Poster 1310

35th ESCMID Global 2025
Vienna, Austria
11–15 April, 2025

Cefiderocol Activity Against Resistant Bacteria From Patients With Ventilator-Associated Pneumonia From European and US Hospitals

Sean T. Nguyen[1], Boudewijn L.M. DeJonge[1], Jason J. Bryowsky[1], Joshua M. Maher[2], Rodrigo E. Mendes[2], Christopher Longshaw[3], Hidenori Yamashiro[4], and Yoshinori Yamano[4]

[1]Shionogi Inc., Florham Park, New Jersey, USA; [2] Element, North Liberty, Iowa, USA; [3]Shionogi BV, London, UK; [4]Shionogi & Co., Ltd., Osaka, Japan

Contact: Sean T. Nguyen
Email: sean.nguyen@shionogi.com

SHIONOGI

BACKGROUND

- Treatment of ventilator-associated pneumonia (VAP) caused by Gram-negative pathogens is often complicated by resistance, resulting in limited treatment options.
- Cefiderocol is a siderophore cephalosporin with activity against Gram-negative bacteria, including multidrug-resistant isolates.

OBJECTIVE

- In this study, the activity of cefiderocol and comparators was evaluated against carbapenem-non-susceptible (CarbNS) and difficult-to-treat resistant (DTR) phenotypes of Gram-negative bacteria isolated from hospitalized patients with VAP from European and US hospitals.

METHODS

- 3,475 Gram-negative isolates from patients with VAP were collected in the SENTRY Antimicrobial Surveillance Program from 2020 to 2023; Enterobacterales (n=1,877; 54%), *Pseudomonas aeruginosa* (n=1,100; 32%), *Acinetobacter baumannii-calcoaceticus* species complex (n=240; 7%), and *Stenotrophomonas maltophilia* (n=219; 6%) were the most common species collected.
- Minimum inhibitory concentrations (MICs) were determined according to Clinical and Laboratory Standards Institute (CLSI) guidelines using broth microdilution with cation-adjusted Mueller–Hinton broth (CAMHB) for comparator agents and iron-depleted CAMHB for cefiderocol.
- CarbNS subsets were defined as non-susceptibility to meropenem and imipenem based on CLSI criteria.
- DTR phenotype was defined as non-susceptibility to aztreonam (except *A. baumannii*), cefepime, ceftazidime, ceftriaxone (for Enterobacterales only), imipenem, meropenem, ciprofloxacin, and levofloxacin. Susceptibility was assessed according to 2024 CLSI, US Food and Drug Administration (FDA), and European Committee on Antimicrobial Susceptibility Testing (EUCAST) breakpoints.

RESULTS

- 4.3% and 3.3% of Enterobacterales, 25.9% and 5.4% of *P. aeruginosa*, and 63.8% and 62.5% of *A. baumannii-calcoaceticus* species complex were CarbNS and DTR, respectively. 100% of *S. maltophilia* were CarbNS.
- CarbNS and DTR Enterobacterales isolates showed more elevated MIC values for cefiderocol compared with the overall Enterobacterales population, but this was less the case for *P. aeruginosa* and *A. baumannii-calcoaceticus* species complex (**Figure 1**).
- Nonetheless, cefiderocol showed good activity against all CarbNS and DTR isolates regardless of species, with >95% of isolates being susceptible by CLSI breakpoints, which was higher than comparator agents, including β-lactam–β-lactamase inhibitor combinations (**Table 1**).

Figure 1: Cefiderocol MIC distributions for Enterobacterales, *P. aeruginosa*, and *A. baumannii-calcoaceticus* species complex isolates collected from VAP patients, as part of SENTRY 2020–2023

*Criteria as published by CLSI (2024), EUCAST (2024), and US FDA (2024)

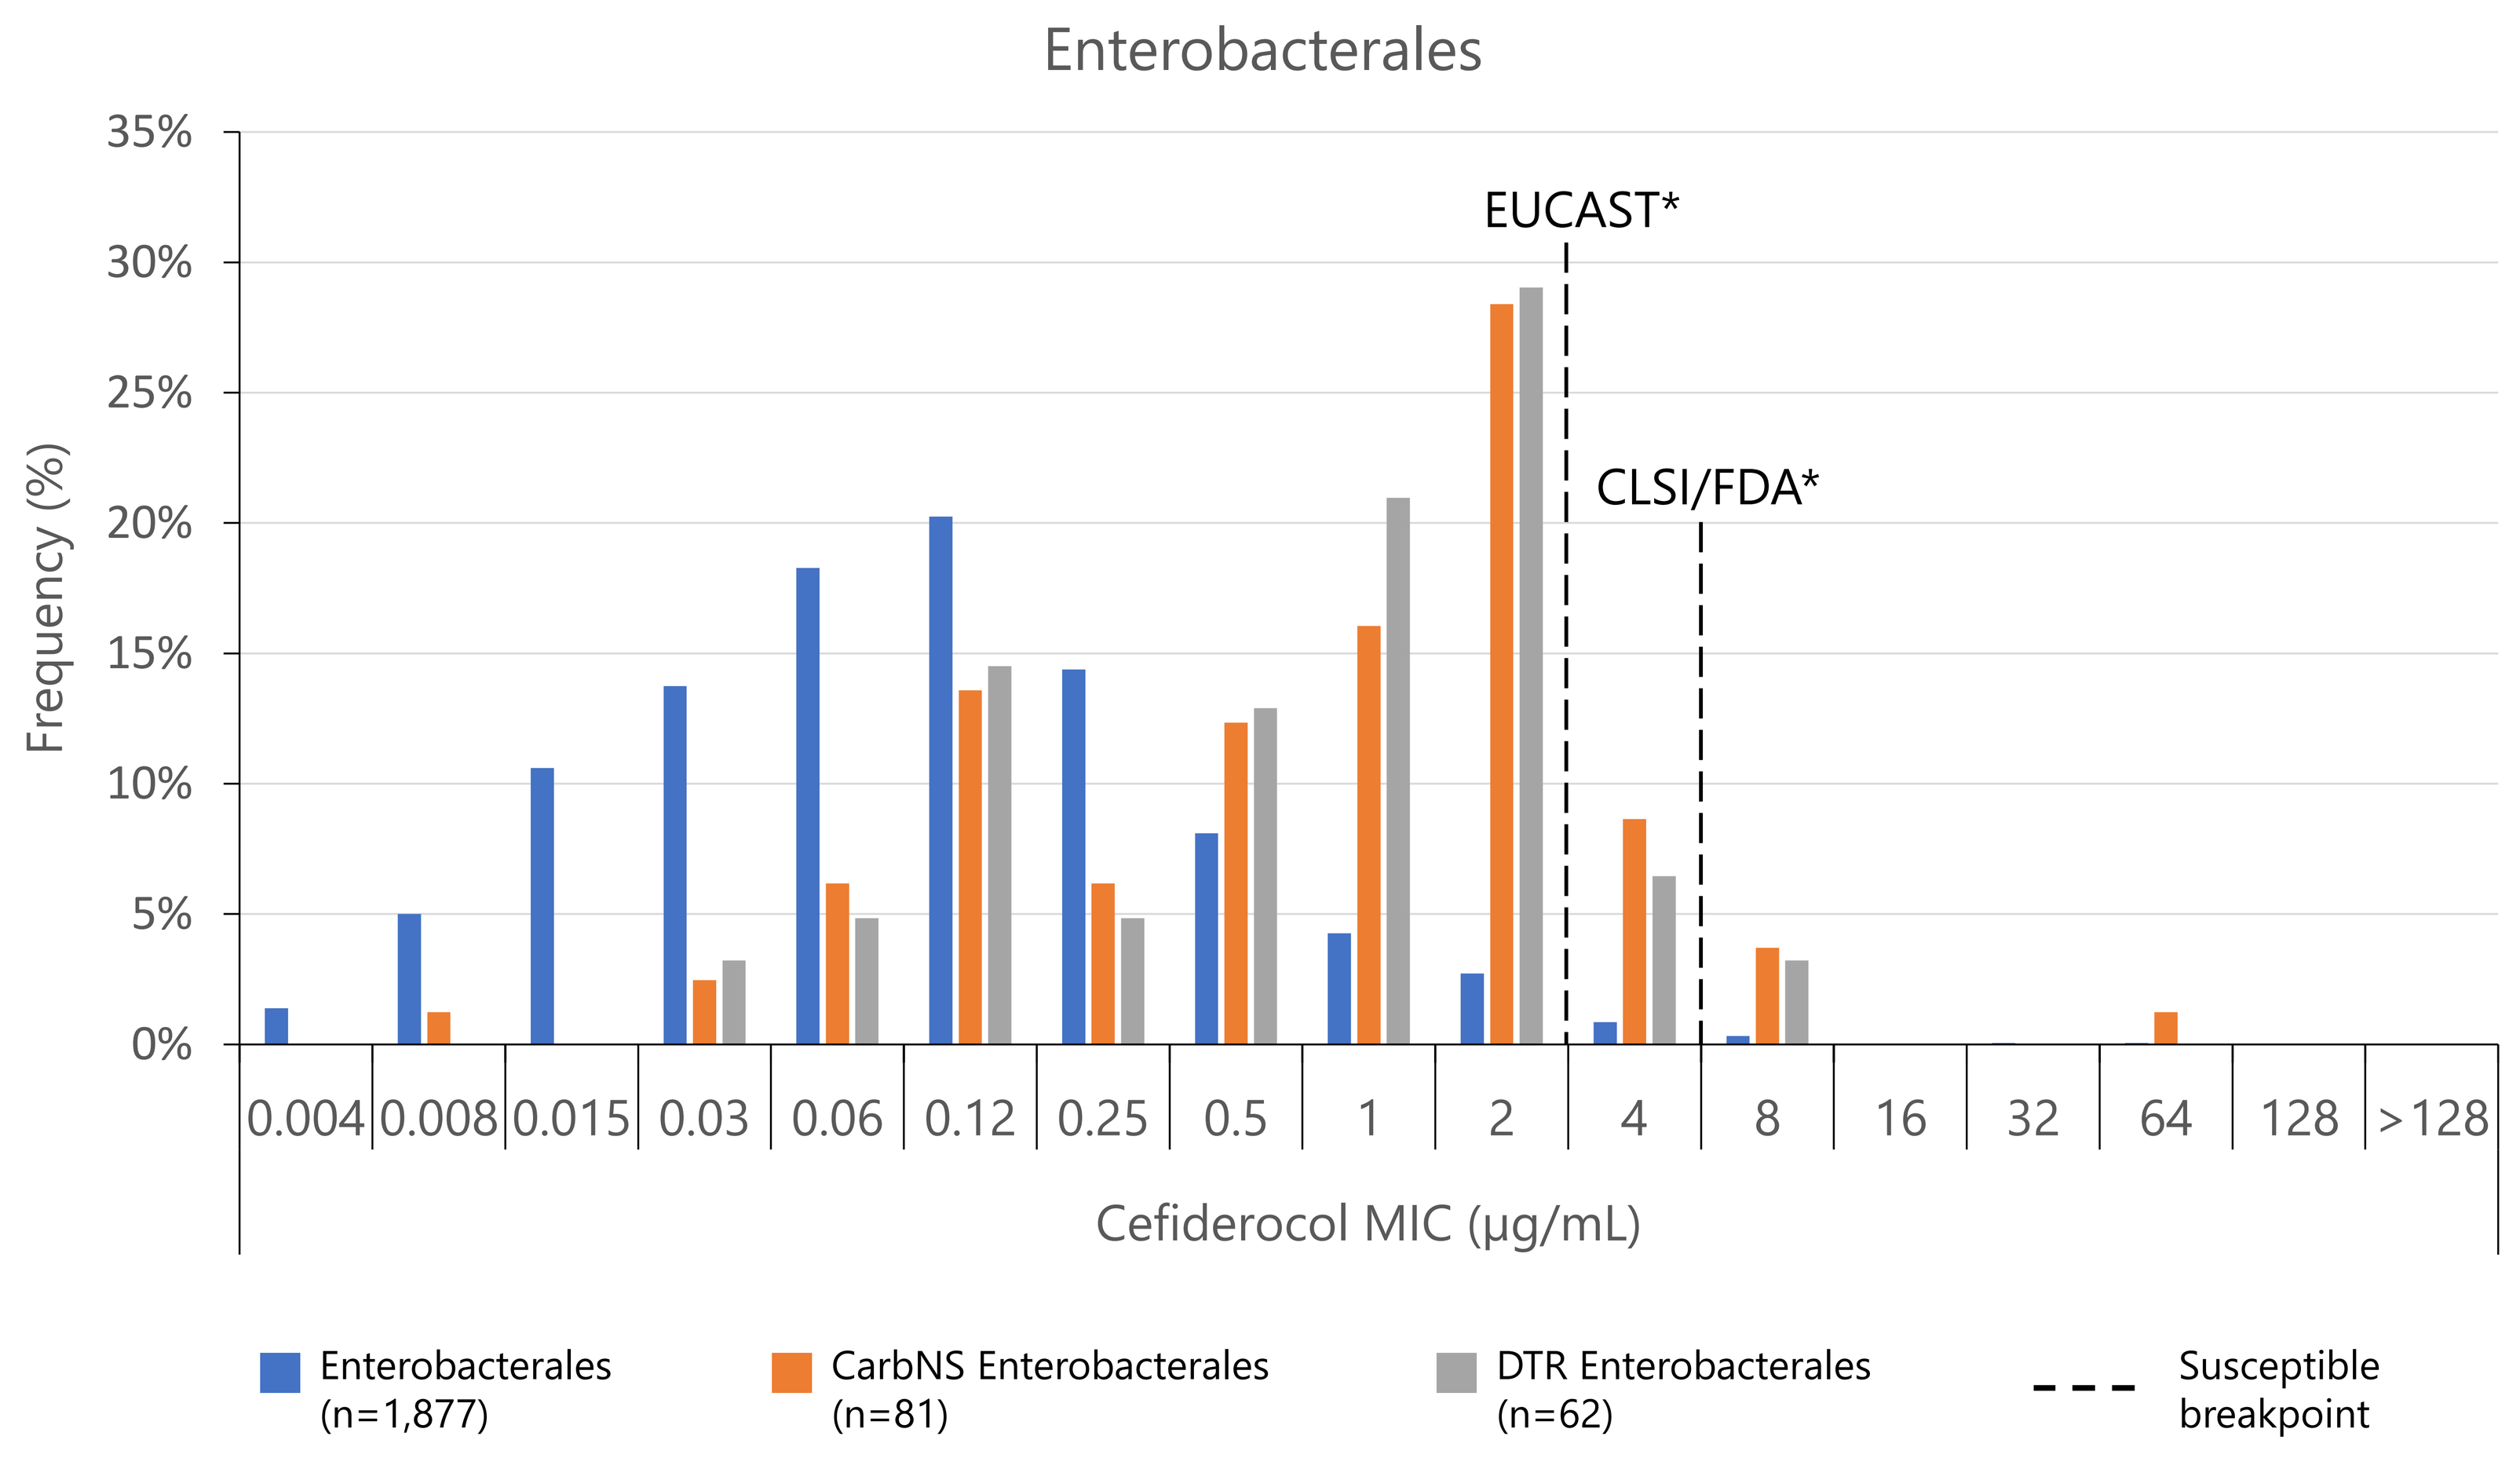

Enterobacterales
Frequency (%)
35%
30%
25%
20%
15%
10%
5%
0%
EUCAST*
CLSI/FDA*
0.004 0.008 0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 64 128 >128
Cefiderocol MIC (µg/mL)
Enterobacterales (n=1,877)
CarbNS Enterobacterales (n=81)
DTR Enterobacterales (n=62)
Susceptible breakpoint

Figure 1 cont.

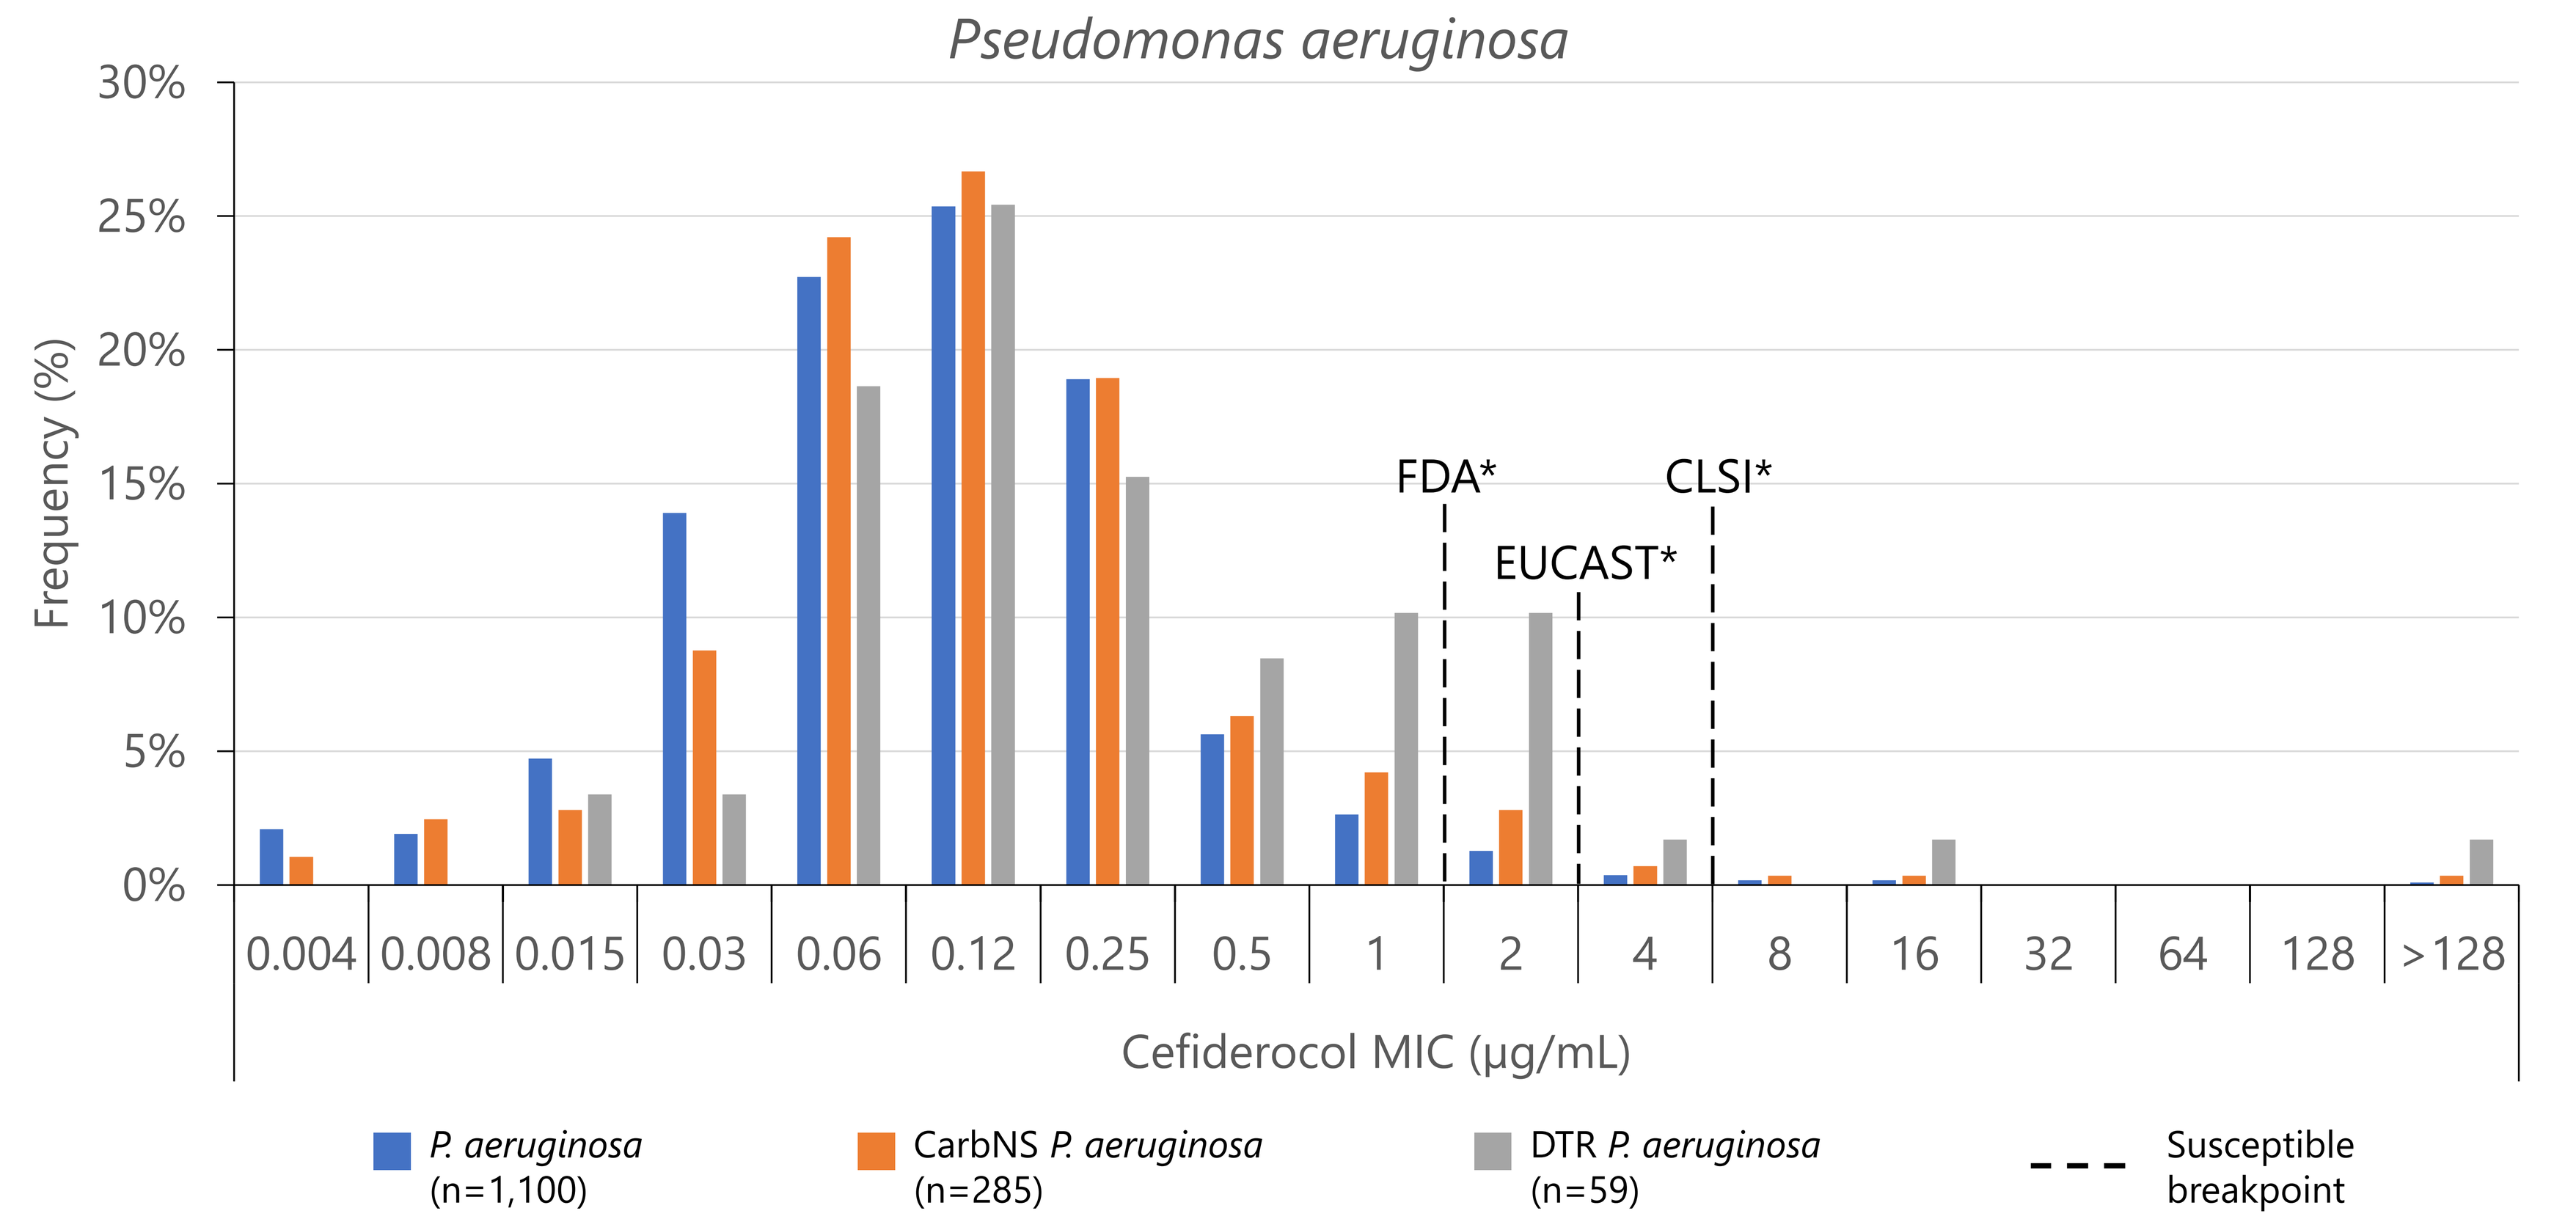

Pseudomonas aeruginosa
Frequency (%)
30%
25%
20%
15%
10%
5%
0%
FDA*
EUCAST*
CLSI*
0.004 0.008 0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 64 128 >128
Cefiderocol MIC (µg/mL)
P. aeruginosa (n=1,100)
CarbNS P. aeruginosa (n=285)
DTR P. aeruginosa (n=59)
Susceptible breakpoint

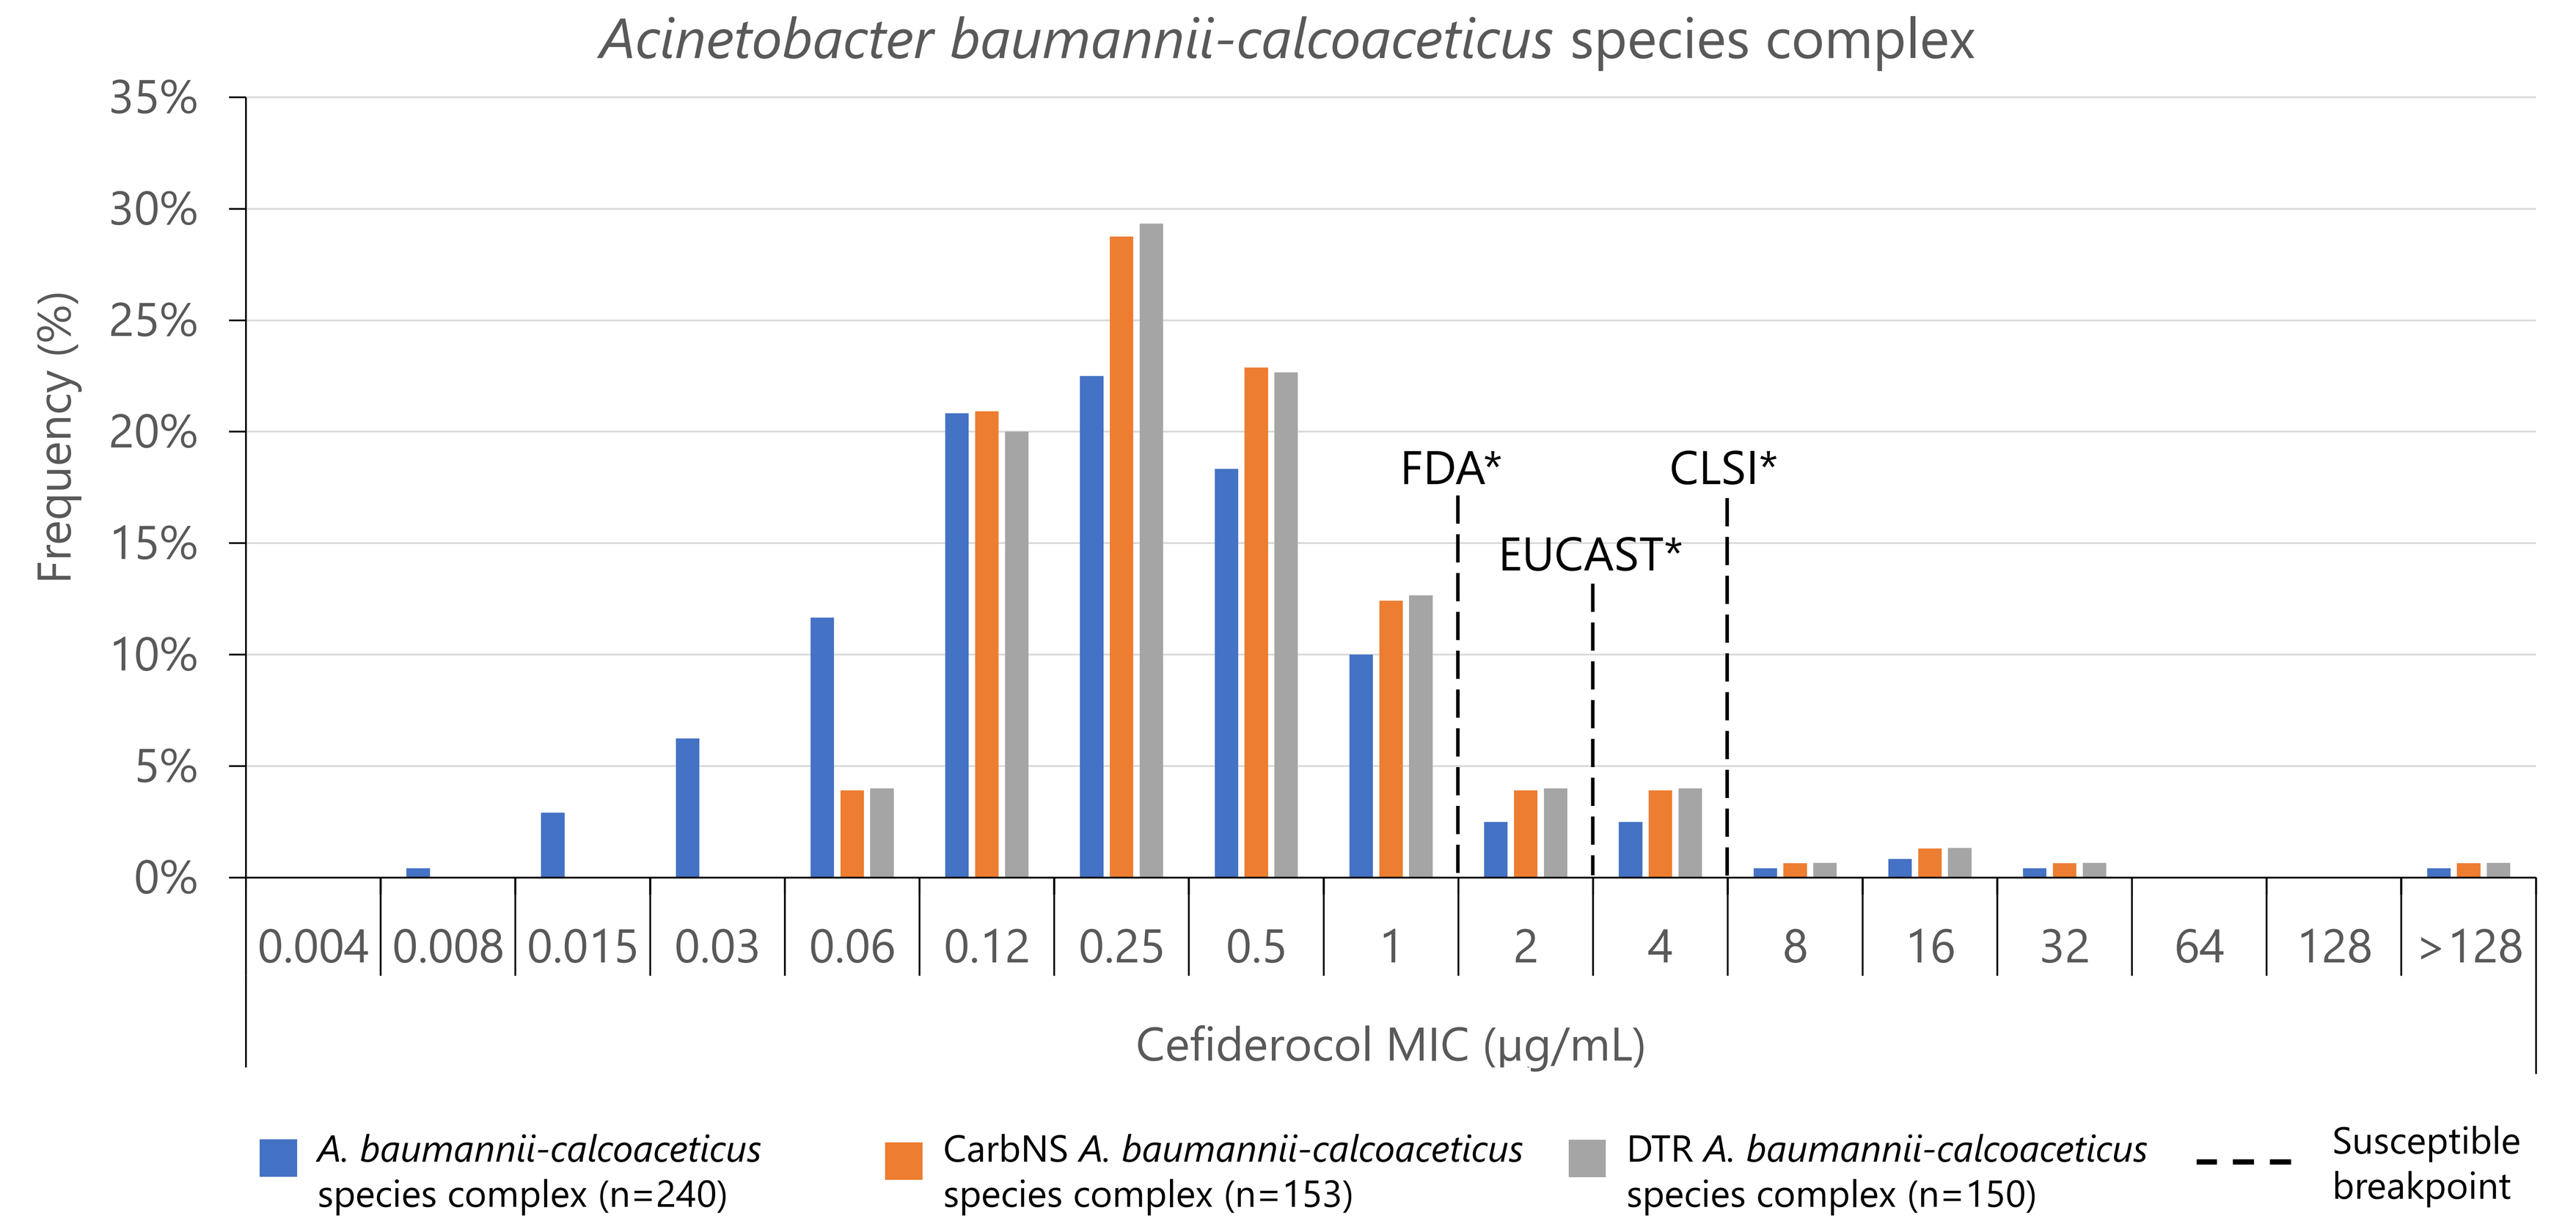

Acinetobacter baumannii-calcoaceticus species complex
Frequency (%)
35%
30%
25%
20%
15%
10%
5%
0%
FDA*
EUCAST*
CLSI*
0.004 0.008 0.015 0.03 0.06 0.12 0.25 0.5 1 2 4 8 16 32 64 128 >128
Cefiderocol MIC (µg/mL)
A. baumannii-calcoaceticus species complex (n=240)
CarbNS A. baumannii-calcoaceticus species complex (n=153)
DTR A. baumannii-calcoaceticus species complex (n=150)
Susceptible breakpoint

Table 1: Antibacterial activity of cefiderocol and comparators against CarbNS and DTR Gram-negative pathogens collected from patients with VAP in the SENTRY Antimicrobial Surveillance Program during 2020–2023

Organism group Agent	MIC_{50} (µg/mL)	MIC_{90} (µg/mL)	MIC range (µg/mL)	%S[a] CLSI	%S[a] FDA	%S[a] EUCAST
CarbNS Enterobacterales (n=81)						
Cefiderocol	1	4	0.008 to 64	95.1	95.1	86.4
Imipenem-relebactam	0.5	>8	0.12 to >8	50.6	50.6	50.6
Meropenem-vaborbactam	2	>8	≤0.015 to >8	53.1	53.1	60.5
Ceftazidime-avibactam	2	>32	0.06 to >32	60.5	60.5	60.5
Ceftolozane-tazobactam	>16	>16	1 to >16	4.9	4.9	4.9
DTR Enterobacterales (n=62)						
Cefiderocol	1	2	0.03 to 8	96.8	96.8	90.3
Imipenem-relebactam	0.5	>8	0.12 to >8	53.2	53.2	53.2
Meropenem-vaborbactam	2	>8	≤0.015 to >8	54.8	54.8	61.3
Ceftazidime-avibactam	2	>32	0.12 to >32	62.9	62.9	62.9
Ceftolozane-tazobactam	>16	>16	8 to >16	0.0	0.0	0.0
CarbNS *P. aeruginosa* (n=285)						
Cefiderocol	0.12	0.5	≤0.004 to >64	98.9	95.4	98.2
Imipenem-relebactam	1	8	0.25 to >8	80.4	80.4	80.4
Meropenem-vaborbactam	8	>8	1 to >8	NA	NA	50.2
Ceftazidime-avibactam	4	16	0.5 to >32	83.1	83.1	83.1
Ceftolozane-tazobactam	1	>16	0.25 to >16	84.9	84.9	84.9
DTR *P. aeruginosa* (n=59)						
Cefiderocol	0.12	2	0.015 to >64	96.6	84.7	94.9
Imipenem-relebactam	4	>8	0.25 to >8	44.1	44.1	44.1
Meropenem-vaborbactam	>8	>8	1 to >8	NA	NA	18.4
Ceftazidime-avibactam	16	>32	2 to >32	47.5	47.5	47.5
Ceftolozane-tazobactam	8	>16	1 to >16	49.2	49.2	49.2
CarbNS *A. baumannii-calcoaceticus* species complex (n=153)						
Cefiderocol	0.25	2	0.06 to >64	96.7	88.9	92.8
Imipenem-relebactam	>8	>8	>8 to >8	NA	0	NA
Ampicillin-sulbactam	64	>64	16 to >64	0	0	NA
Colistin	0.5	>8	0.12 to >8	0	0	69.9
Minocycline	16	16	0.25 to >32	20.9	20.9	NA
DTR *A. baumannii-calcoaceticus* species complex (n=150)						
Cefiderocol	0.25	2	0.06 to >64	96.7	88.7	92.7
Imipenem-relebactam	>8	>8	>8 to >8	NA	0	NA
Ampicillin-sulbactam	64	>64	16 to >64	0	0	NA
Colistin	0.5	>8	0.12 to >8	NA	NA	69.3
Minocycline	16	16	0.25 to >32	20.7	20.7	NA
***S. maltophilia* (n=219)**						
Cefiderocol	0.06	0.25	≤0.004 to 2	99.5	NA	100.0
Levofloxacin	1	4	0.12 to 32	84.9	NA	NA
Minocycline	0.5	1	0.12 to 8	93.2	NA	NA
Trimethoprim-sulfamethoxazole	≤0.12	0.5	≤0.12 to >4	96.3	NA	NA

S, susceptible; n, number of isolates; NA, not applicable.
[a]According to 2024 CLSI, FDA, and EUCAST breakpoints.

CONCLUSIONS

- Cefiderocol demonstrated greater activity than β-lactam–β-lactamase inhibitor combinations against Gram-negative isolates from patients with VAP, including those with a CarbNS or DTR phenotype for which treatment options are limited.
- No cross-resistance between cefiderocol and β-lactam–β-lactamase inhibitor combinations was observed.
- Cefiderocol is an important treatment option for hospitalized patients on mechanical ventilation at risk for an infection caused by Gram-negative pathogens.

Acknowledgments

This research was funded by Shionogi. STN, BJMD, JJB, CL, HY, YY are employees of the SHIONOGI Group. Editorial support was provided by Highfield, Oxford, UK; this support was funded by Shionogi & Co., Ltd., Osaka, Japan.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ESCMID Global 2025 and the authors of the poster.