In vitro antiviral activity of ensitrelvir against novel Omicron strains of SARS-CoV-2

Teruhisa Kato[1], Haruaki Nobori[1], Kae Inoue[2], Takashi Hashimoto[2]
[1] Shionogi & Co., Ltd., Osaka, Japan
[2] Shionogi TechnoAdvance Research, Co., Ltd., Osaka, Japan

SHIONOGI

Background

Severe acute respiratory syndrome coronavirus 2 (SARS-CoV-2), which causes the novel coronavirus infection (COVID-19), has shown repeated mutations in the spike protein, and new mutant variants are constantly emerging. Ensitrelvir, developed by Shionogi & Co., Ltd. discovered through joint research with Hokkaido University, is a COVID-19 therapeutic agent that specifically inhibits the 3CL protease of SARS-CoV-2. It was approved in Japan in November 2022 as an emergency approval. We have already evaluated the antiviral activity in various mutant strains. We also evaluated in vitro efficacy of ensitrelvir against newly emerging Omicron strains.

Methods

In vitro antiviral activity of ensitrelvir against the SARS-CoV-2 Omicron strains were evaluated using VeroE6 expressing type II transmembrane serine protease (VeroE6/TMPRSS2) cells and human primary airway epithelial cells (hAEC). Antiviral activity was assessed by cytopathic effect reduction in VeroE6/TMPRSS2 cells and the virus released to the apical side in hAEC cells.

Results

Ensitrelvir was effective against SARS-CoV-2 Omicron BA.4.6, BA.5.2.1, BF.7, BF.7.4.1, BQ.1.1, CH.1.1.11, XBB.1.5, XBB.1.9.1, XBB.1.16, and XBF strains with EC_{50} = 0.30, 0.37, 0.51, 0.55, 0.48, 0.38, 0.57, 0.99, 0.33, and 0.29 μmol/L in VeroE6/TMPRSS2 cells. In addition, the antiviral activity of ensitrelvir against the Omicron BE.1 (BA.5-like) and XBB.1.5 strain in hAEC showed EC_{90} = 60.1 nmol/L.

CellTiter Glo assay using VeroE6/TMPRSS2 cells

VeroE6/TMPRSS2 cells
infected with SARS-CoV-2
(omicron strain)

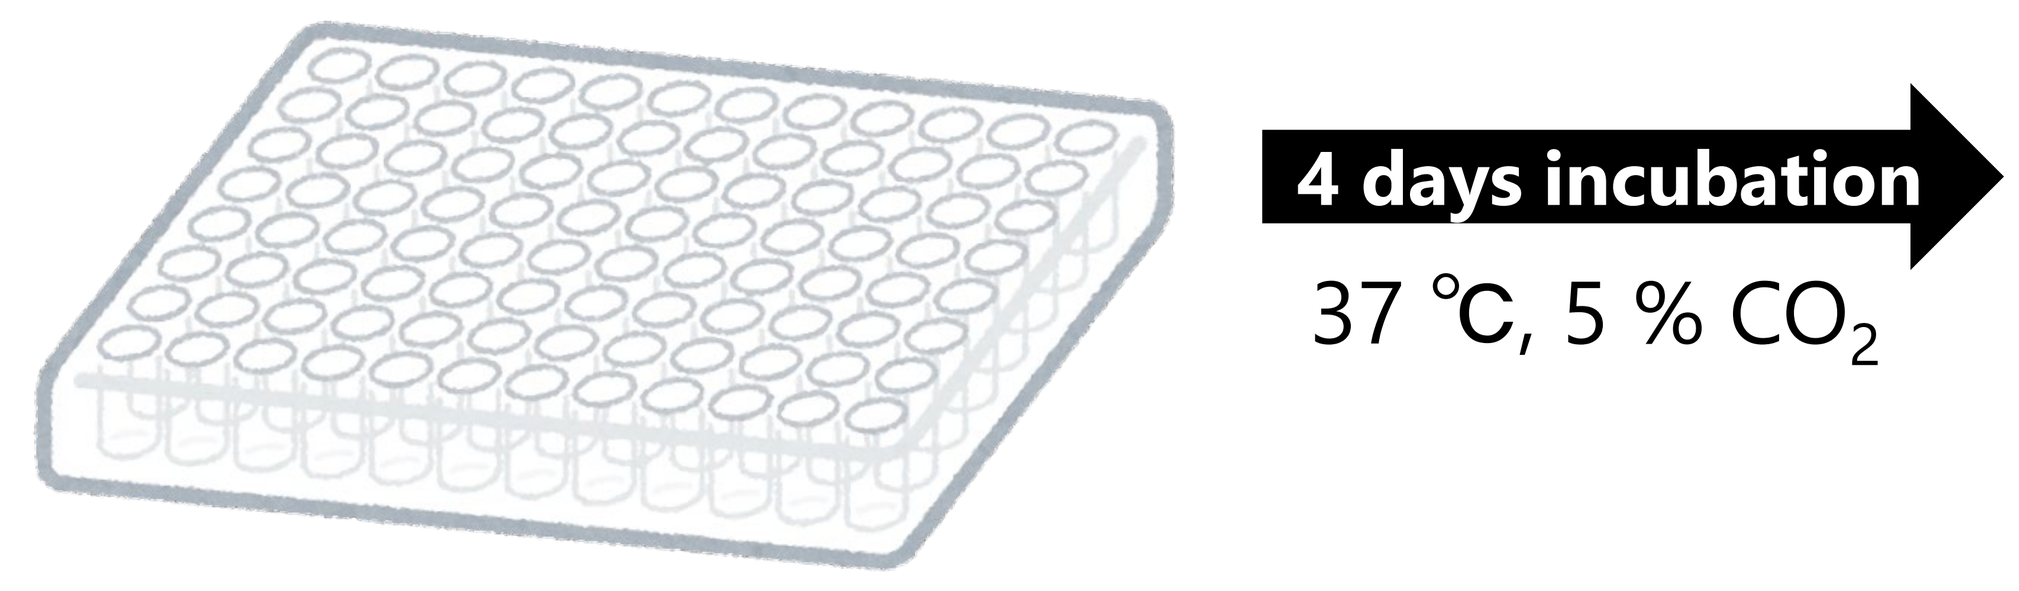
4 days incubation
37 ℃, 5 % CO2

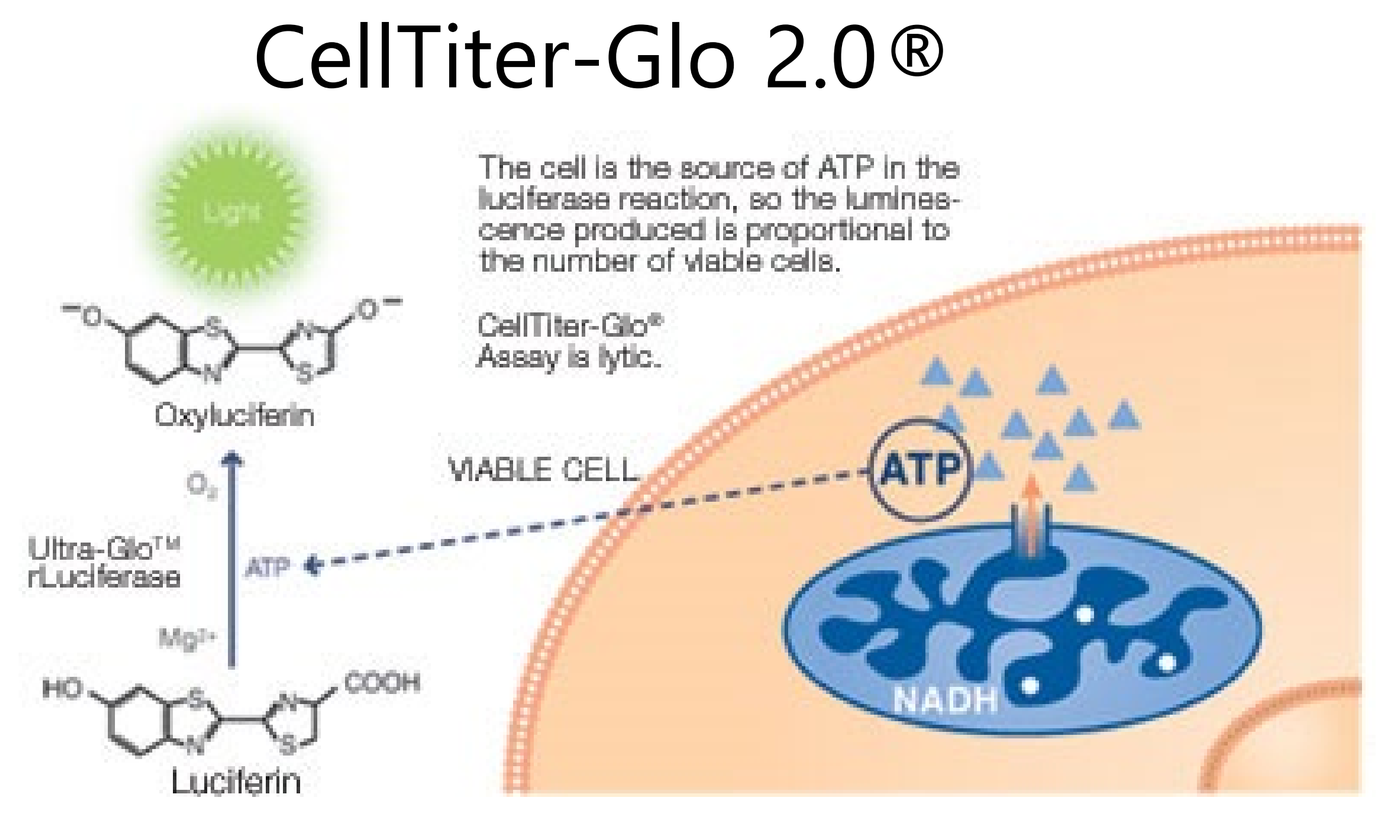
CellTiter-Glo 2.0®
The cell is the source of ATP in the luciferase reaction, so the luminescence produced is proportional to the number of viable cells.
CellTiter-Glo® Assay is lytic.
Oxyluciferin
VIABLE CELL
ATP
NADH
Luciferin

Ref. CellTiter-Glo® Luminescent Cell Viability Assay (promega.jp)

Table 1 *In vitro* antiviral activities against SARS-CoV-2 in VeroE6/TMPRSS2

WHO label	Pango lineage	Strain name	EC_{50} (μmol/L, CPE) Ensitrelvir	EC_{50} (μmol/L, CPE) Remdesivir	3CLpro mutation
Wuhan	A	WK-521	0.37 ± 0.06	1.9 ± 0.1	-
Omicron	BA.1.18	TY38-873	0.29 ± 0.05	1.1 ± 0.3	P132Y
Omicron	BA.1.1	TY38-871	0.36 ± 0.08	1.0 ± 0.1	P132H
Omicron	BA.2	TY40-385	0.52 ± 0.09	1.0 ± 0.2	P132H
Omicron	BA.2.12.1	TY41-721	0.24 ± 0.08	0.49 ± 0.20	P132H
Omicron	BA.2.75	TY41-716	0.30 ± 0.03	0.91 ± 0.08	P132H
Omicron	BA.4.1	TY41-703	0.22 ± 0.07	0.65 ± 0.19	P132H
Omicron	**BA.4.6**	**TY41-763**	**0.30 ± 0.07**	**0.87 ± 0.07**	**P132H**
Omicron	**BA.5.2.1**	**TY41-704**	**0.37 ± 0.02**	**1.7 ± 0.3**	**P132H**
Omicron	BE.1 (BA.5-like)	TY41-702	0.40 ± 0.08	1.3 ± 0.5	P132H
Omicron	**BF.7**	**TY41-820**	**0.51 ± 0.07**	**1.2 ± 0.1**	**P132H**
Omicron	**BF.7.4.1**	**TY41-828**	**0.55 ± 0.08**	**1.6 ± 0.5**	**P132H**
Omicron	**BQ.1.1**	**TY41-796**	**0.48 ± 0.04**	**2.2 ± 0.7**	**P132H**
Omicron	**CH.1.1.11**	**TY41-832**	**0.38 ± 0.09**	**1.2 ± 0.3**	**P132H**
Omicron	XBB.1	TY41-795	0.33 ± 0.10	0.95 ± 0.08	P132H
Omicron	**XBB.1.5**	**23-018**	**0.57 ± 0.07**	**1.0 ± 0.2**	**P132H**
Omicron	**XBB.1.9.1**	**TY41-951**	**0.99 ± 0.11**	**3.0 ± 0.3**	**P132H**
Omicron	**XBB.1.16**	**TY41-984**	**0.33 ± 0.03**	**1.1 ± 0.2**	**P132H**
Omicron	**XBF**	**TY41-831**	**0.29 ± 0.01**	**1.0 ± 0.2**	**P132H**
Omicron	XE	TY41-686	0.44 ± 0.04	1.1 ± 0.4	P132H

Mean ± SD, The mean and SD were calculated from 3 independent experiments.

Antiviral activity using hAEC cells (MucilAir™ nasal)

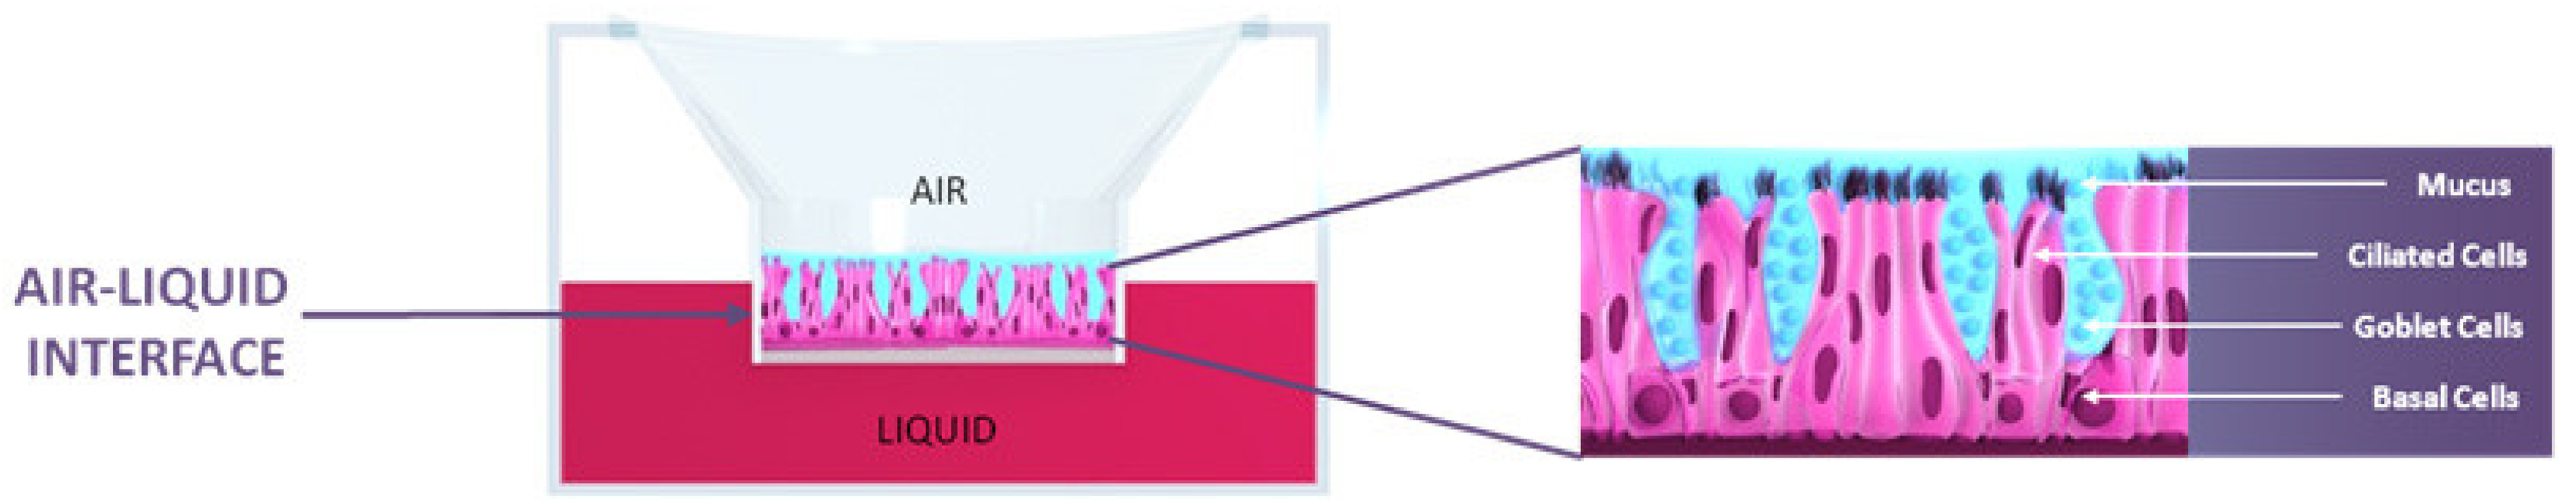
AIR-LIQUID INTERFACE
AIR
LIQUID
Mucus
Ciliated Cells
Goblet Cells
Basal Cells

Ref. MucilAir™ is an in vitro cell model of the human upper airway epithelium cultured at the air liquid interface (epithelix.com)

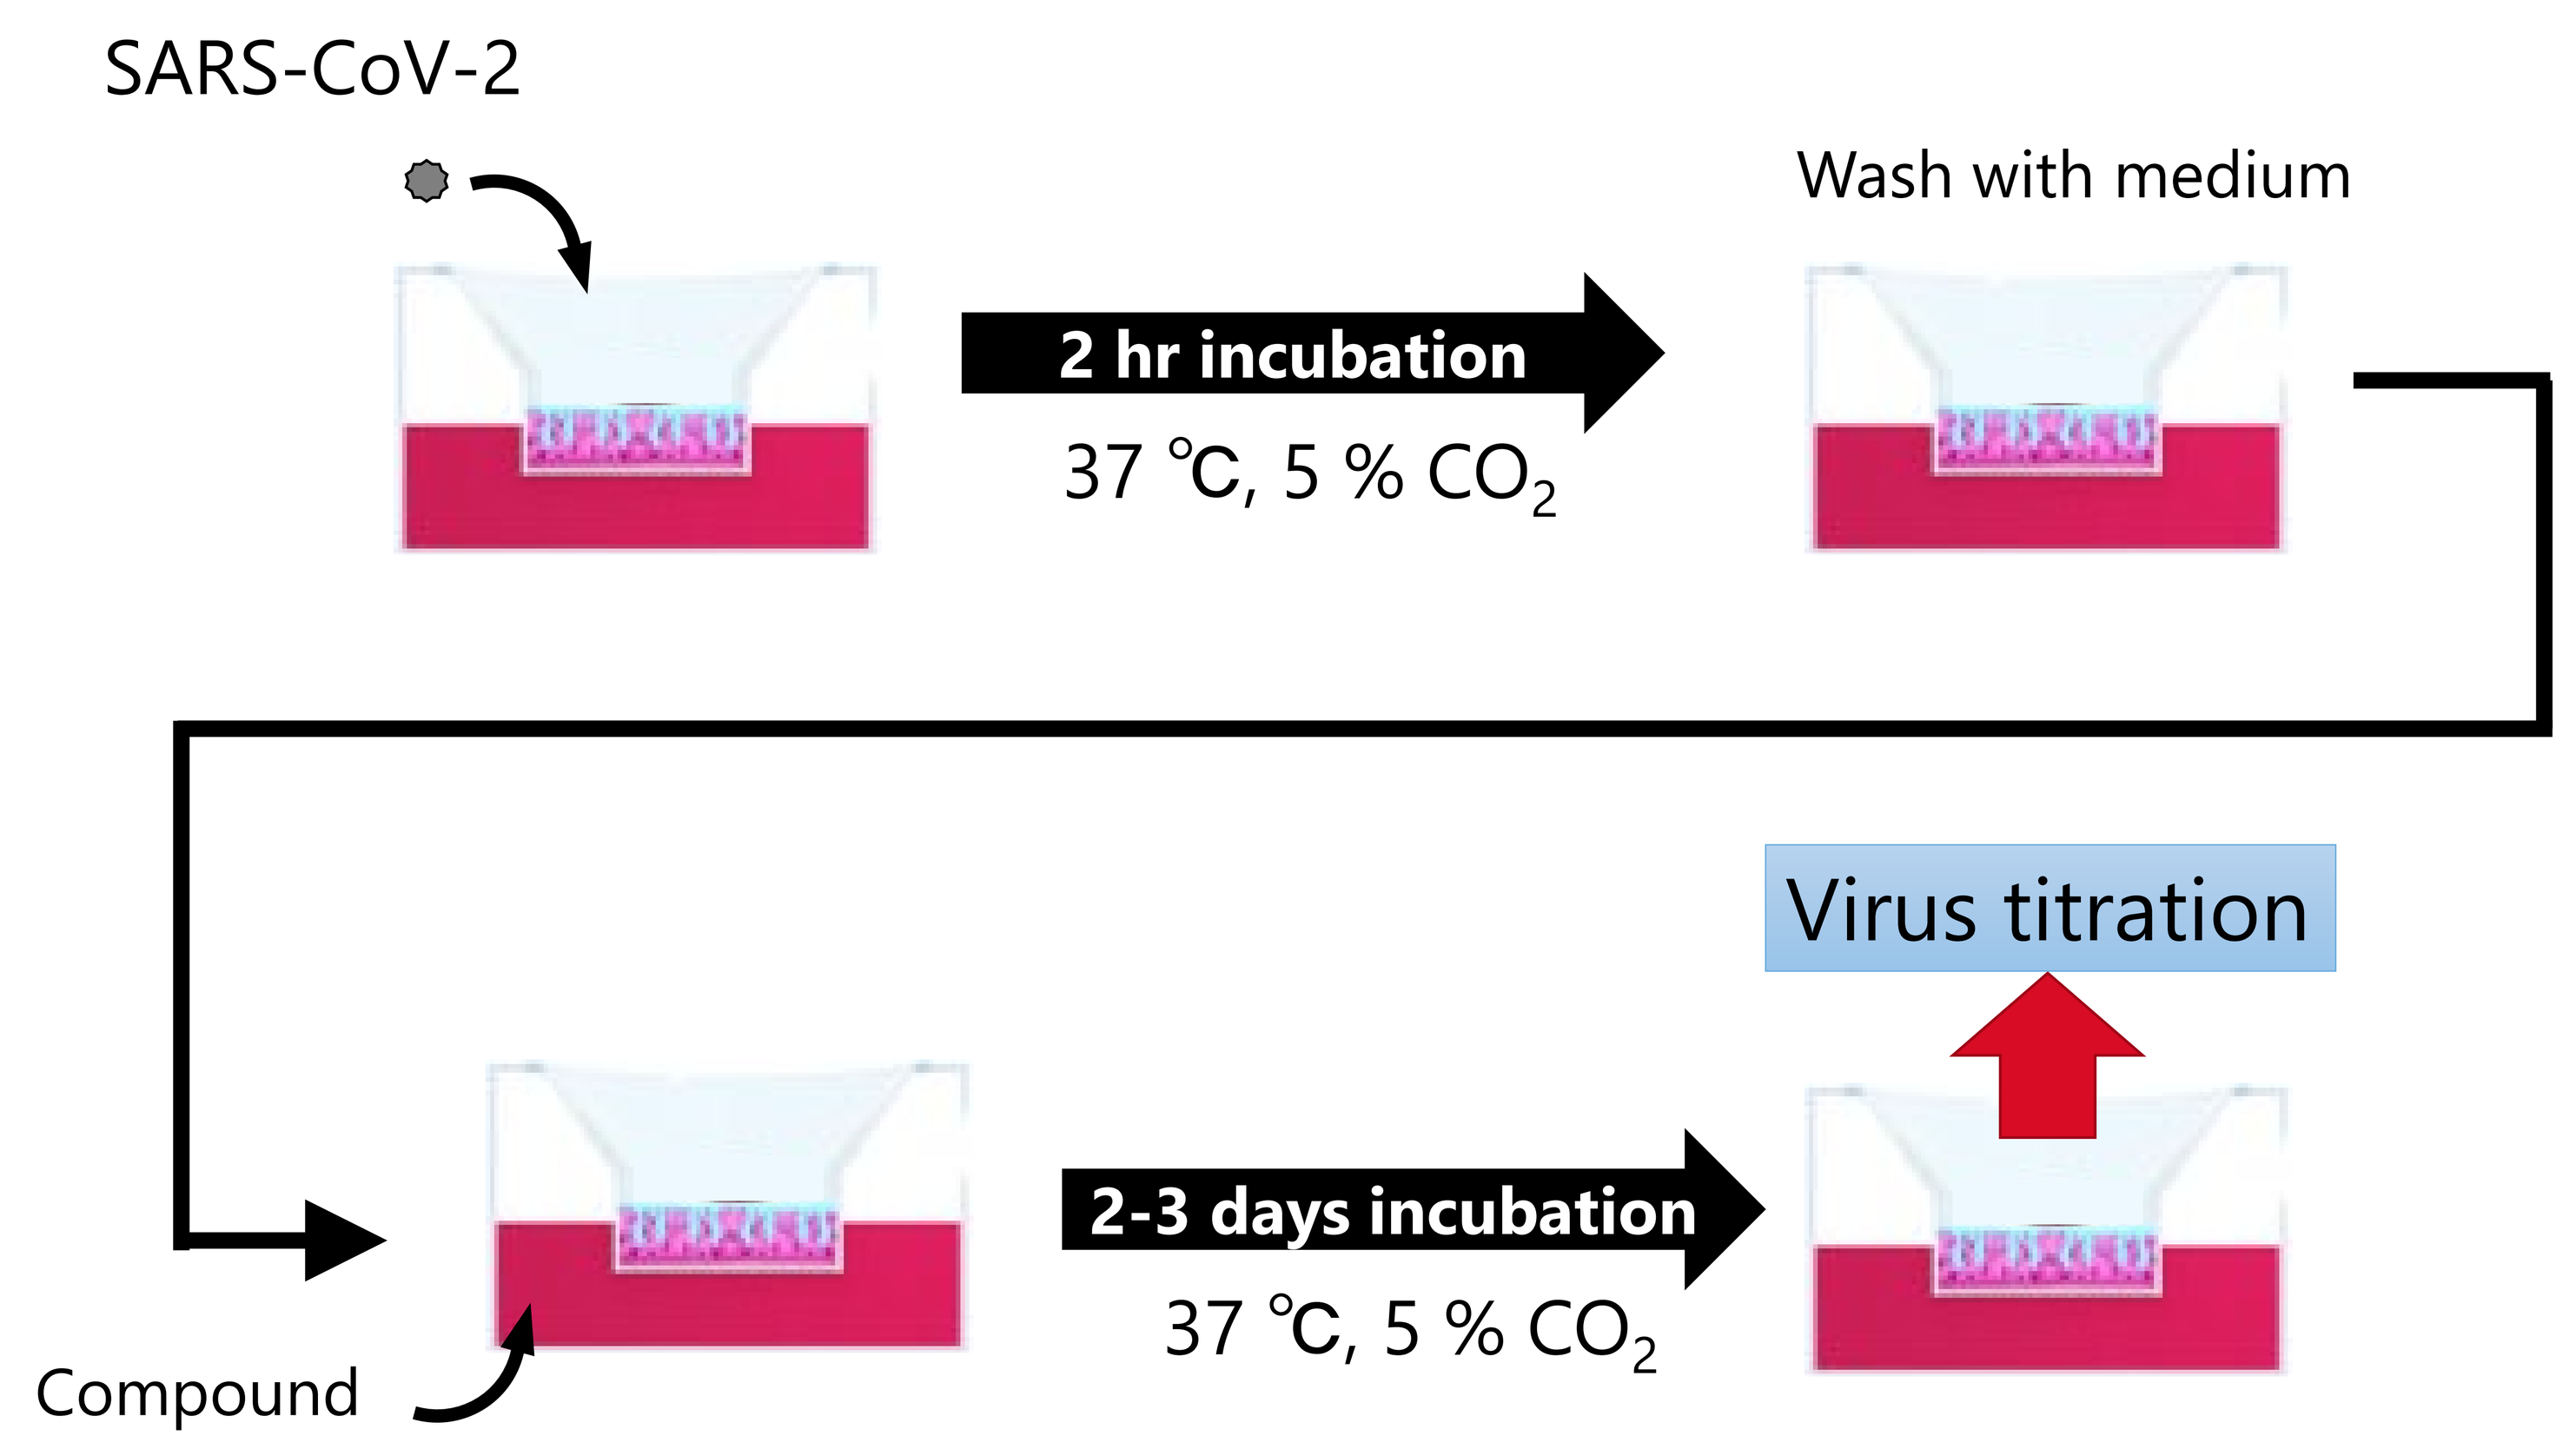
SARS-CoV-2
2 hr incubation
37 ℃, 5 % CO2
Wash with medium
Virus titration
2-3 days incubation
37 ℃, 5 % CO2
Compound

Table 2 *In vitro* antiviral activities in hAEC (MucilAir™ nasal)

WHO label	Pango lineage	Strain name	EC_{90} (nmol/L, viral titer) Ensitrelvir	EC_{90} (nmol/L, viral titer) Nirmatrelvir	EC_{90} (nmol/L, viral titer) Remdesivir
Delta		TY11-927	117 ± 26	46.8 ± 10.2	35.9 ± 14.0
Omicron	BA.1.18	TY38-873	160 ± 57	69.2 ± 14.1	21.1 ± 9.0
Omicron	**BE.1 (BA.5-like)**	**TY41-702**	**60.1 ± 32.8**	**45.1 ± 21.2**	NT

Mean ± SD, The mean and SD were calculated from 3 independent experiments.

Conclusion

Conclusion: Ensitrelvir has shown antiviral activity against novel Omicron strains in this study and the activity was comparable to that of the previous mutant variants. The sequences of these Omicron strains are registered in the database GISAID and have shown no mutations in the active domain of 3CL protease, which is the target site of ensitrelvir. It is possible that novel mutant strains will emerge in the future, but the antiviral activity of ensitrelvir is not likely to be affected by mutations in the spike protein. It is expected that ensitrelvir shows comparable antiviral activity against novel variants unless mutations are observed near the active center of 3CL protease.

Acknowledgement

SARS-CoV-2 strains were kindly gifts from National Institute of Infectious Diseases (NIID).

COI disclosure

Authors are employees of Shionogi & Co., Ltd. or Shionogi TechnoAdvance Research, Co., Ltd. Some authors are shareholder of Shionogi & Co., Ltd.