P0543

Prediction of transmission mitigation based on the antiviral effect of Ensitrelvir and simulation of its impact on epidemics

ECCMID 2024
27-30 Apr. 2024

Shogo Miyazawa[1], Masaya Saito[2], Ayano Hata[1], Takamichi Baba[1], Yoshitake Kitanishi[1]

1. Data Science Department, Shionogi & Co., Ltd., Osaka, Japan
2. University of Nagasaki, Nagasaki, Japan

SHIONOGI

Contact: Shogo Miyazawa
Shionogi & Co., Ltd., Japan
Email: shogo.miyazawa@shionogi.co.jp

Background

- Ensitrelvir is an oral SARS-CoV-2 3CL protease inhibitor that has demonstrated a significant reduction in change from baseline in viral RNA at day 4 of treatment compared to placebo in patients with mild to moderate COVID-19.
- In general, it is known that transmission of infectious diseases is associated with higher viral load in infected individuals. To control the spread of infectious diseases, it is important to achieve early negative status for the virus at an early stage through early diagnosis and treatment.
- Here, we attempted to evaluate the long-term impact of the antiviral drug, ensitrelvir, infection control by simulation.

Methods

Concept of this study

Individual viral shedding curves per treatment start time
- ensitrelvir vs Placebo
- The treatment start time was shifted every 0.1 day (every 2.4 hours) in simulation

The ratio of AUC of infectiousness profiles for each treatment start time
- Assuming that the AUC ratio of the infection profile could be converted to transmission mitigation
- Estimated using AUC ratios between ensitrelvir group and placebo

Scenario analysis by SIRS model
- The parameters were the transmission mitigation, the treatment rate and immune duration

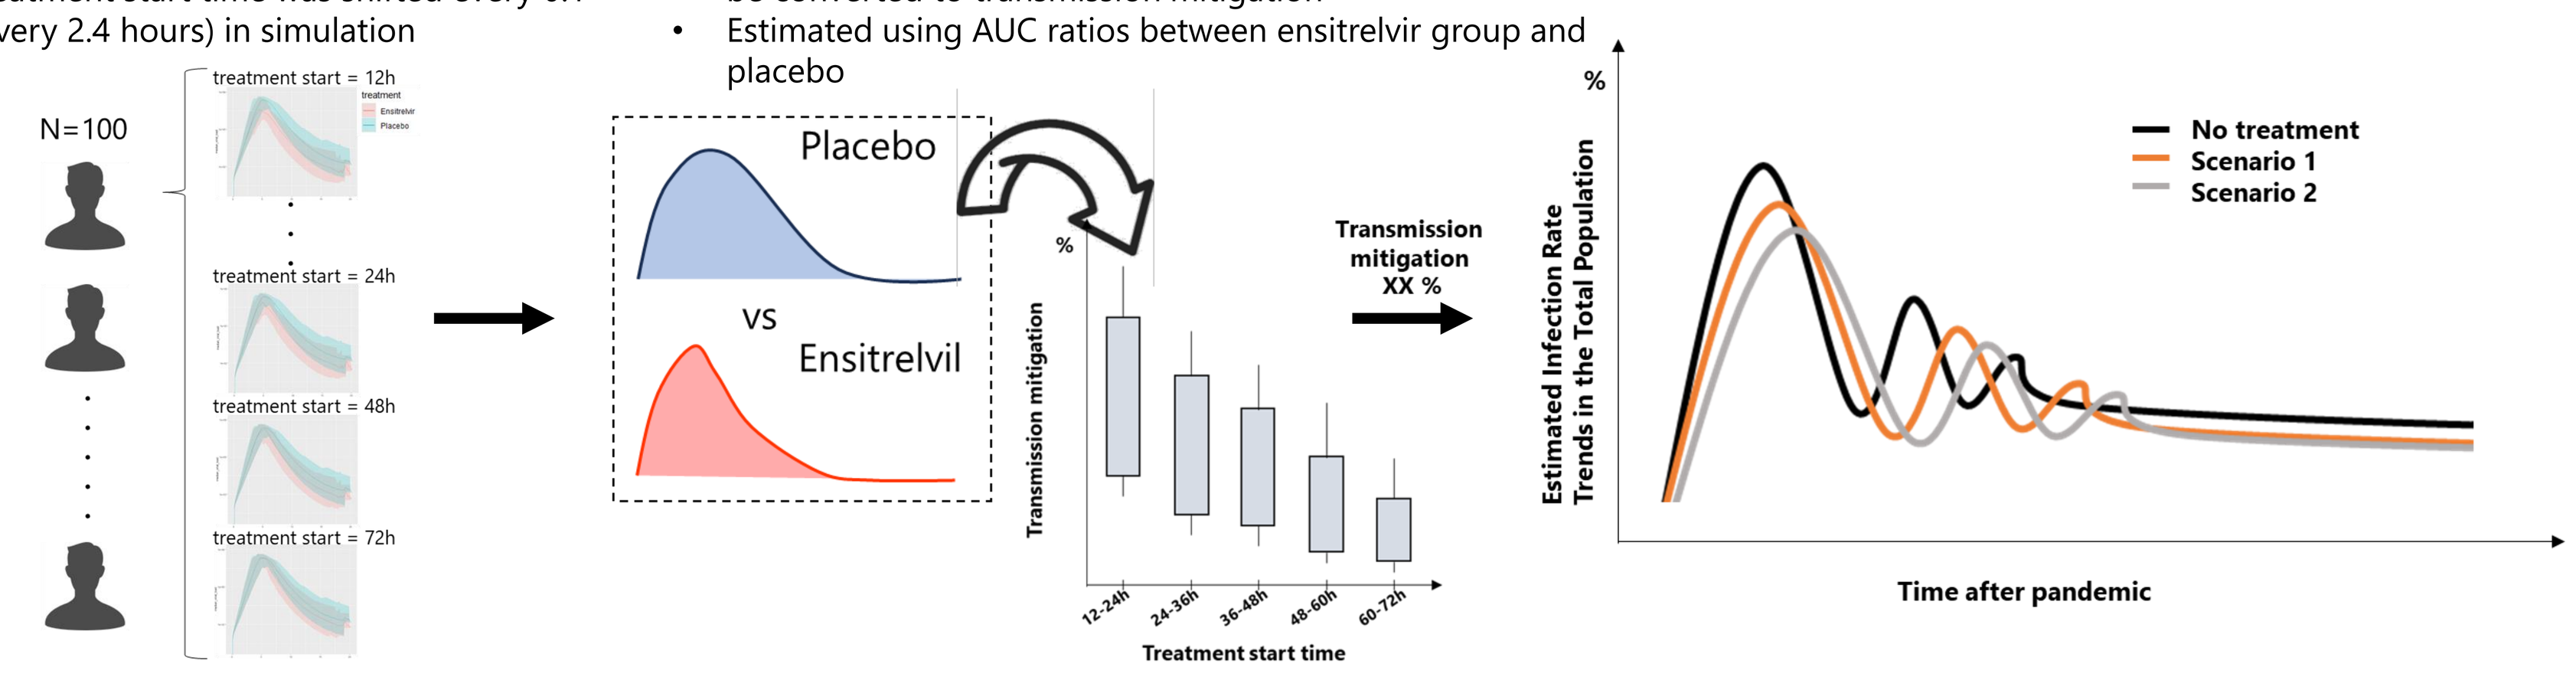
N=100
treatment start = 12h
treatment start = 24h
treatment start = 48h
treatment start = 72h
Placebo
vs
Ensitrelvil
Transmission mitigation
12-24h
24-36h
36-48h
48-60h
60-72h
Treatment start time
Transmission mitigation XX %
Estimated Infection Rate Trends in the Total Population
No treatment
Scenario 1
Scenario 2
Time after pandemic

Simulation of viral RNA

- Based on PPK model (Population Pharmacokinetics model) and Viral Dynamic model of SARS-CoV-2, a drug effect model was constructed to explain viral kinetics after ensitrelvir administration [1,2].
- The drug effect model was constructed using 2167 patients (1447 in ensitrelvir group and 720 in the placebo group) in clinical trials (Phase 2a/2b and Phase 3) of ensitrelvir.
- In this study, we used data from viral kinetic simulations by drug effect model for each dose timing and ensitrelvir/placebo groups.

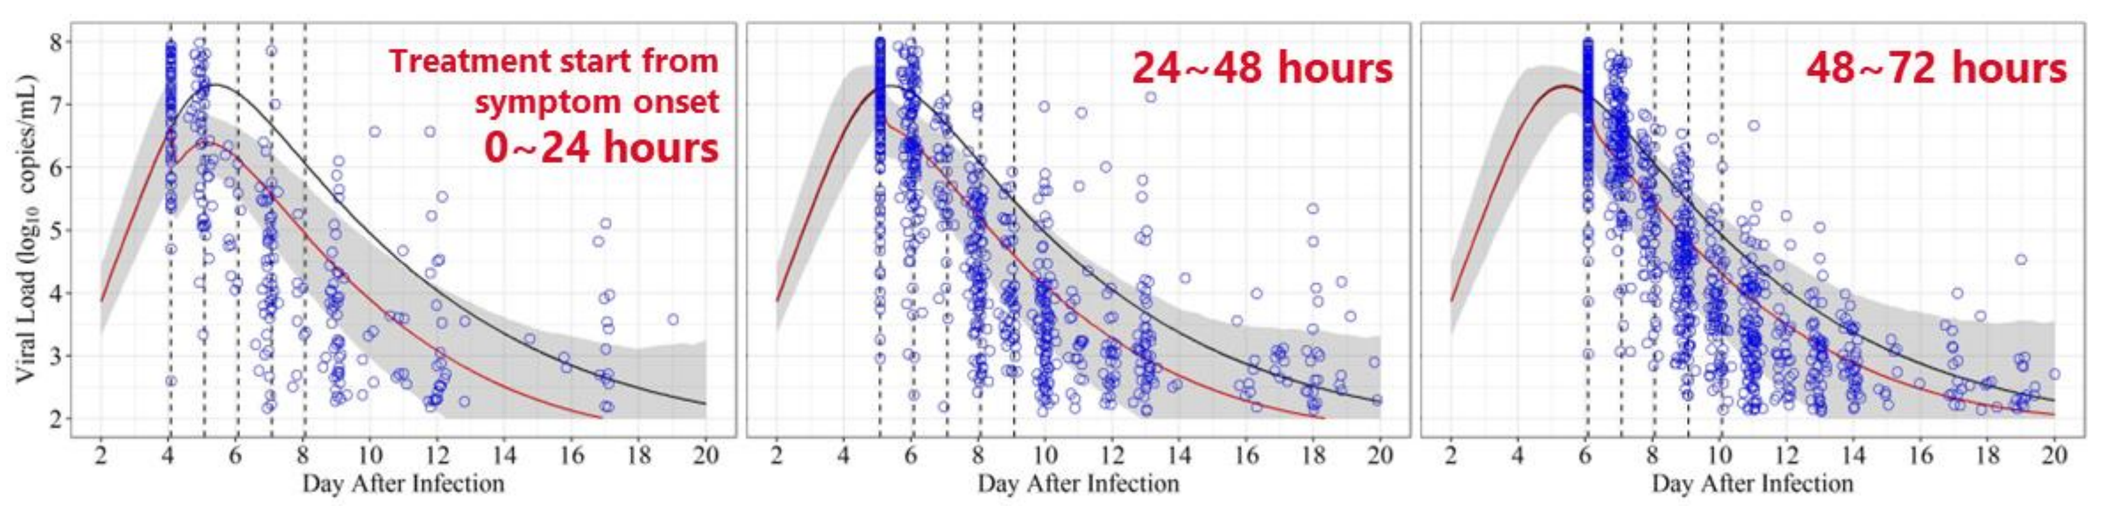
Treatment start from symptom onset 0~24 hours
24~48 hours
48~72 hours
Day After Infection

Calculation of transmission mitigation

- The AUC (area under the curve) of infectiousness profiles was calculated for each treatment start time as the total infectiousness.
- The AUC of ensitrelvir group and that of placebo were calculated for each treatment start time, taking ratio to estimate transmission mitigation. We performed these steps on three scales, on a natural-scale ($V(t)$), logarithmic-scale ($\log_{10} V(t)$), or to a dose–response ($1-\ e^{-\alpha V(t)}$) transform of viral RNA. Where $V(t)$ was viral RNA and $\alpha = 10^{-4}$ was adopted [3]
- The estimates obtained here were used as the effect of ensitrelvir in predicting the number of infections at the population level.
- Based on these infectiousness profiles, we concluded that the natural scale did not reflect the characteristics of the omicron variant due to short infectious period and that others were more applicable.

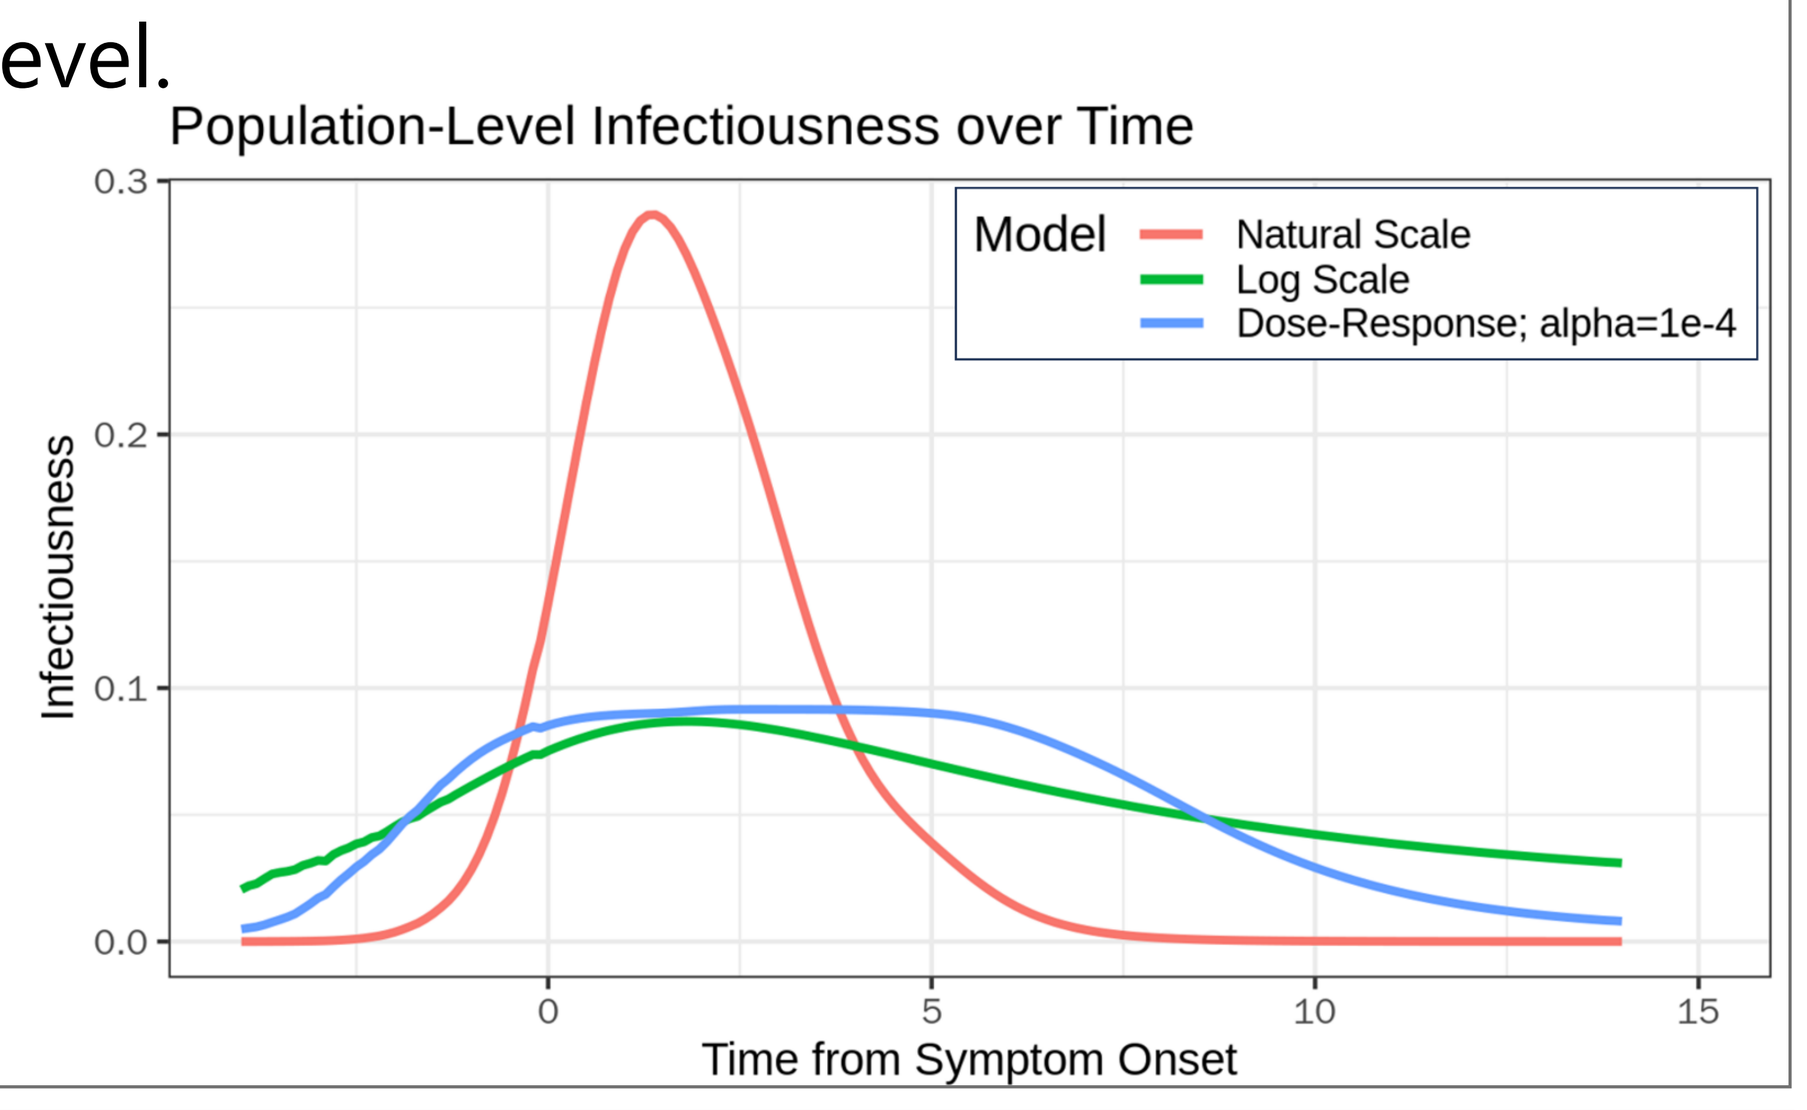
Population-Level Infectiousness over Time
Model
Natural Scale
Log Scale
Dose-Response; alpha=1e-4
Infectiousness
Time from Symptom Onset

Scenario analysis number of infections using transmission mitigation

- Using estimated the effect of transmission mitigation by ensitrelvir, a long-term scenario analysis was performed to assess the impact of ensitrelvir on the number of infections at the population level and estimate reduction ratio in the annual number of infections per treatment rate compared to no treatment.
- In conducting the scenario analysis, the age-structured SIRS model was adopted for this study. This model is a mathematical model of infectious diseases that can represent the epidemic dynamics of infectious diseases while accounting for reinfection[4].
- We assume that a person is born in the youngest age category in the total population N_iat birth rate μ_i. Let α_i be the rate of moving up one age category per year.

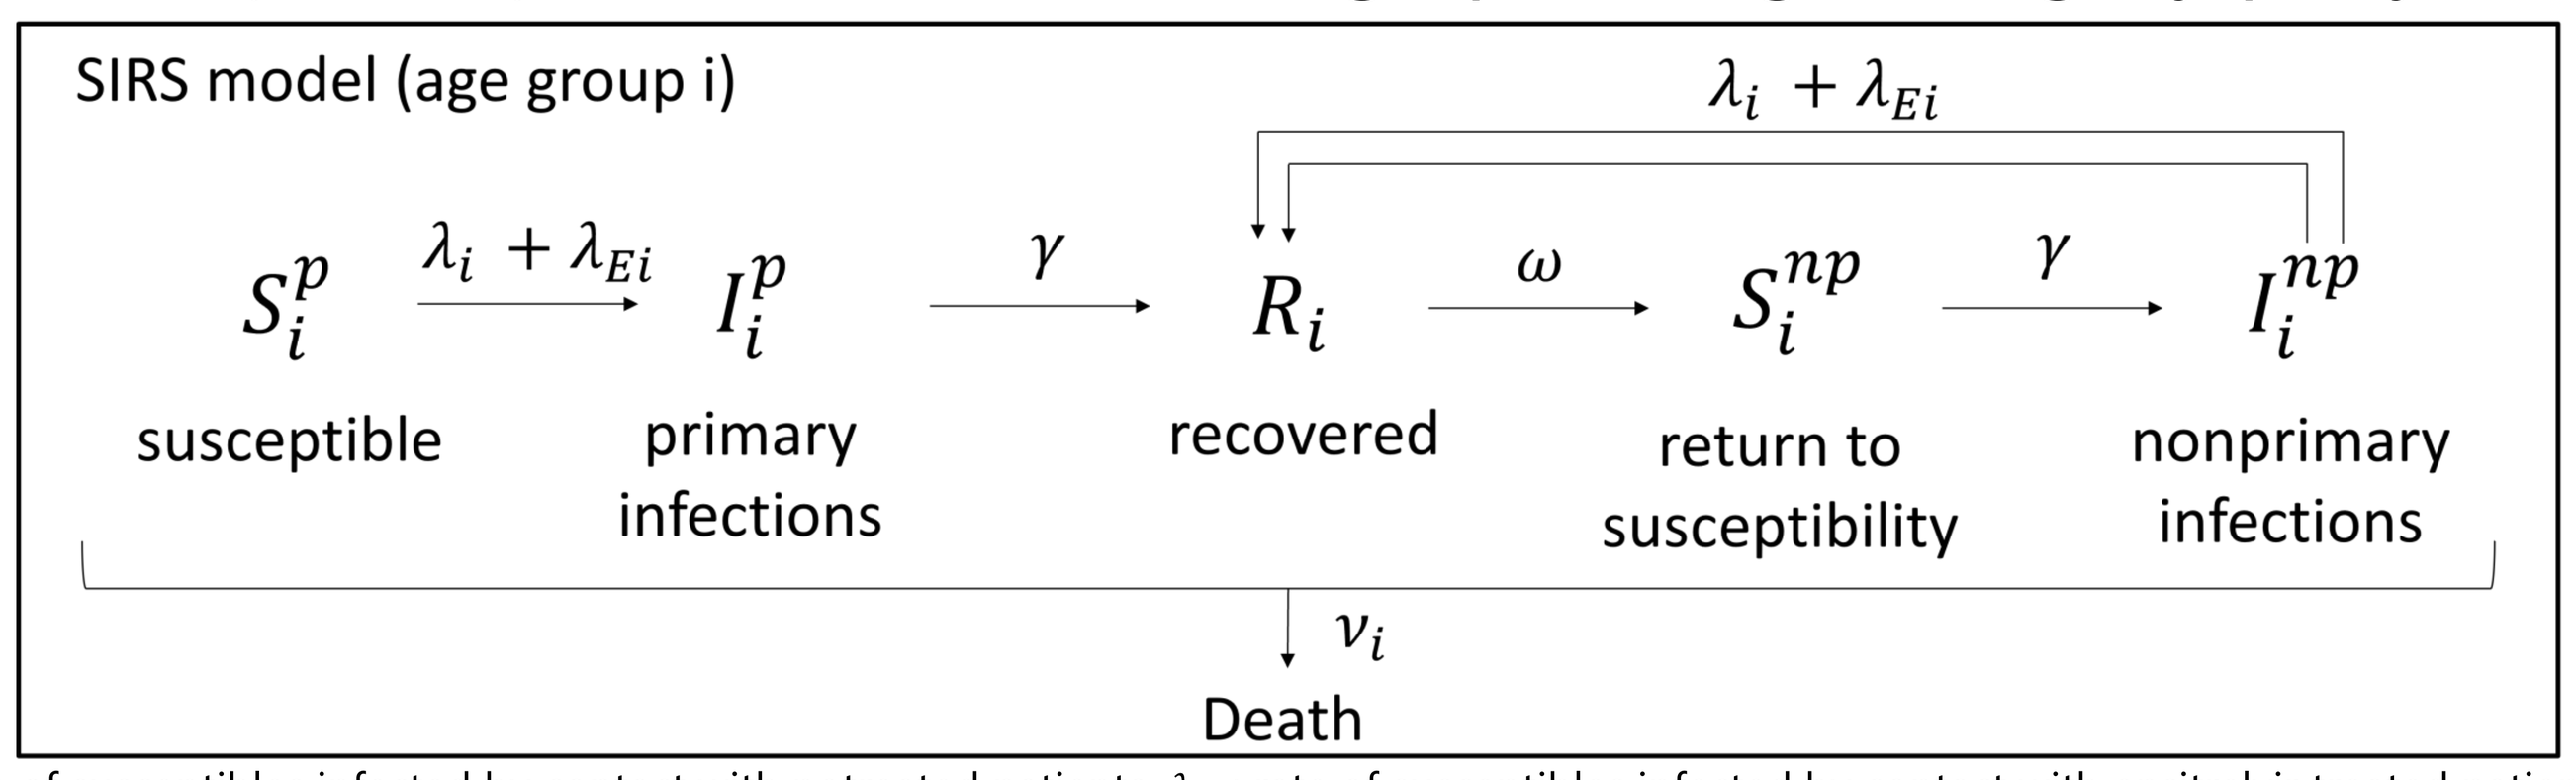
SIRS model (age group i)
$\lambda_i + \lambda_{Ei}$
S_i^p
susceptible
$\lambda_i + \lambda_{Ei}$
I_i^p
primary infections
γ
R_i
recovered
ω
S_i^{np}
return to susceptibility
γ
I_i^{np}
nonprimary infections
ν_i
Death

λ_i: rate of susceptibles infected by contact with notreated patients, λ_{Ei} : rate of susceptibles infected by contact with ensitrelvir treated patients, γ: rate of recovery, ω: rate of Immune lost, ν_i: rate of death

- Simulations were conducted considering the population structure and age-specific contact conditions in Japan. Parameters incorporated into the model included the estimated effect of transmission mitigation , the rate of treatment with ensitrelvir, and the duration of immunity after reinfection.
- Other parameters for SIRS model were set by applying to Omicron variants.

Results

Figure 1. Transmission mitigation by treatment start time

- The earlier treatment from the onset of symptoms, the greater efficacy of ensitrelvir in transmission-mitigation was shown.
- Based on the results of transmission mitigation estimation, we figured that the Log scale or the Dose-response scale of infectiousness profiles was appropriate in the case of Omicron variant. So, we assumed that transmission mitigation would be 10-15%, supposing that early treatment would be achieved, and conducted a long-term simulations.

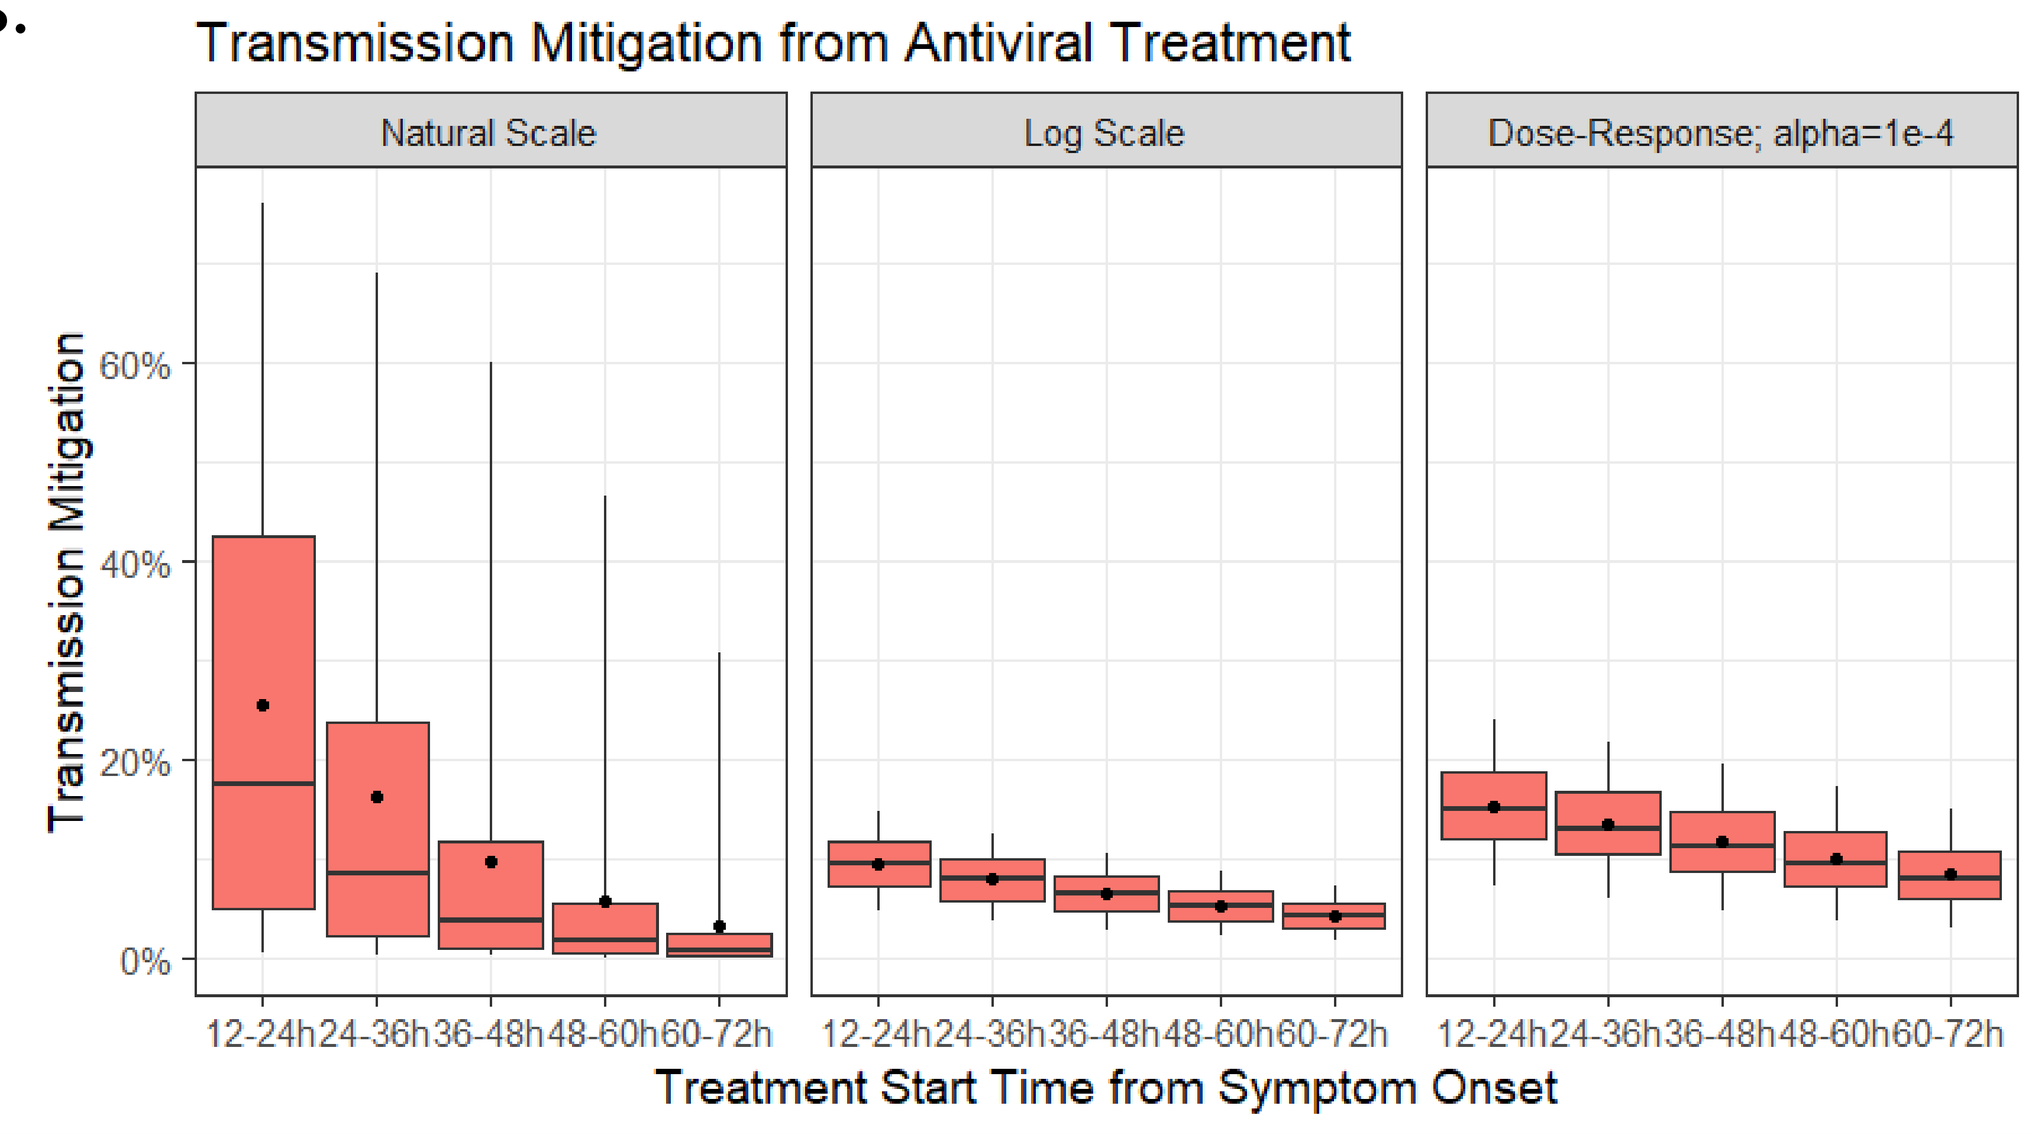
Transmission Mitigation from Antiviral Treatment
Natural Scale
Log Scale
Dose-Response; alpha=1e-4
Transmission Mitigation
60%
40%
20%
0%
12-24h 24-36h 36-48h 48-60h 60-72h
Treatment Start Time from Symptom Onset

	Natural Scale					Log Scale					Dose-Response; alpha=1e-4				
Treatment start time	12-24h	24-36h	36-48h	48-60h	60-72h	12-24h	24-36h	36-48h	48-60h	60-72h	12-24h	24-36h	36-48h	48-60h	60-72h
Median transmission mitigation (%)	17.6	8.5	3.8	1.7	0.8	9.4	7.9	6.5	5.2	4.1	14.9	13.0	11.2	9.5	7.9

Figure 2. Daily infection rates for the total population in Japan

- The SIRS model was used to estimate the effect of treatment of COVID-19 cases over a period of 20 years after the pandemic in Japan under several scenarios, considering the effect of transmission mitigation (10% and 15%), durations of immunity (6 month and 1 year) and treatment rate with ensitrelvir (10, 40 and 70%).
- During the endemic period, it was estimated that there would be approximately 1-2% of the total population with constant infection in all scenarios.

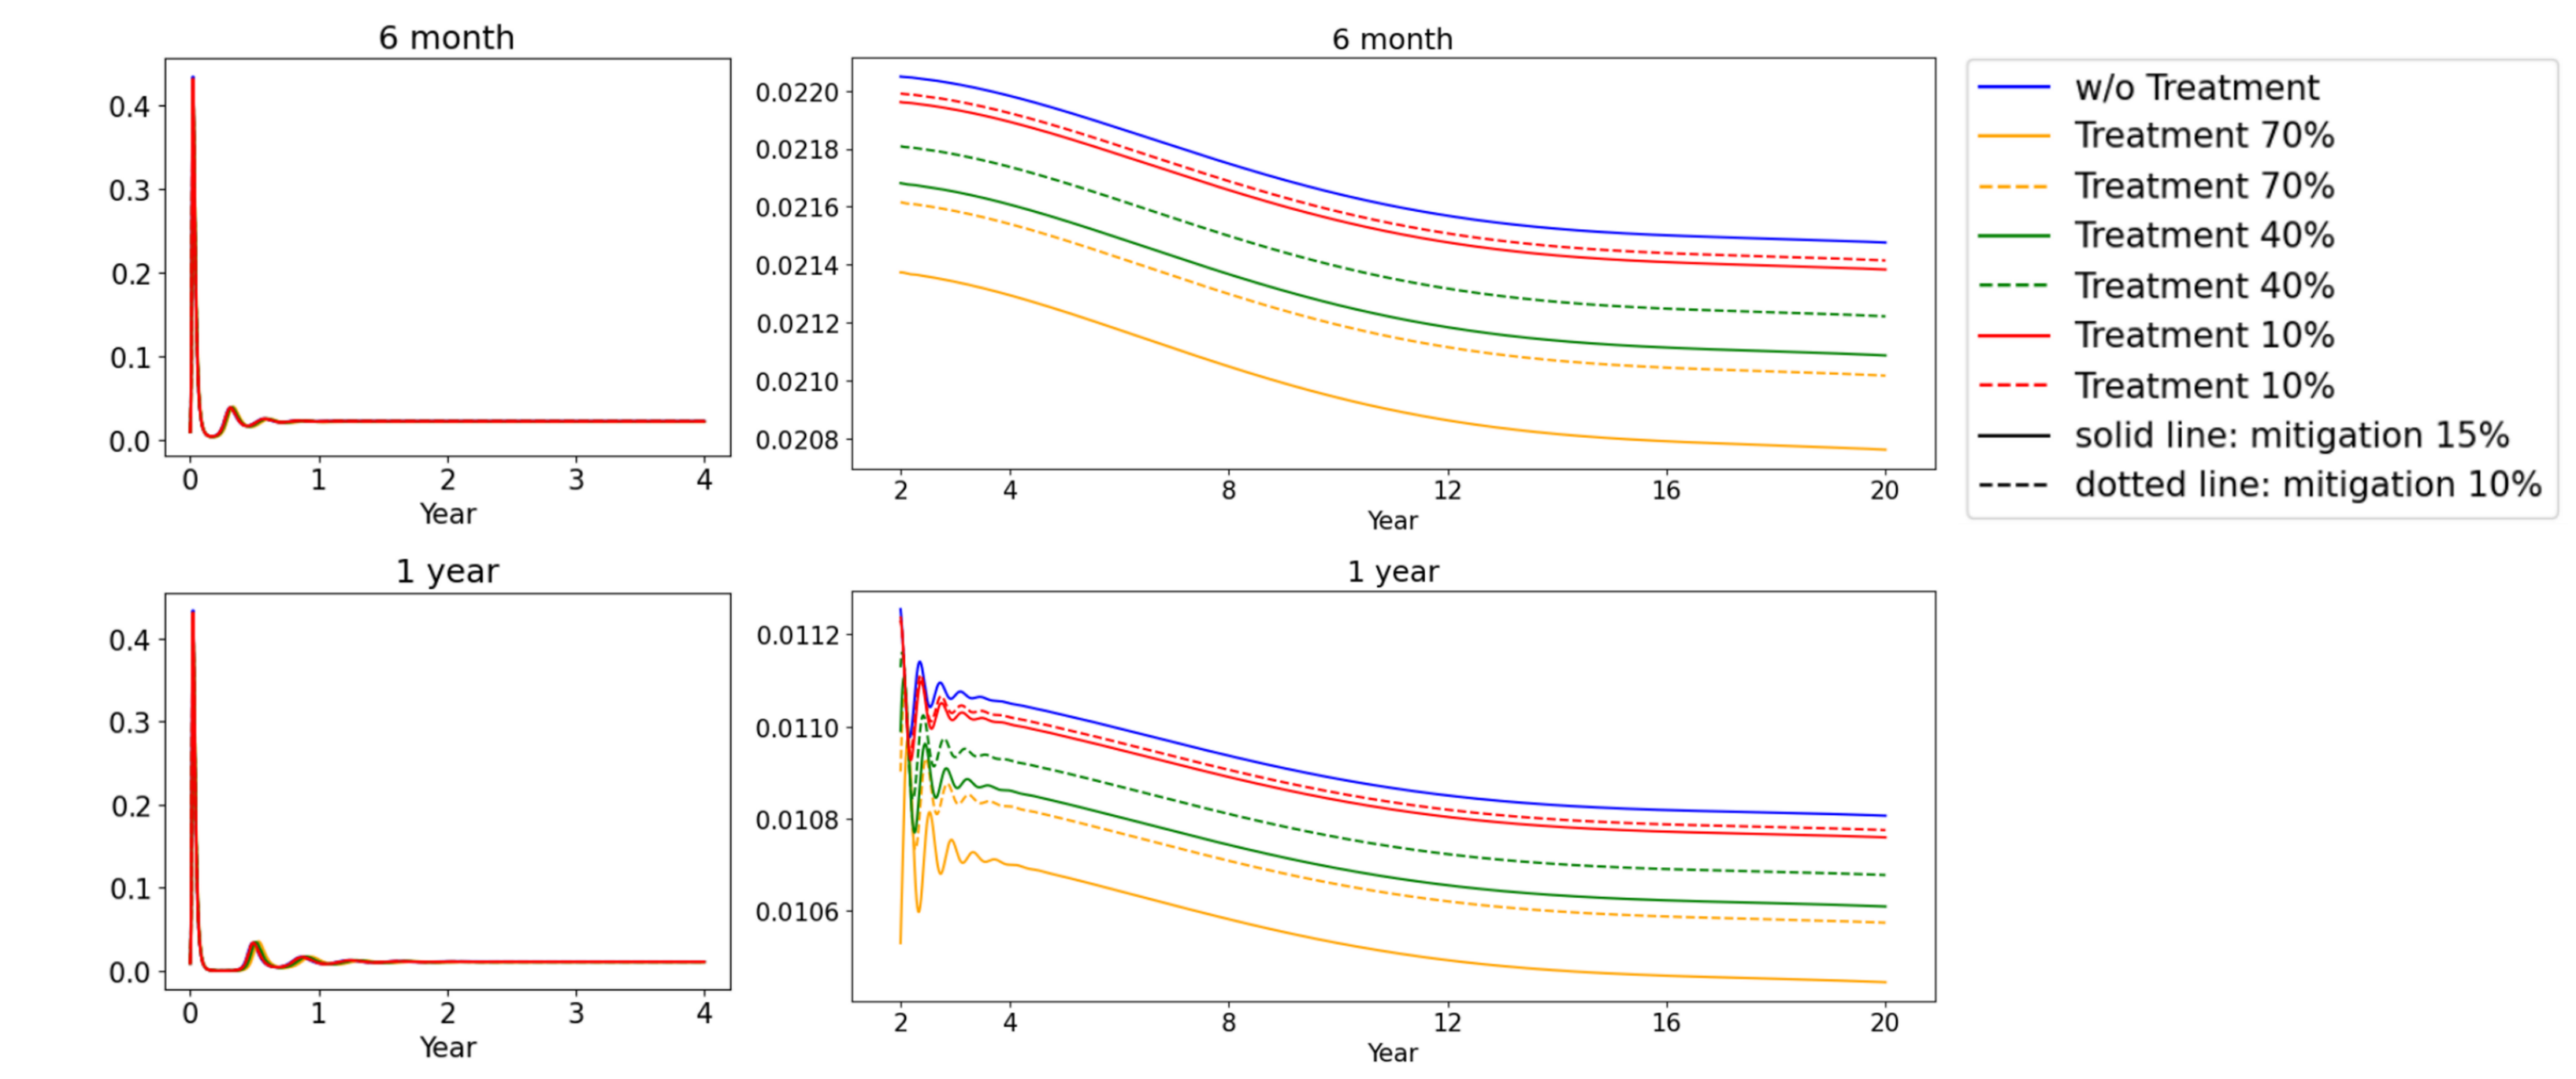
6 month
6 month
1 year
1 year
Year
w/o Treatment
Treatment 70%
Treatment 70%
Treatment 40%
Treatment 40%
Treatment 10%
Treatment 10%
solid line: mitigation 15%
dotted line: mitigation 10%

Table 1. Estimated reduction ratio (%) in the annual number of infections per treatment rate

- Based on the simulation, we estimated the annual reduction ratio in the number of infections per treatment rate compared to no treatment in Japan.
- Regardless of the duration of immunity, the reduction ratio in the number of infections at 10%, 40%, and 70% treatment rates with ensitrelvir was approximately 0.3%, 1.2%, and 2.1% at 10% transmission mitigation and 0.4%, 1.8%, and 3.2% at 15% transmission mitigation.
- This ratio of decline remained almost constant until 20 years after the pandemic to endemic phase.

	Mitigation: 10%						Mitigation: 15%					
	Immune duration: 6 month			Immune duration: 1 year			Immune duration: 6 month			Immune duration: 1 year		
Year	Treatment :70%	Treatment :40%	Treatment :10%	Treatment :70%	Treatment :40%	Treatment :10%	Treatment :70%	Treatment :40%	Treatment :10%	Treatment :70%	Treatment :40%	Treatment :10%
1	1.881	1.046	0.255	1.419	0.729	0.171	2.918	1.598	0.393	2.498	1.172	0.255
5	2.019	1.12	0.277	2.049	1.138	0.295	3.142	1.715	0.407	3.177	1.739	0.418
10	2.086	1.156	0.284	2.101	1.161	0.279	3.248	1.77	0.427	3.276	1.775	0.427
15	2.116	1.168	0.283	2.134	1.186	0.287	3.295	1.792	0.431	3.31	1.814	0.43
20	2.136	1.177	0.283	2.15	1.194	0.287	3.318	1.813	0.431	3.342	1.827	0.427

Conclusion

We concluded following suggestions in this study under several assumptions, including the effect of transmission mitigation, duration of immunity and treatment rate. These results should be confirmed with actual data in the near future.

- Early taking of antiviral drugs, including ensitrelvir, could have effect on reducing transmission.
- Approximately 10-15% risk reduction in transmission could be expected with ensitrelvir treatment.
- In case of 70% treatment rate among infected persons, it is estimated to reduce the number of infections by approximately 3% per year compared with no treatment.

Reference

[1] Yamaguchi D et al. Poster Abstracts, 10303. PAGE 2023.
[2] Yamaguchi D et al. Abstracts, The 44th Annual Scientific Meeting of The Japanese Society of Clinical Pharmacology and Therapeutics 2023.12.
[3] Asher J et al. PLOS Comput. Biol 19(1) 2023 e1010797
[4] Li R et al. Sci. Adv. 7(33) 2021 eabf9040

Conflict of interest statement

- M. Saito have no conflicts of interest directly relevant to the content of this research.
- S.Miyazawa, A. Hata, T. Baba, and Y. Kitanishi are employees of Shionogi & Co., Ltd.

ECCMID 2024, Barcelona, Spain