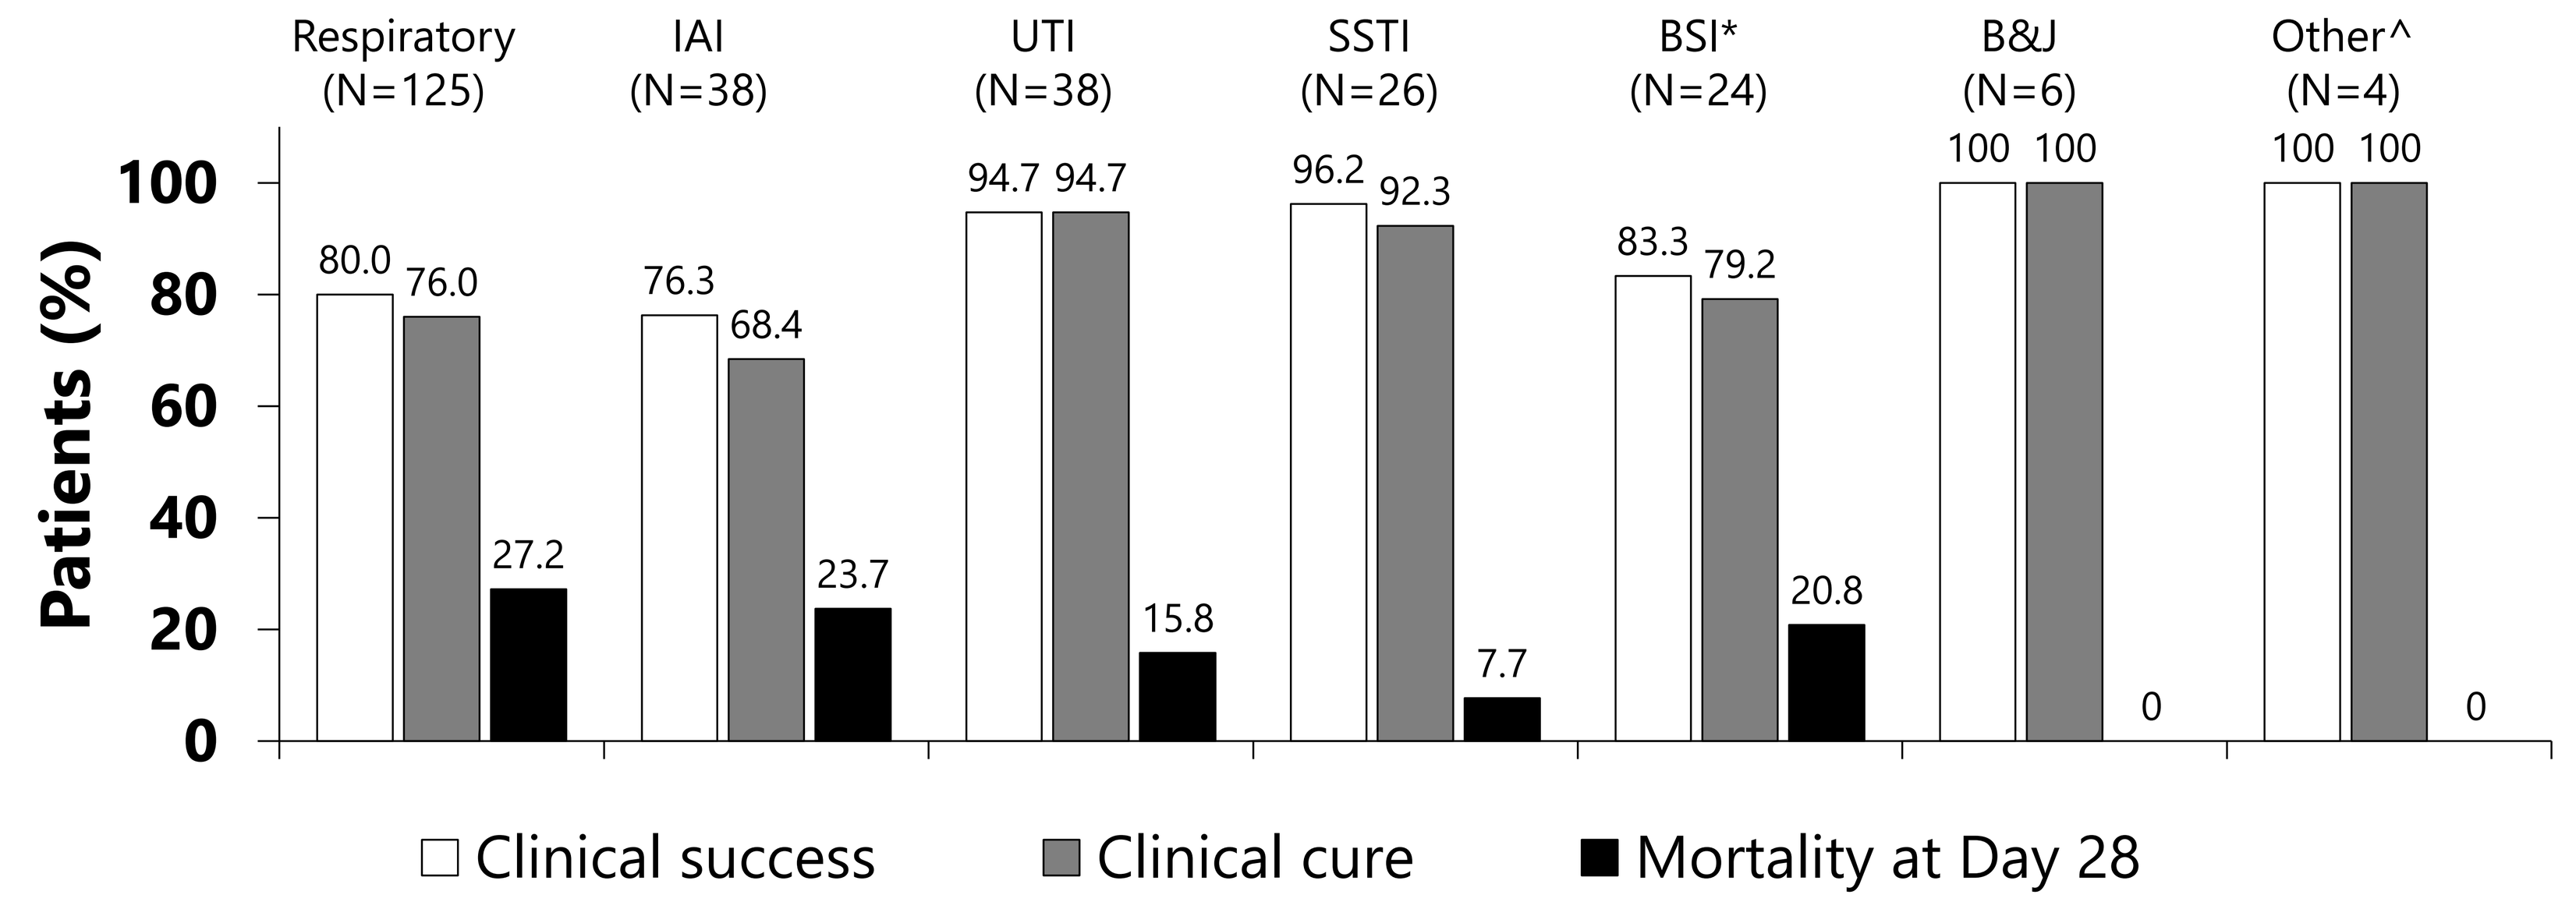
Respiratory (N=125)
IAI (N=38)
UTI (N=38)
SSTI (N=26)
BSI* (N=24)
B&J (N=6)
Other^ (N=4)
Patients (%)
100
80
60
40
20
0
80.0 76.0 27.2
76.3 68.4 23.7
94.7 94.7 15.8
96.2 92.3 7.7
83.3 79.2 20.8
100 100 0
100 100 0
Clinical success
Clinical cure
Mortality at Day 28

*Includes catheter-related (15), unknown source (9); ^Central nervous system infection (*n*=2), mediastinitis (*n*=2).

Clinical outcomes by antibiotic use

Number of days with prior antibiotics	Overall n (%) N=212	Clinical success n (%)	Clinical cure n (%)	Mortality Day 28 n (%)
≤3	55 (25.9)	49 (89.1)	49 (89.1)	9 (16.4)
4–7	70 (33.0)	62 (88.6)	59 (84.3)	13 (18.6)
>7	87 (41.0)	67 (77.0)	60 (69.0)	25 (28.7)
Cefiderocol as first line	N=261			
No	219 (83.9)	182 (83.1)	172 (78.5)	50 (22.8)
Yes	42 (16.1)	38 (90.5)	38 (90.5)	6 (14.3)
Combination treatment	N=261			
No	170 (65.1)	150 (88.2)	143 (84.1)	30 (17.6)
Yes	91 (34.9)	70 (76.9)	67 (73.6)	26 (28.6)

Clinical outcomes by resistance to BL–BLIs

Overall	Resistance phenotype	Clinical success	Clinical cure	Mortality Day 28
C/T-R, n/N′ (%)	99/130* (76.2)	85/99 (85.9)	82/99 (82.8)	17/99 (17.2)
CZA-R, n/N′ (%)	134/160* (83.8)	111/134 (82.8)	107/134 (79.9)	31/134 (23.1)
Patients with *P. aeruginosa*, n (%)				
C/T-R, n/N′ (%)	75/105* (71.4)	67/75 (89.3)	65/75 (86.7)	10/75 (13.3)
CZA-R, n/N′ (%)	96/112* (85.7)	83/96 (86.5)	80/96 (83.3)	18/96 (18.8)

*Number of patients with known susceptibility results.

CONCLUSIONS

- Cefiderocol treatment was effective with high clinical success and clinical cure rates in patients with serious Gram-negative bacterial infections, including patients with MDR and BL–BLI-resistant *P. aeruginosa* and other non-fermenters.
- Early administration of cefiderocol showed a numerical trend towards higher clinical cure and lower mortality rates.

Acknowledgements

The PERSEUS study was funded by Shionogi. Sarda J, Verardi S, Gonzalez AJ are employees of Shionogi. Editorial support was provided by Highfield, Oxford, UK and this support was funded by Shionogi.

Reference

1. ClinicalTrials.gov: NCT05789199.

Abbreviations

APACHE, Acute Physiology and Chronic Health Evaluation; BL–BLIs, beta-lactam–beta-lactamase inhibitors; BSI, bloodstream infection; B&J, bone and joint; CCI, Charlson Comorbidity Index; COPD, chronic obstructive pulmonary disease; COVID-19, coronavirus disease 2019; C/T-R, ceftolozane-tazobactam resistant; CZA-R, ceftazidime-avibactam resistant; ECMO, extracorporeal membrane oxygenation; IAI, intra-abdominal infection; LOS, length of stay; MDR, multidrug resistant; Q, quartile; RTI, respiratory tract infection; SSTI, skin and soft tissue infection; UTI, urinary tract infection.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ECCMID 2024 and the authors of the poster.